Abstract #7336

B7-H7 is a novel ADC target for solid tumors and shows potent activity with multiple payload-linker technologies

Emilien Loeuillard[1], Bijan Etemad-Gilbertson[1], Susannah L Hewitt[1], Jamie Strand[1], Riale Gilligan[1], Silvia Ferrati[1], Tatiana Novobrantseva[1], Matthew Rausch[1].
[1]NextPoint Therapeutics, Cambridge, Massachusetts, USA

Background

B7-H7 (or HHLA2) has a high potential for tumor targeting:

- B7 family member ligand with both suppressive and stimulatory immune functions
- Limited expression in normal tissues
- Overexpressed in multiple cancer indications and its upregulation is associated with poor patient outcomes *(Wei L et al., Hum Cell 2020)*

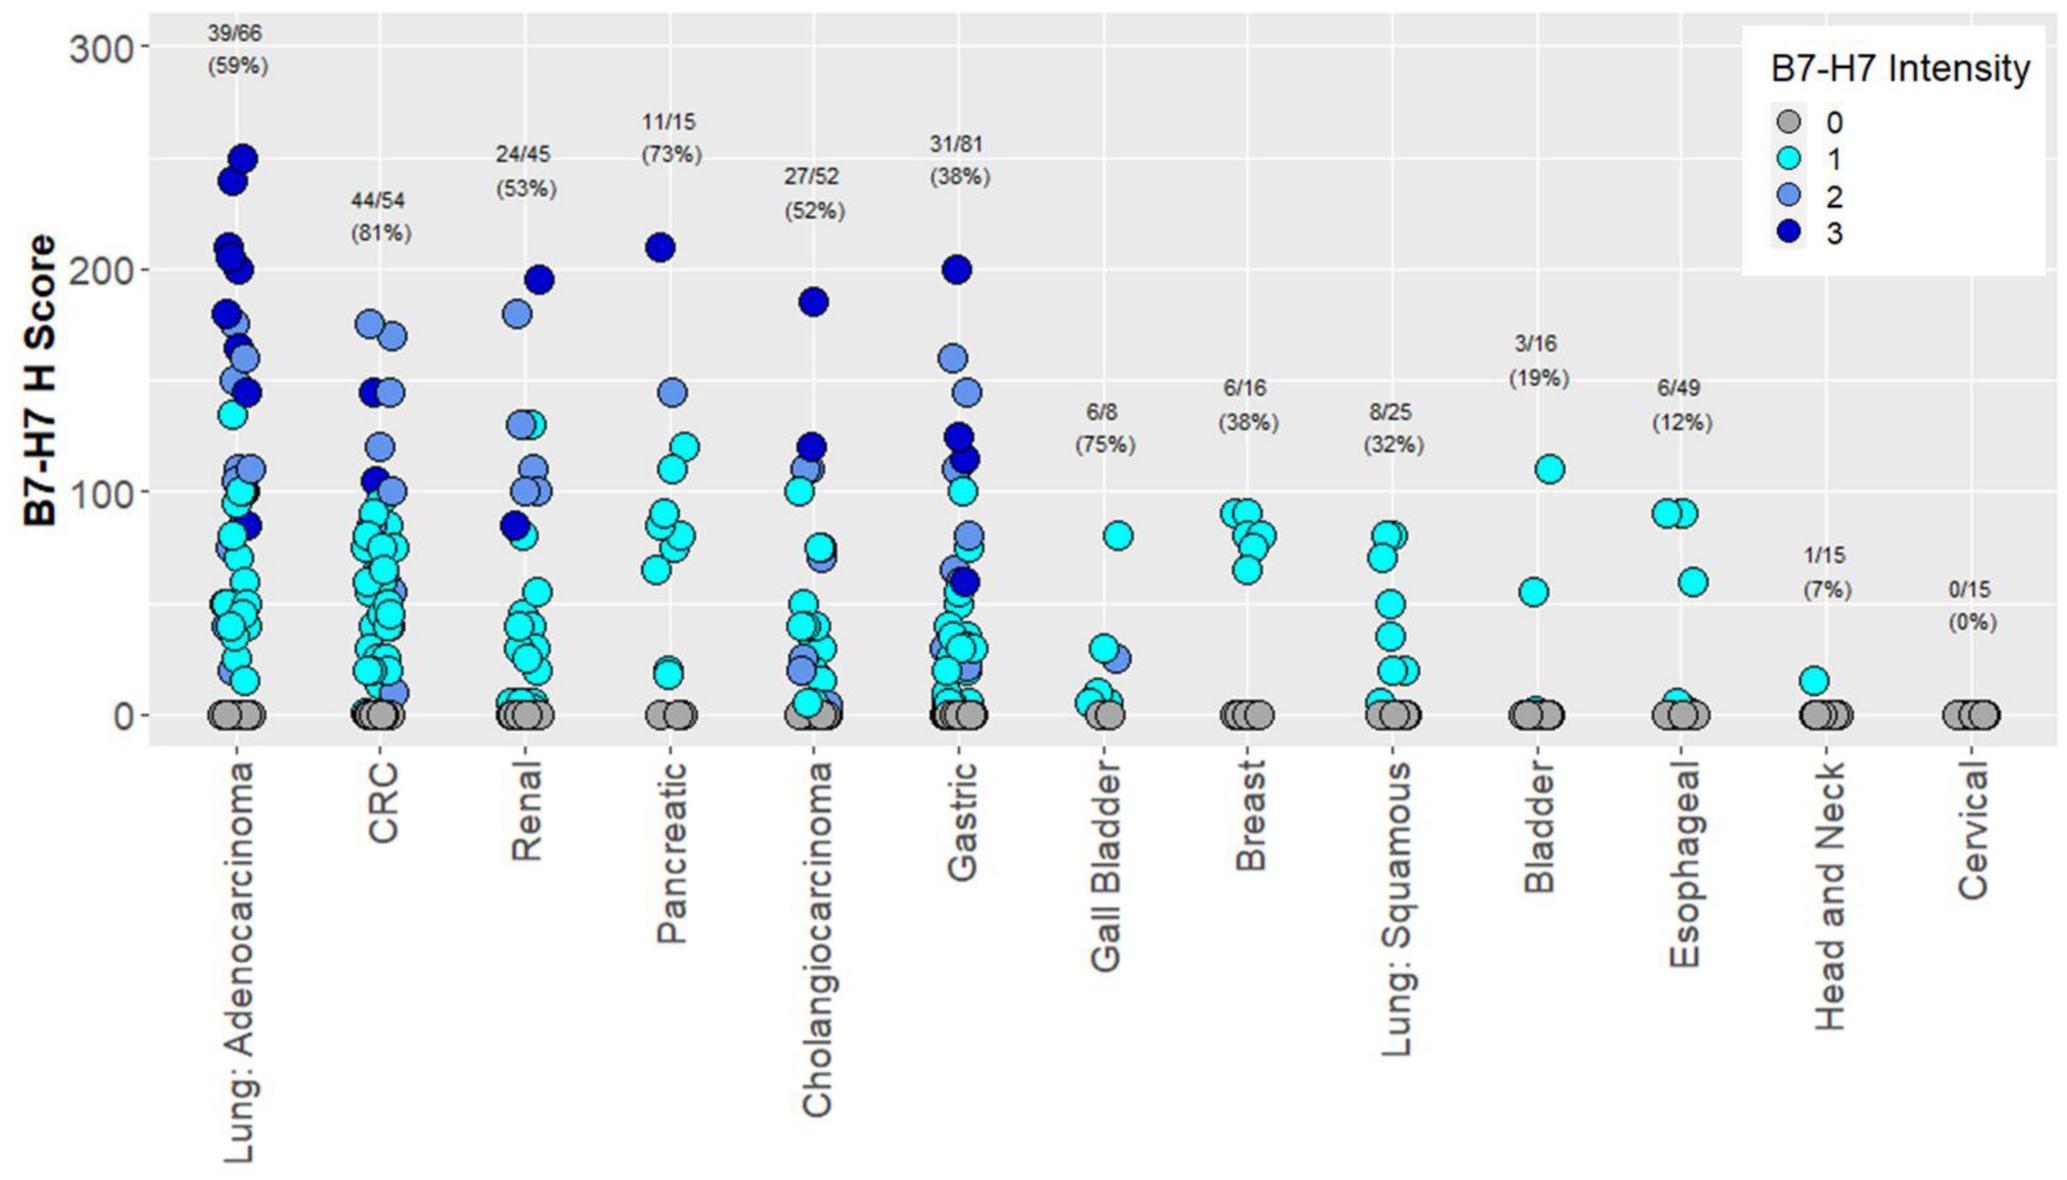
B7-H7 Intensity
0
1
2
3
B7-H7 H Score
300
200
100
0
39/66 (59%)
44/54 (81%)
24/45 (53%)
11/15 (73%)
27/52 (52%)
31/81 (38%)
6/8 (75%)
6/16 (38%)
8/25 (32%)
3/16 (19%)
6/49 (12%)
1/15 (7%)
0/15 (0%)
Lung: Adenocarcinoma
CRC
Renal
Pancreatic
Cholangiocarcinoma
Gastric
Gall Bladder
Breast
Lung: Squamous
Bladder
Esophageal
Head and Neck
Cervical

H score of B7-H7 assessed by immunochemistry across different human tumor types, showing high and broad expression of B7-H7 (poster #3302).

Antibody Drug Conjugates (ADCs) are targeted cancer therapies:

- ADCs are composed of an antibody, chemical linker and cytotoxic payload
- Ideal ADCs only release their payload to targeted cells
- ADCs induce apoptosis by both direct and bystander cytotoxicity
- Key components for ADC development:
 - Target antigen expression :
 → low on normal cells and high on tumor cells
 - Antibody :
 → high affinity, efficient internalization, long half-life
 - Linker and conjugation methods :
 → selected cleavability, no induction of aggregation
 - Cytotoxic payload :
 → high potency, stable, can be conjugated to antibody

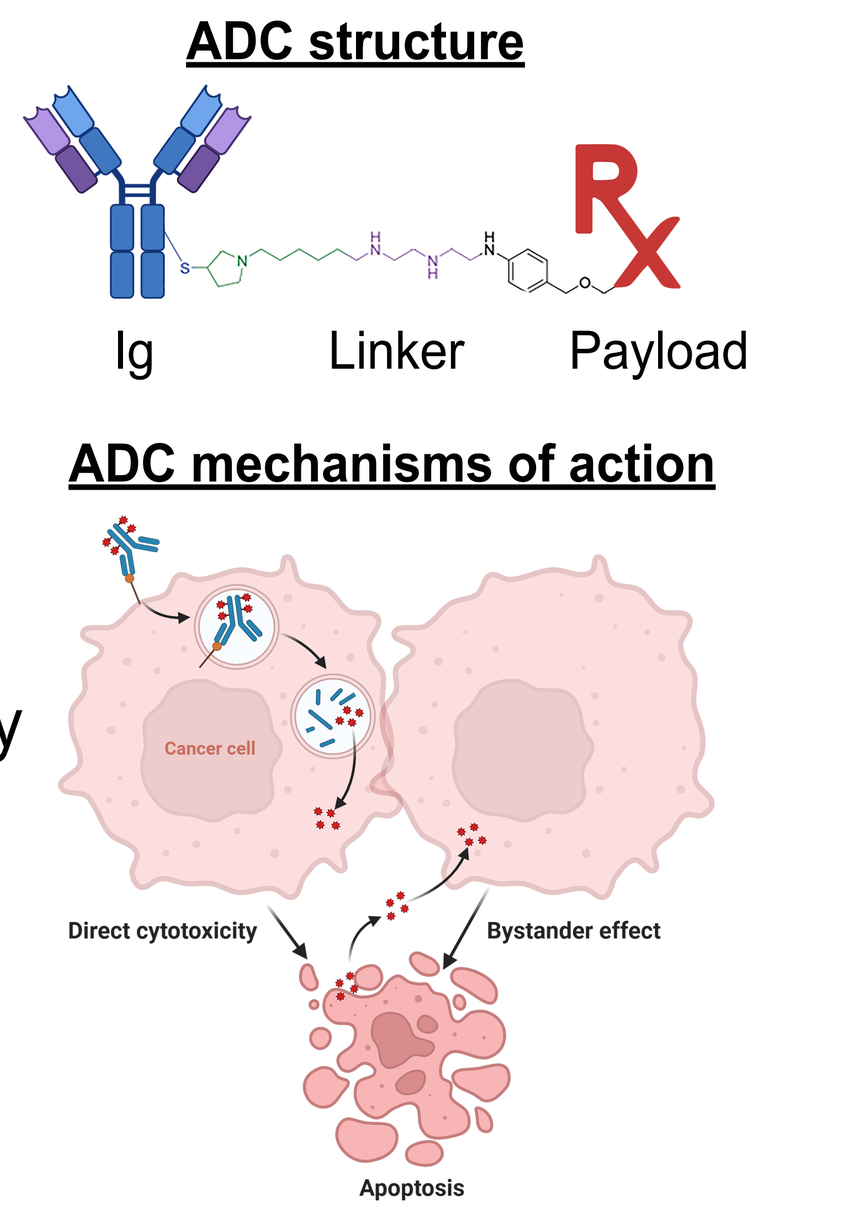
ADC structure
Ig
Linker
Payload
ADC mechanisms of action
Cancer cell
Direct cytotoxicity
Bystander effect
Apoptosis

NextPoint Therapeutics has developed B7-H7 targeted ADCs using:

- Antibodies that bind distinct B7-H7 epitopes, with cross-reacivity to cynomolgus protein
- Multiple linker-payload technologies
 - These were tested for *in vitro* and *in vivo* activity, and for their developability, enabling lead selection of the most potent compounds.

NPX125 Conserves On-Cell Binding Capacities

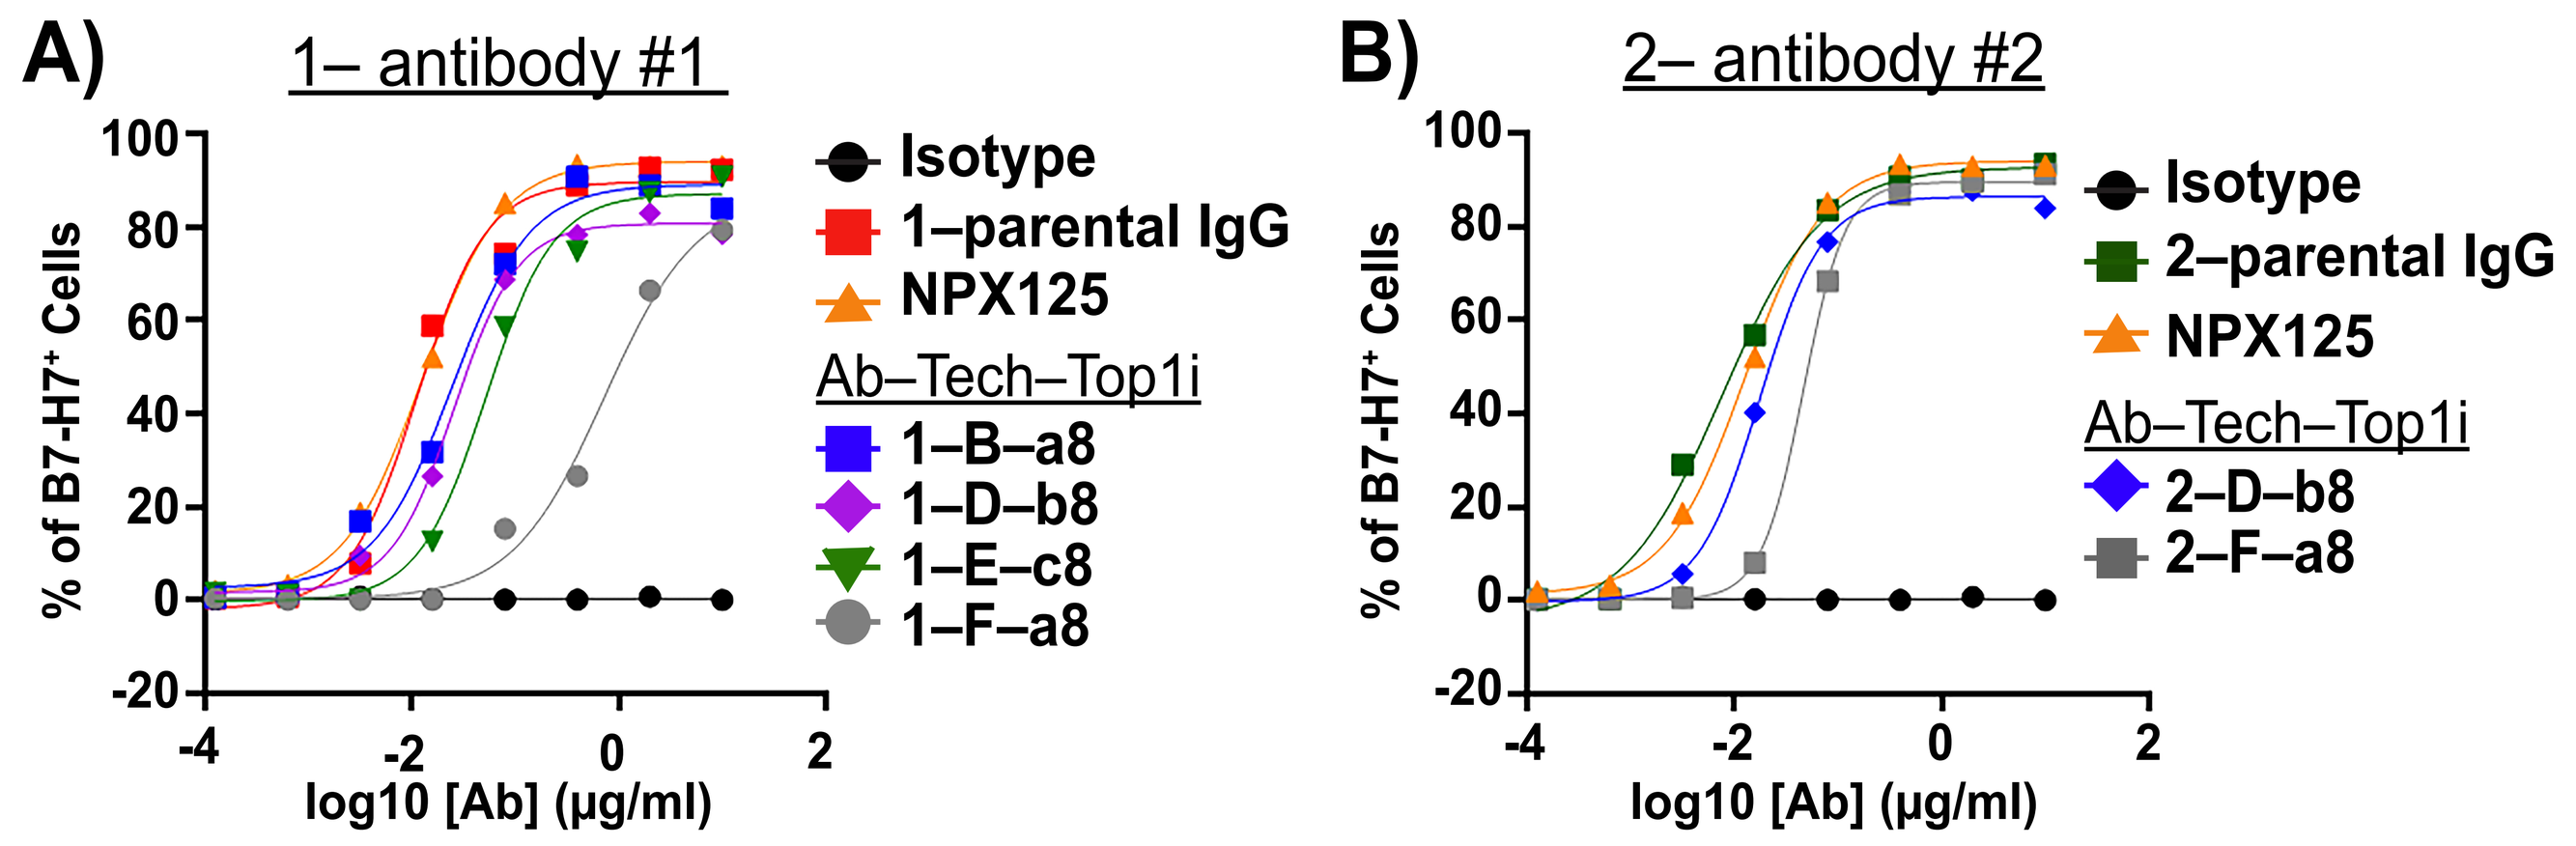
A)
1– antibody #1
Isotype
1–parental IgG
NPX125
Ab–Tech–Top1i
1–B–a8
1–D–b8
1–E–c8
1–F–a8
% of B7-H7+ Cells
log10 [Ab] (µg/ml)
B)
2– antibody #2
Isotype
2–parental IgG
NPX125
Ab–Tech–Top1i
2–D–b8
2–F–a8

(A–B) Similar binding capacity between parental antibodies and NPX125, with comparison to other ADC formats. A204 cells overexpressing B7-H7 were incubated with antibodies or ADCs and analyzed by flow cytometry. Binding levels were detected using a secondary human IgG Fc antibody. Key to ADC names: 1 or 2 represents the parental antibody binder, A–F is the linker technology used, and a, b or c details the Topoisomerase 1 inhibitor payload-type with DAR 4 or 8 indicated.

NPX125 is Internalized by Different Tumor Cell Types

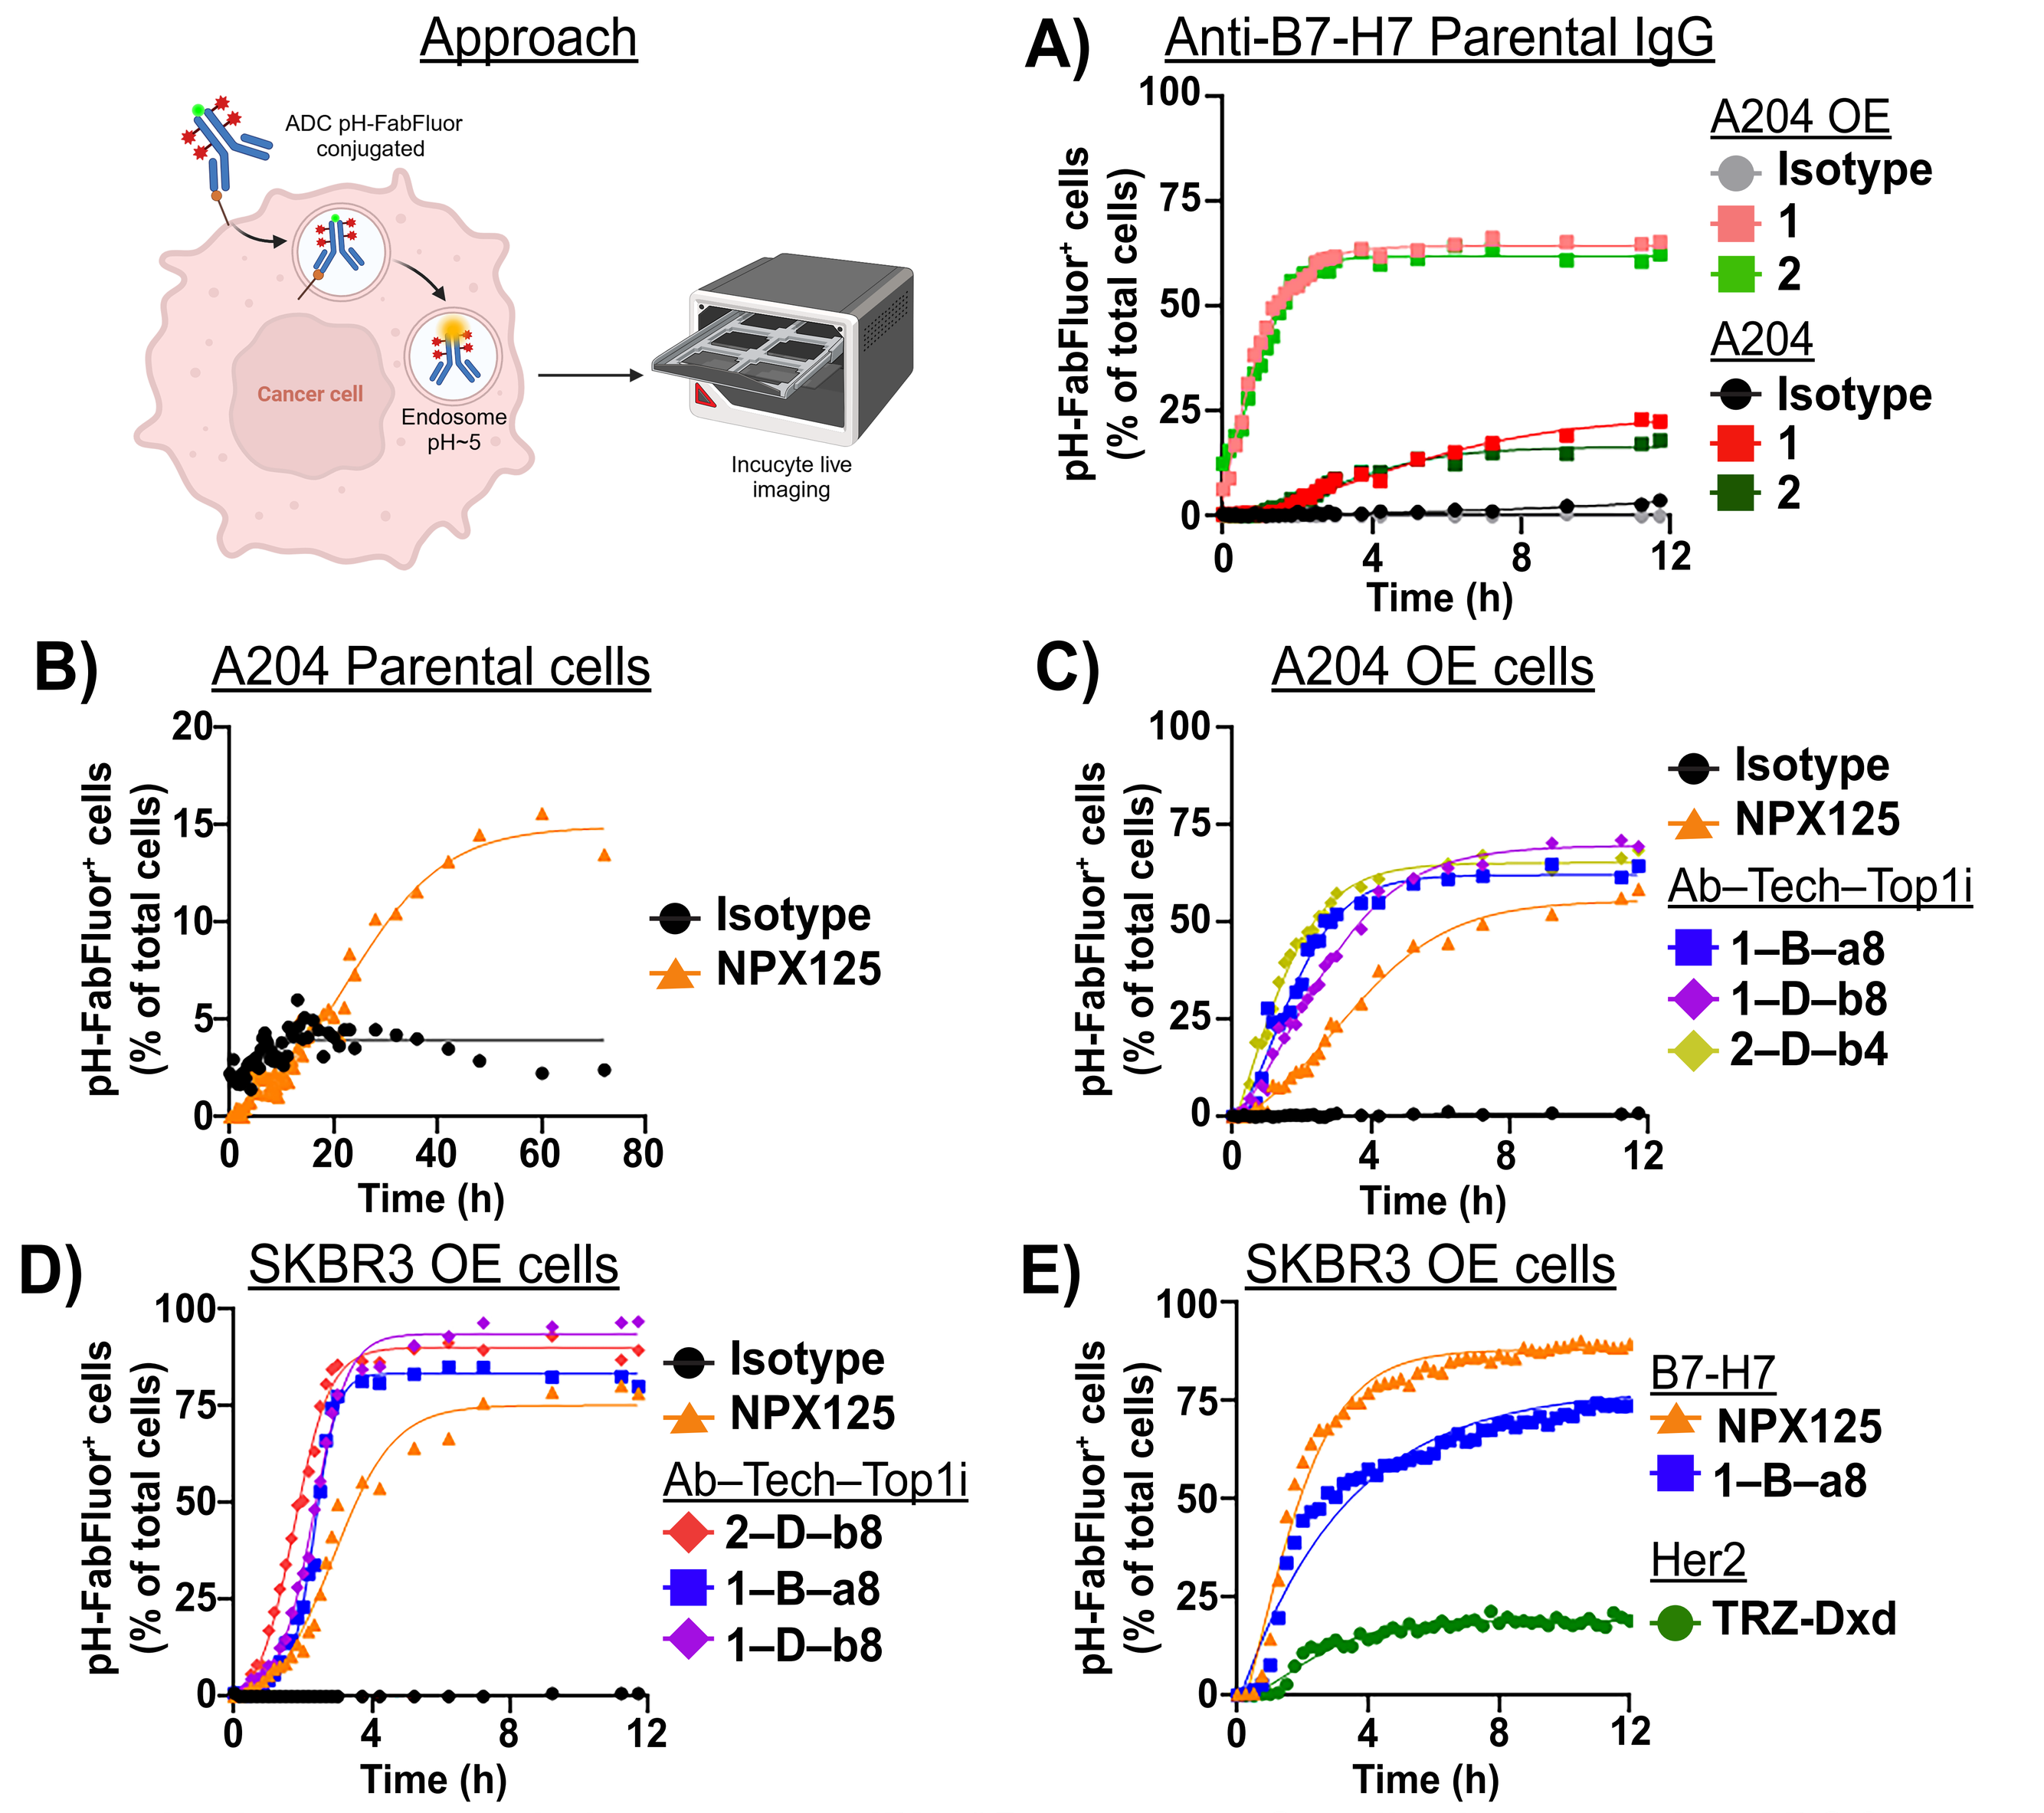
Approach
ADC pH-FabFluor conjugated
Cancer cell
Endosome pH~5
Incucyte live imaging
A)
Anti-B7-H7 Parental IgG
A204 OE
Isotype
1
2
A204
Isotype
1
2
pH-FabFluor+ cells (% of total cells)
Time (h)
B)
A204 Parental cells
Isotype
NPX125
C)
A204 OE cells
Isotype
NPX125
Ab–Tech–Top1i
1–B–a8
1–D–b8
2–D–b4
D)
SKBR3 OE cells
Isotype
NPX125
Ab–Tech–Top1i
2–D–b8
1–B–a8
1–D–b8
E)
SKBR3 OE cells
B7-H7
NPX125
1–B–a8
Her2
TRZ-Dxd

Internalization rates are similar between NPX125, parental IgG antibodies and other ADC formats. Parental A204, A204 or SKBR3 overexpresssing B7-H7 (A204 OE and SKBR3 OE, respectively) cell lines were treated with constructs conjugated to orange pH-Fabfluor, and tracked via IncuCyte. **(A)** Comparison of internalization rates of parental B7-H7 IgG in A204 (low to moderate B7-H7 expression) vs. A204 OE cells. **(B–D)** Comparison of internalization rates of ADCs targeting B7-H7 in A204 cells (B), A204 OE cells (C) or SKBR3 OE cells (D) (both high expression). **(E)** Comparison of internalization rates of ADCs targeting B7-H7 and Trastuzumab-Dxd targeting Her2 in SKBR3 OE cells.

NPX125 Induces Cytotoxicity in Different Tumor Cell Types

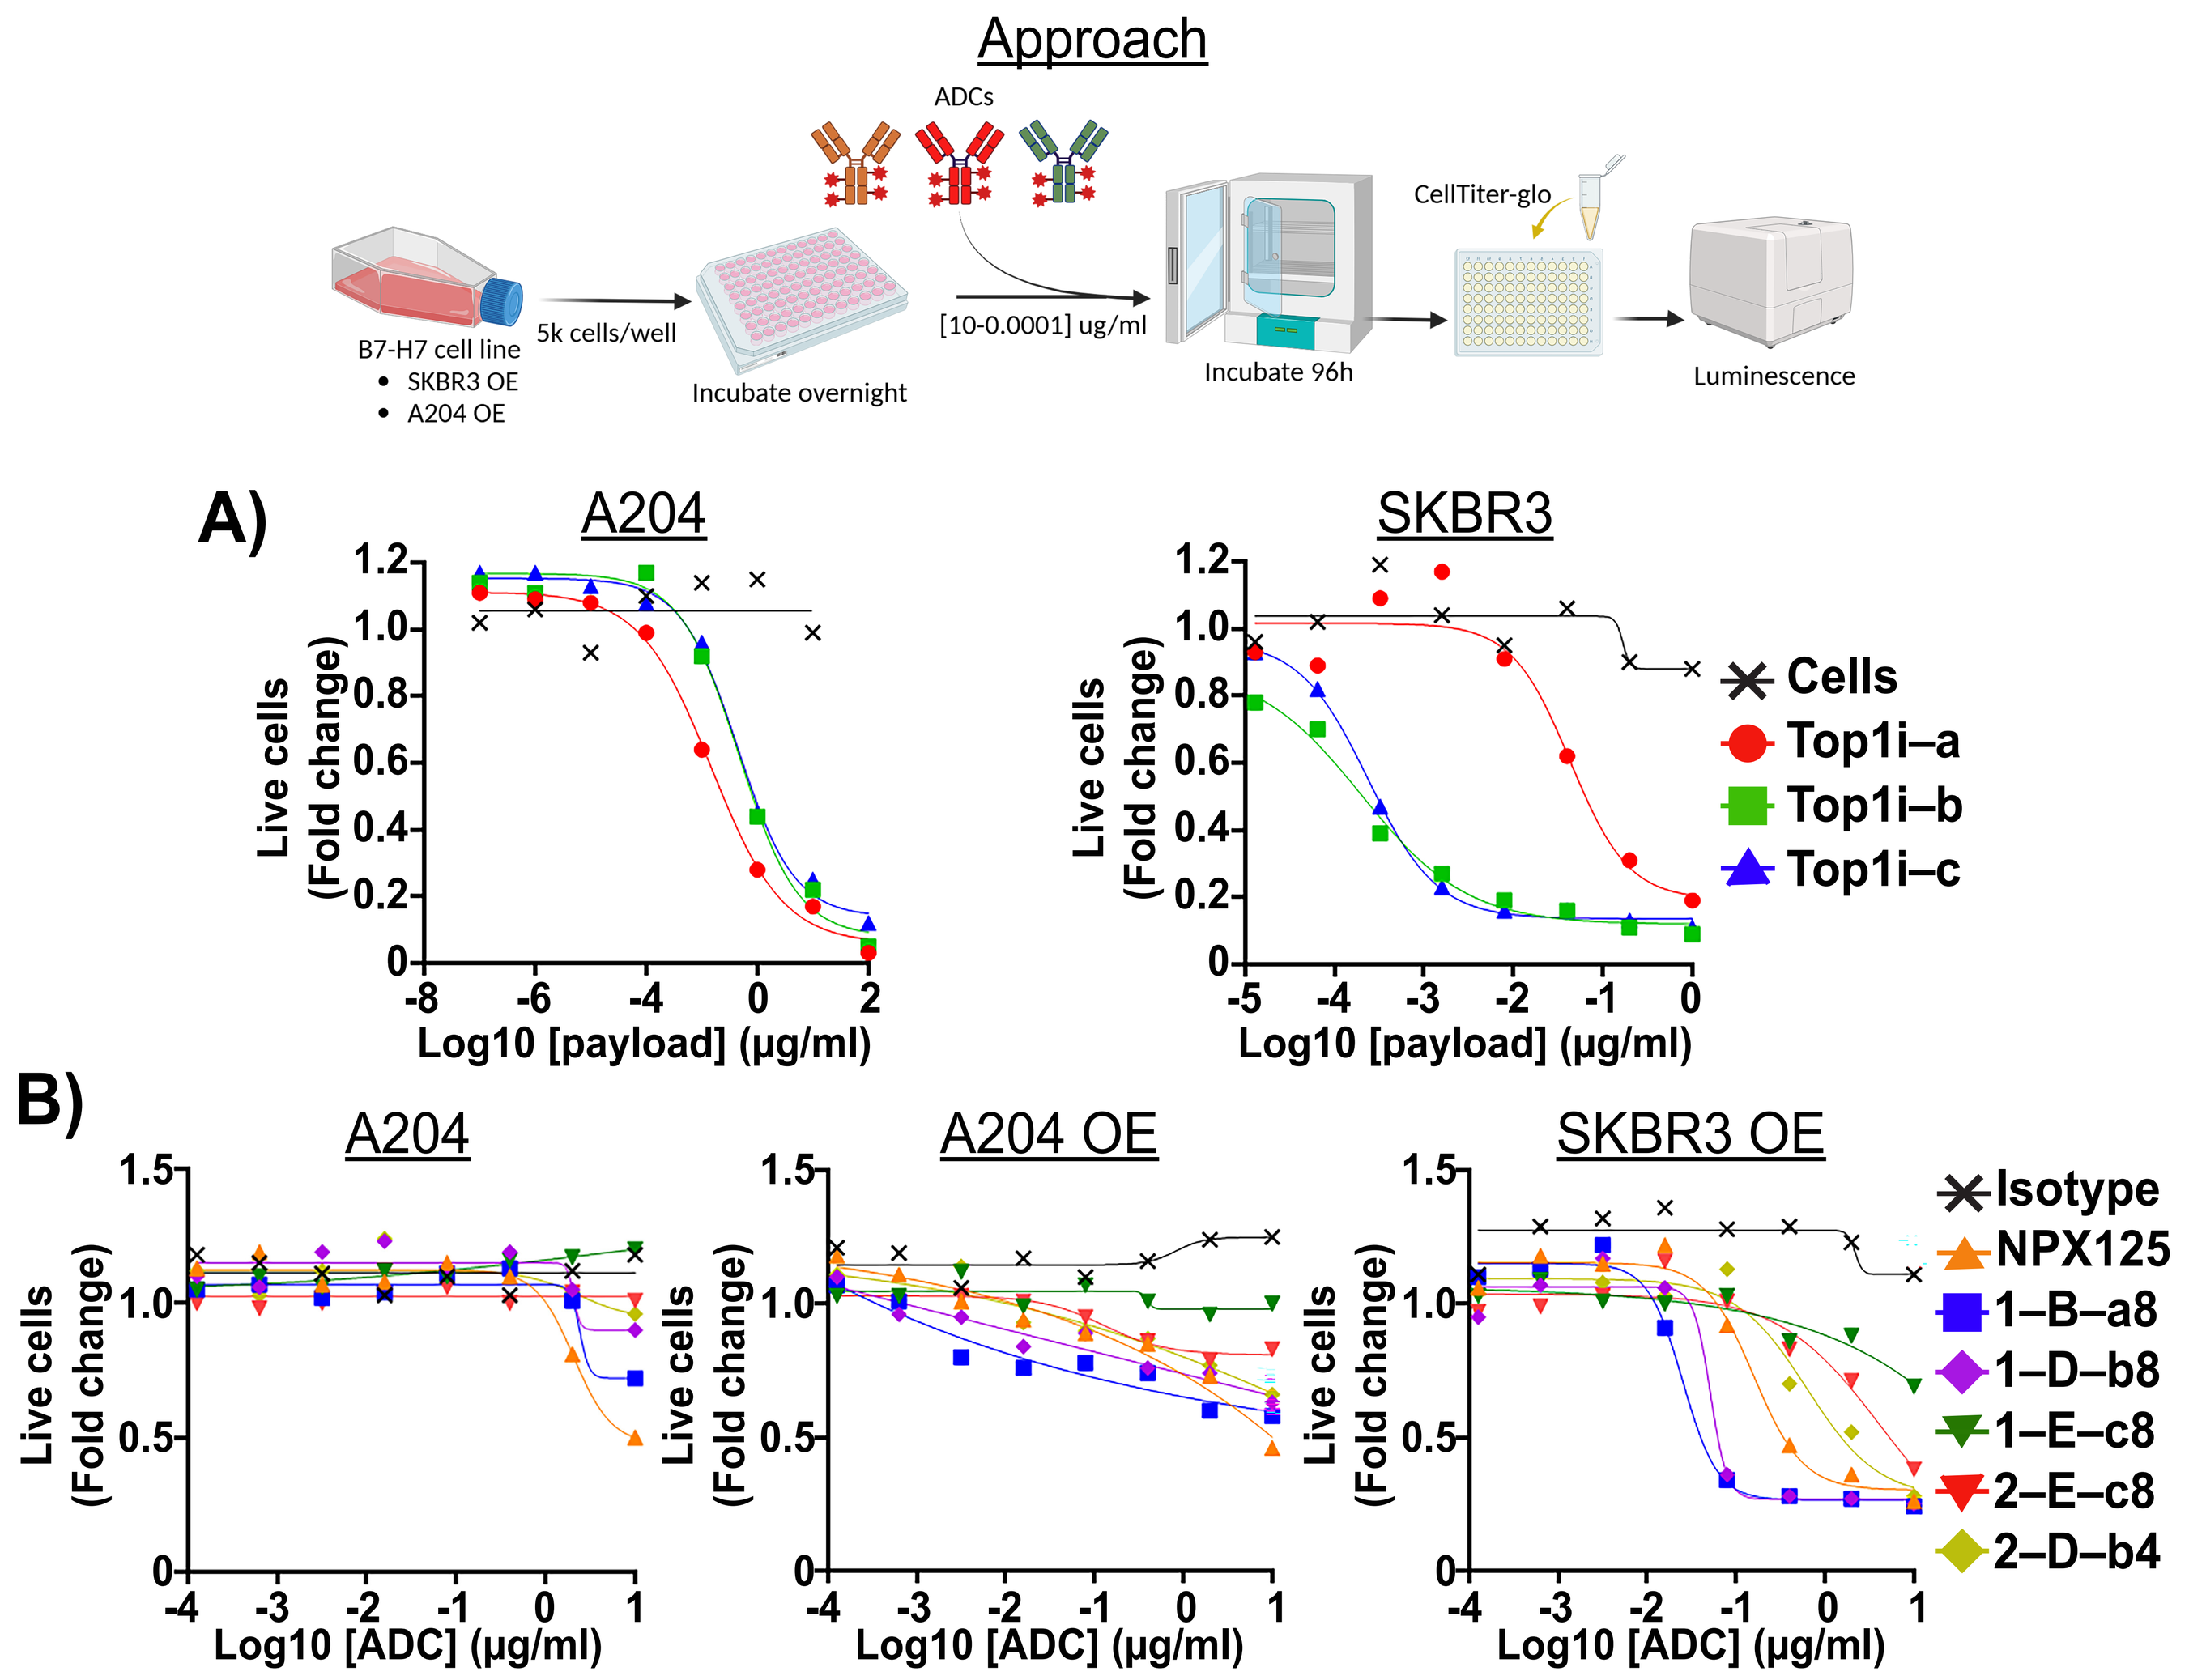
Approach
ADCs
B7-H7 cell line
SKBR3 OE
A204 OE
5k cells/well
Incubate overnight
[10-0.0001] ug/ml
Incubate 96h
CellTiter-glo
Luminescence
A)
A204
SKBR3
Live cells (Fold change)
Log10 [payload] (µg/ml)
Cells
Top1i–a
Top1i–b
Top1i–c
B)
A204
A204 OE
SKBR3 OE
Log10 [ADC] (µg/ml)
Isotype
NPX125
1–B–a8
1–D–b8
1–E–c8
2–E–c8
2–D–b4

B7-H7 expressing cells are sensitive to B7-H7 targeted ADCs, including NPX125 and other ADC formats. (A) Parental A204 and SKBR3 sensitivity to different Topoisomerase 1 inhibitors (free payload). **(B)** ADC-mediated cytotoxicity in A204, A204 OE and SKBR3 OE cells. Cytotoxicity is determined by the Cell-Titer-Glo assay and is presented as fold change in treated cells compared to untreated controls.

NPX125 Induces Bystander Cytotoxicity

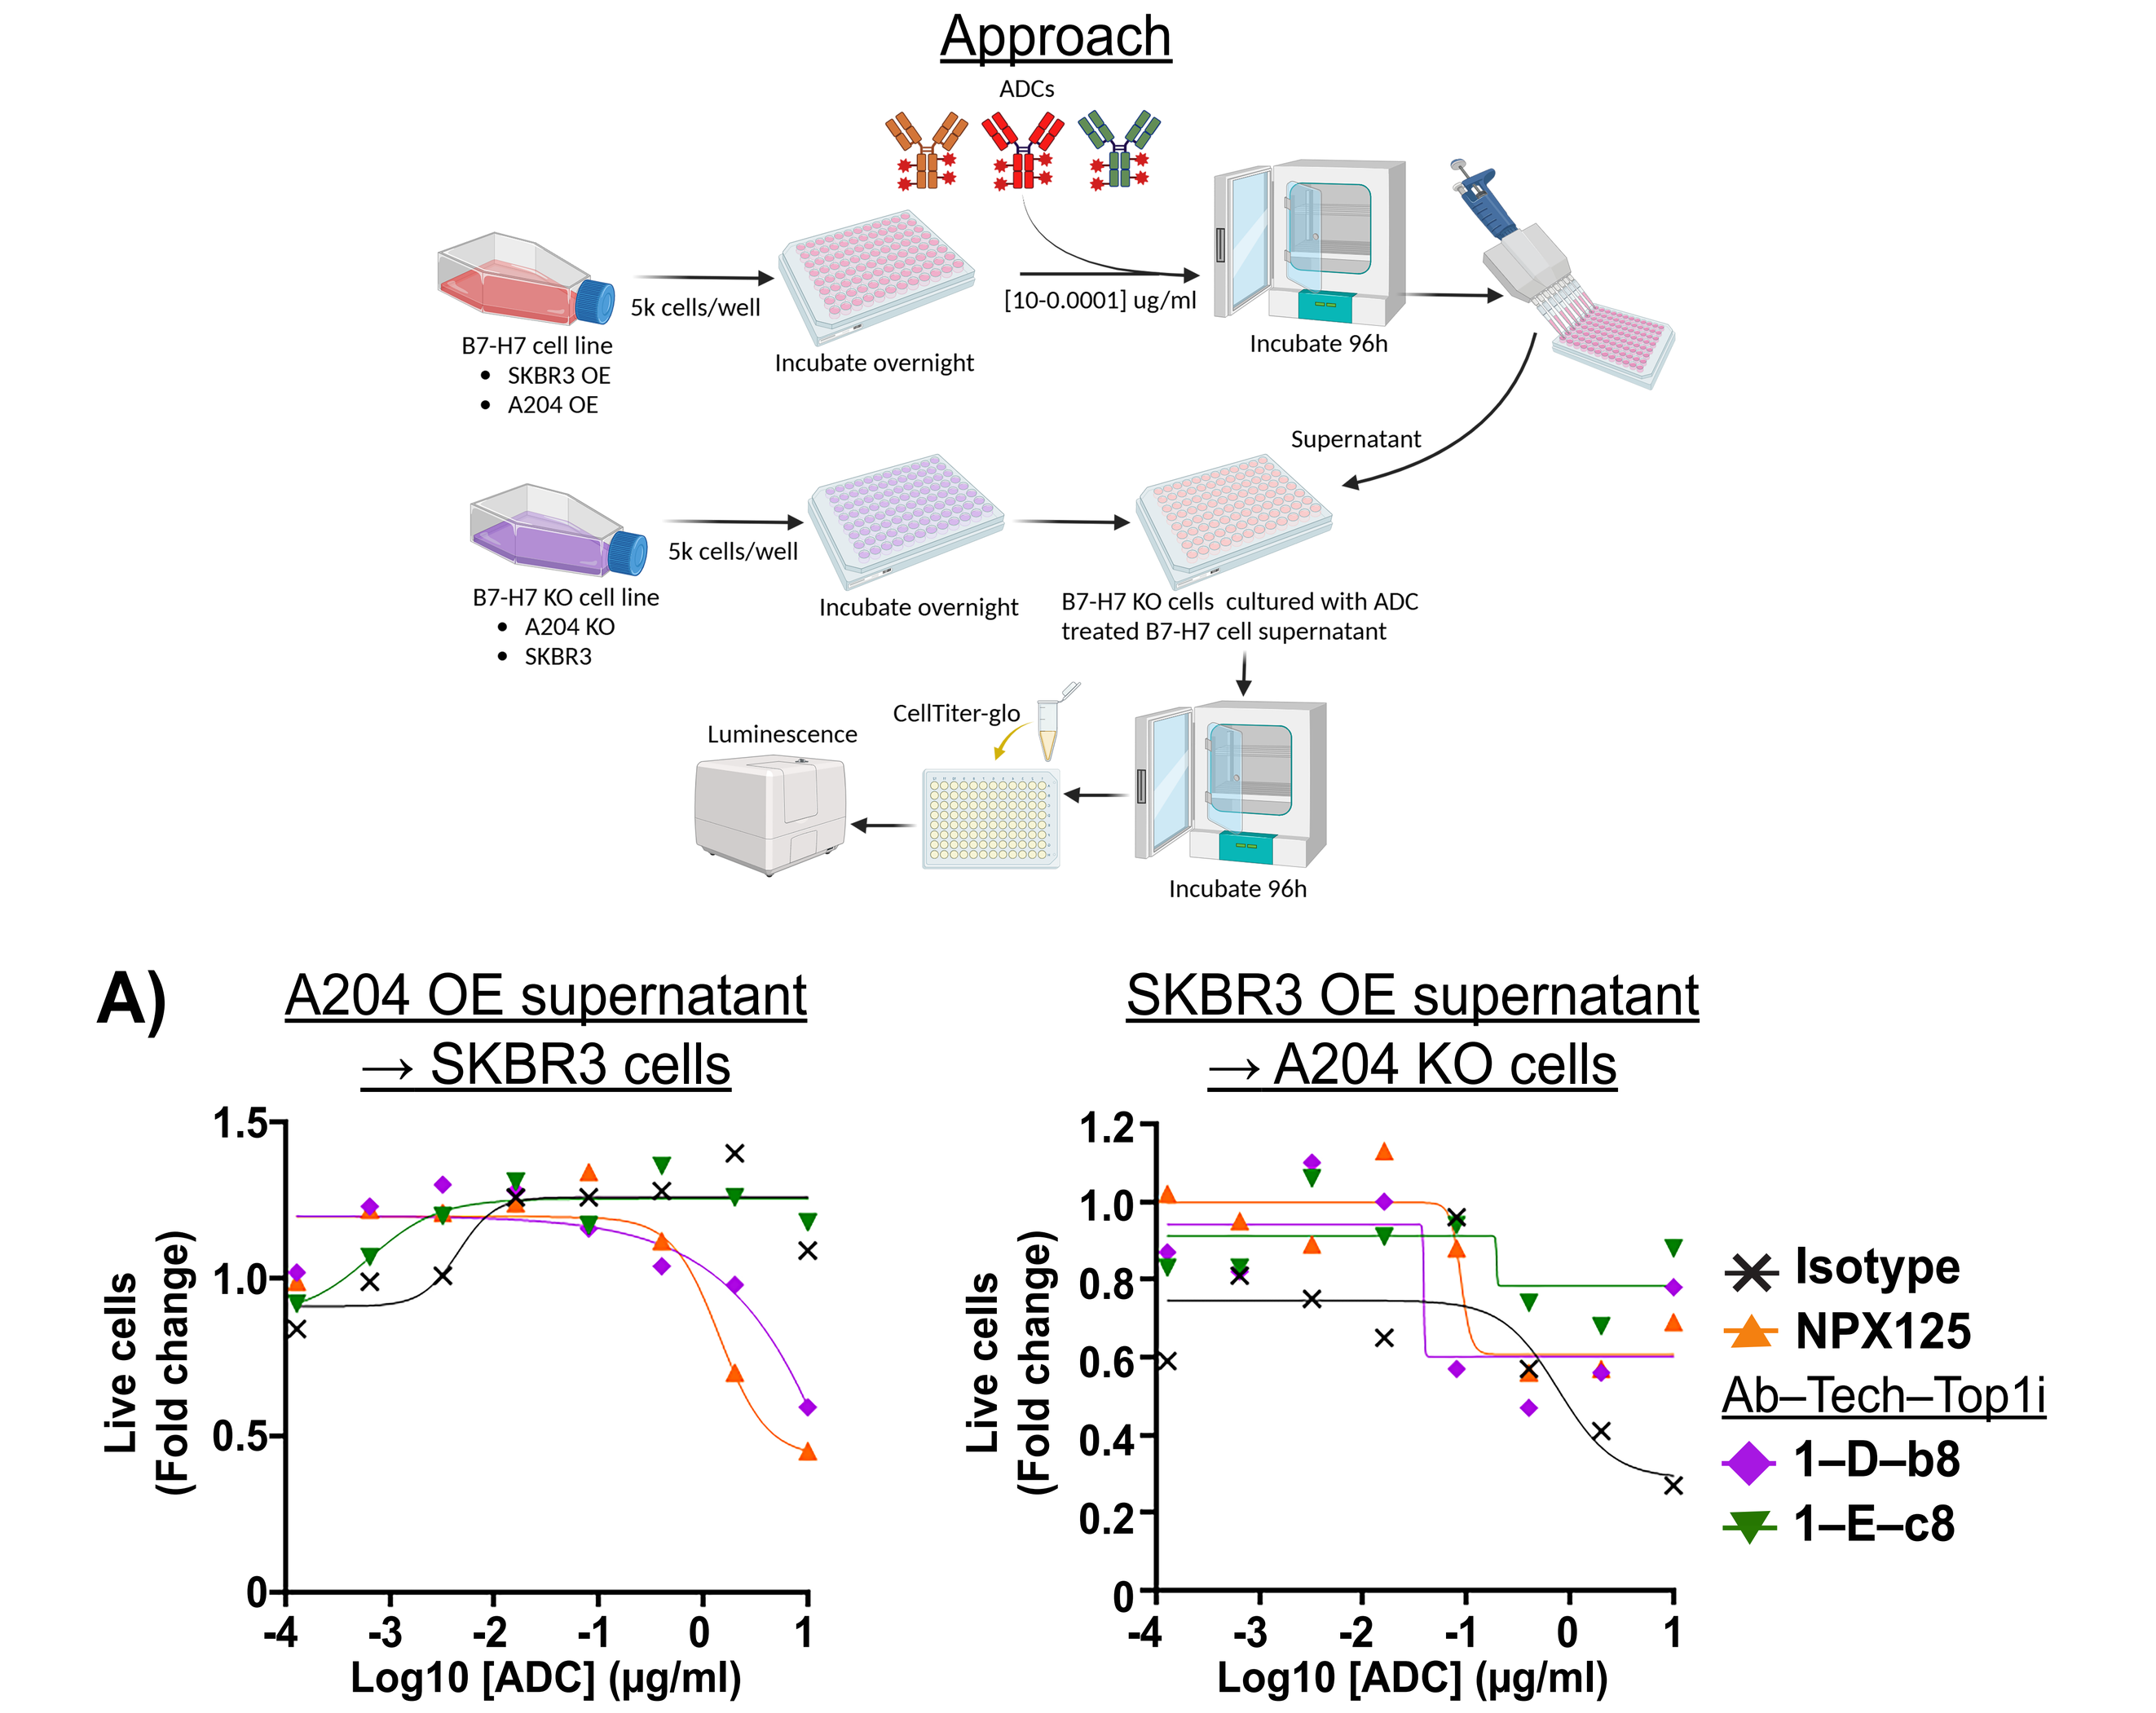
Approach
ADCs
B7-H7 cell line
SKBR3 OE
A204 OE
5k cells/well
Incubate overnight
[10-0.0001] ug/ml
Incubate 96h
Supernatant
B7-H7 KO cell line
A204 KO
SKBR3
B7-H7 KO cells cultured with ADC treated B7-H7 cell supernatant
Luminescence
CellTiter-glo
A)
A204 OE supernatant → SKBR3 cells
SKBR3 OE supernatant → A204 KO cells
Live cells (Fold change)
Log10 [ADC] (µg/ml)
Isotype
NPX125
Ab–Tech–Top1i
1–D–b8
1–E–c8

Bystander cytotoxicity is induced by culturing B7-H7–negative cell lines with the supernatant fromB7-H7–positive cells treated with NPX125, as compared to other ADC formats. (A) A204 B7-H7 knockout (KO) cells were treated with the supernatant from SKBR3 OE cells. **(B)** SKBR3 cells (which do not express B7-H7) were treated with the supernatant from A204 OE cells, to avoid residual ADC internalization. Data are presented as fold change of treated cells compared to untreated controls.

Rat PK Confirms NPX125 Serum Stability

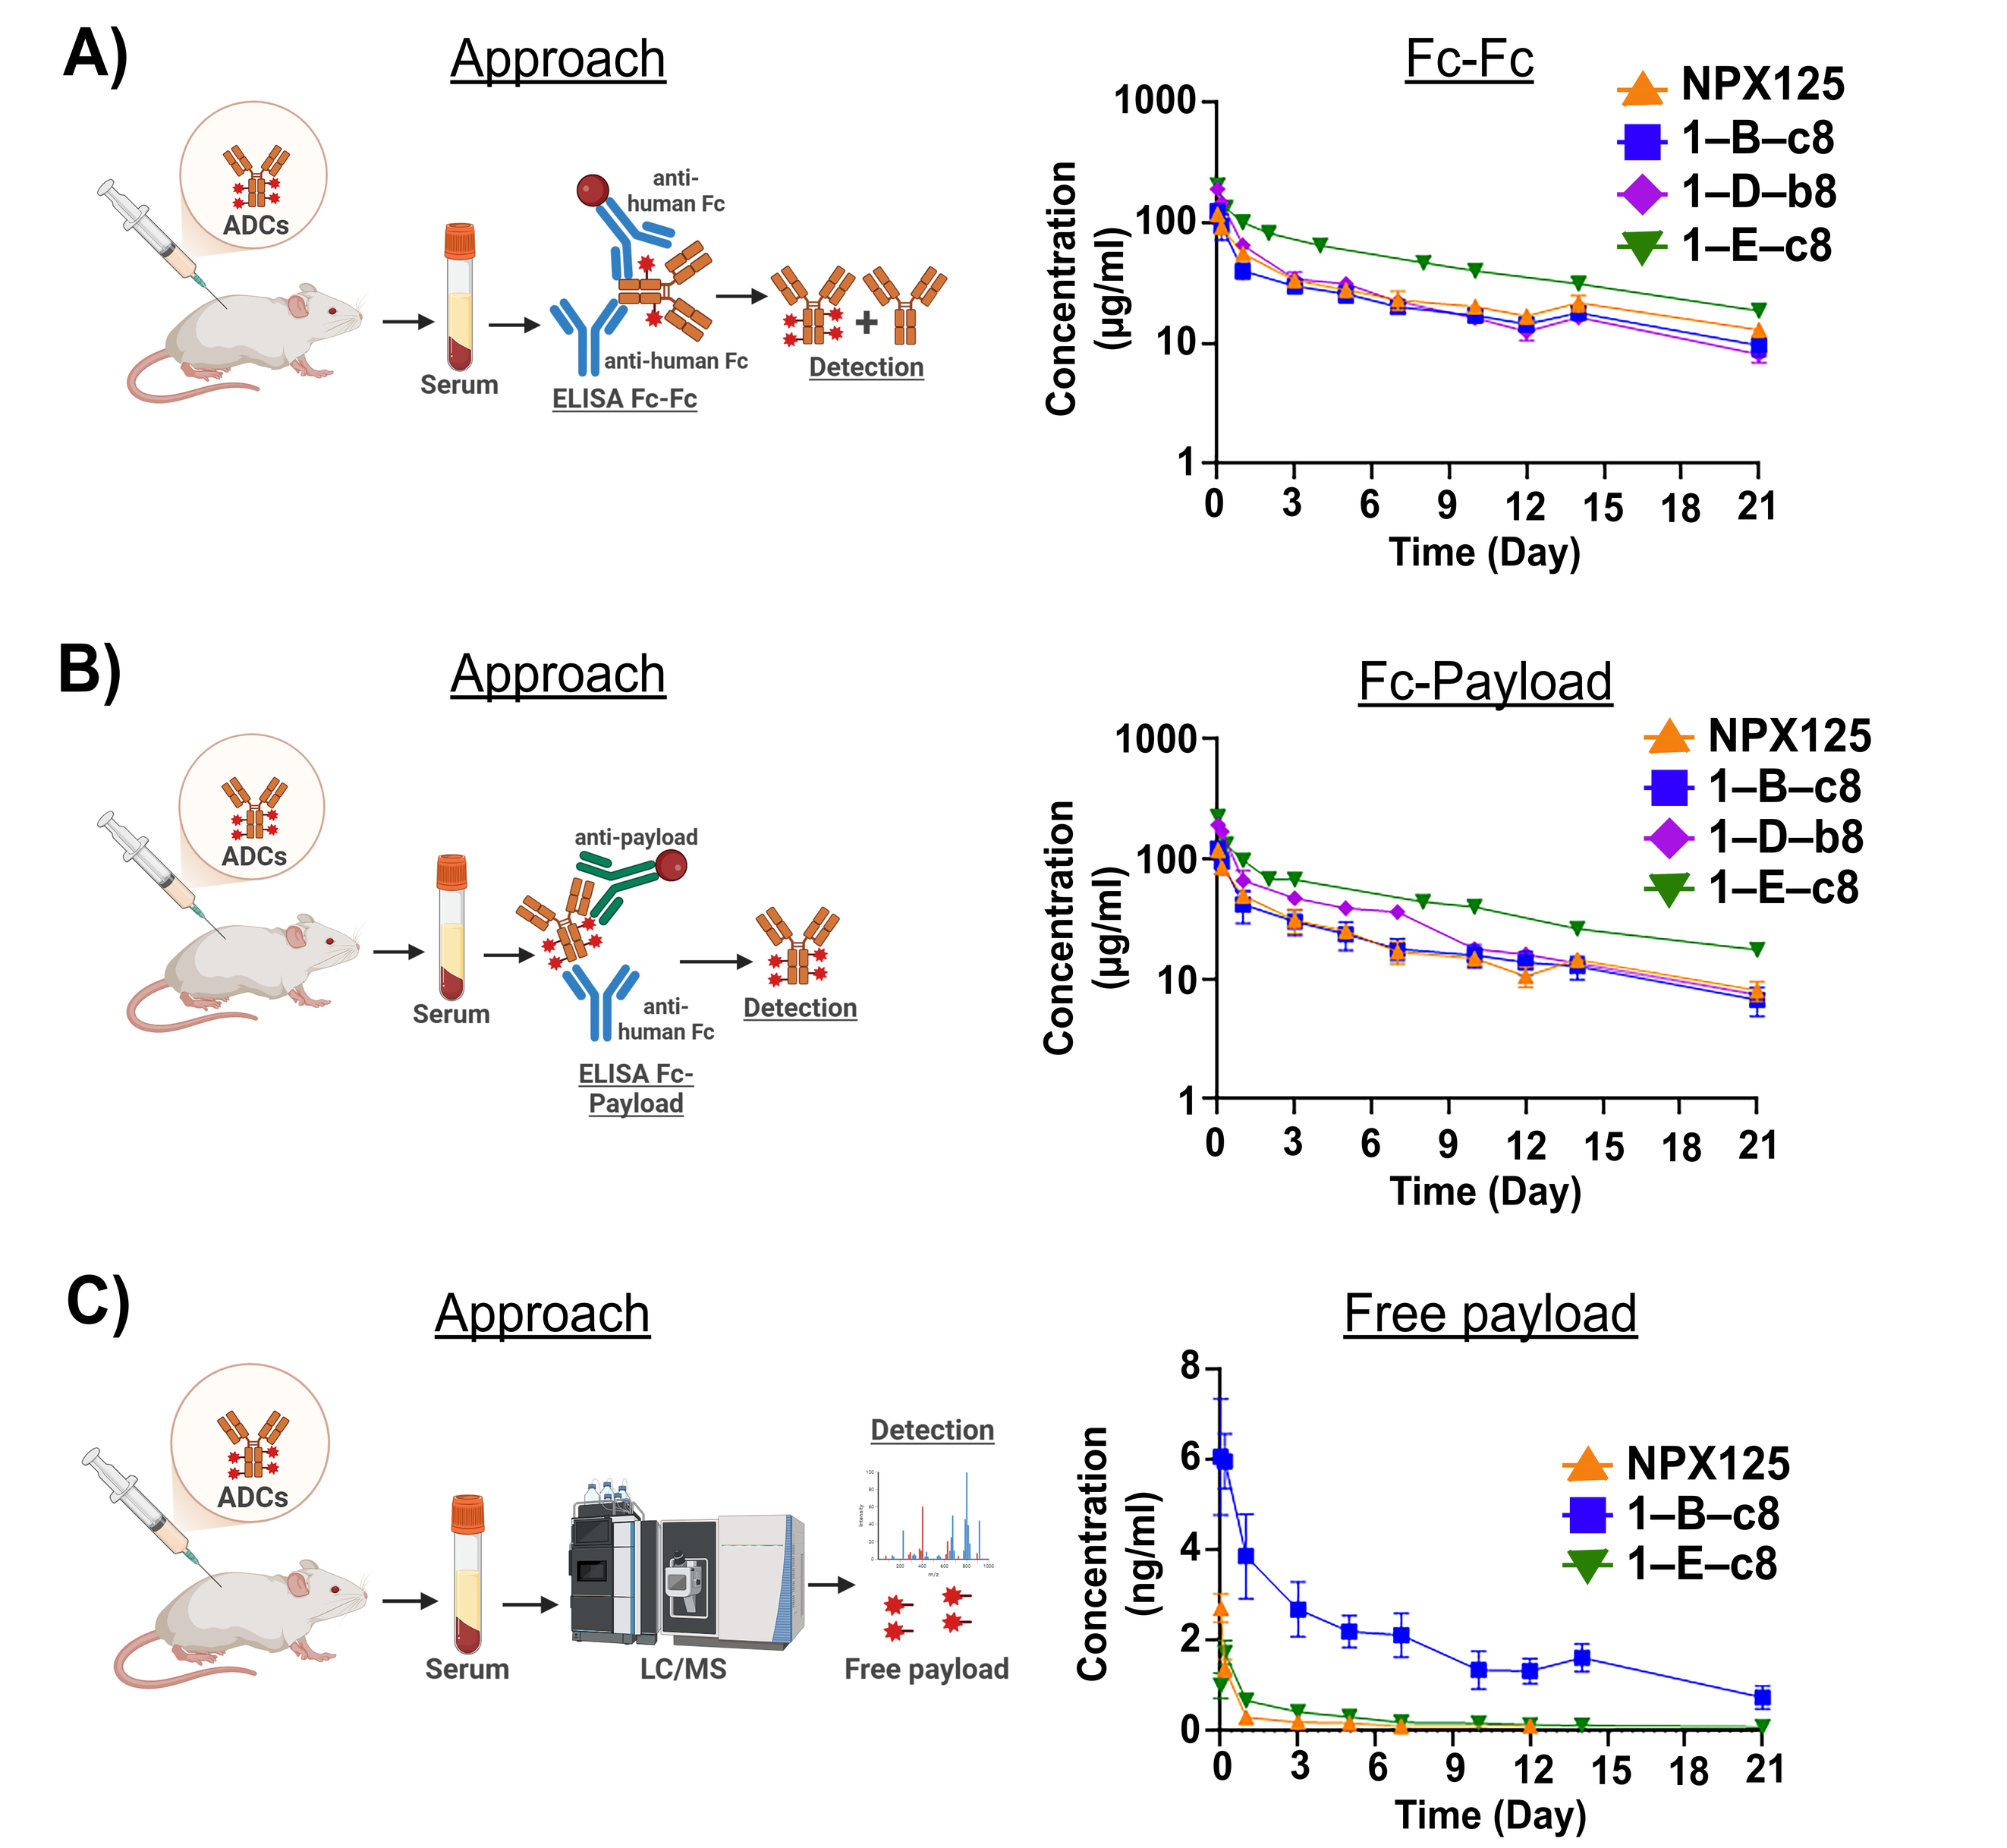
A)
Approach
ADCs
Serum
anti-human Fc
ELISA Fc-Fc
Detection
Fc-Fc
NPX125
1–B–c8
1–D–b8
1–E–c8
Concentration (µg/ml)
Time (Day)
B)
Approach
anti-payload
ELISA Fc-Payload
Fc-Payload
C)
Approach
LC/MS
Free payload
Free payload
Concentration (ng/ml)

Rat PK studies showed that NPX125 has high stability in vivo compared to other B7-H7 targeted ADCs tested. (A) Concentration curve of human IgG from ADC treatment over time in rat serum. **(B)** Concentration curve of ADC over time in rat serum. **(C)** Concentration curve of free payload from ADC treatment over time in rat serum.

Strong anti-tumor efficacy with NPX125 in multiple models

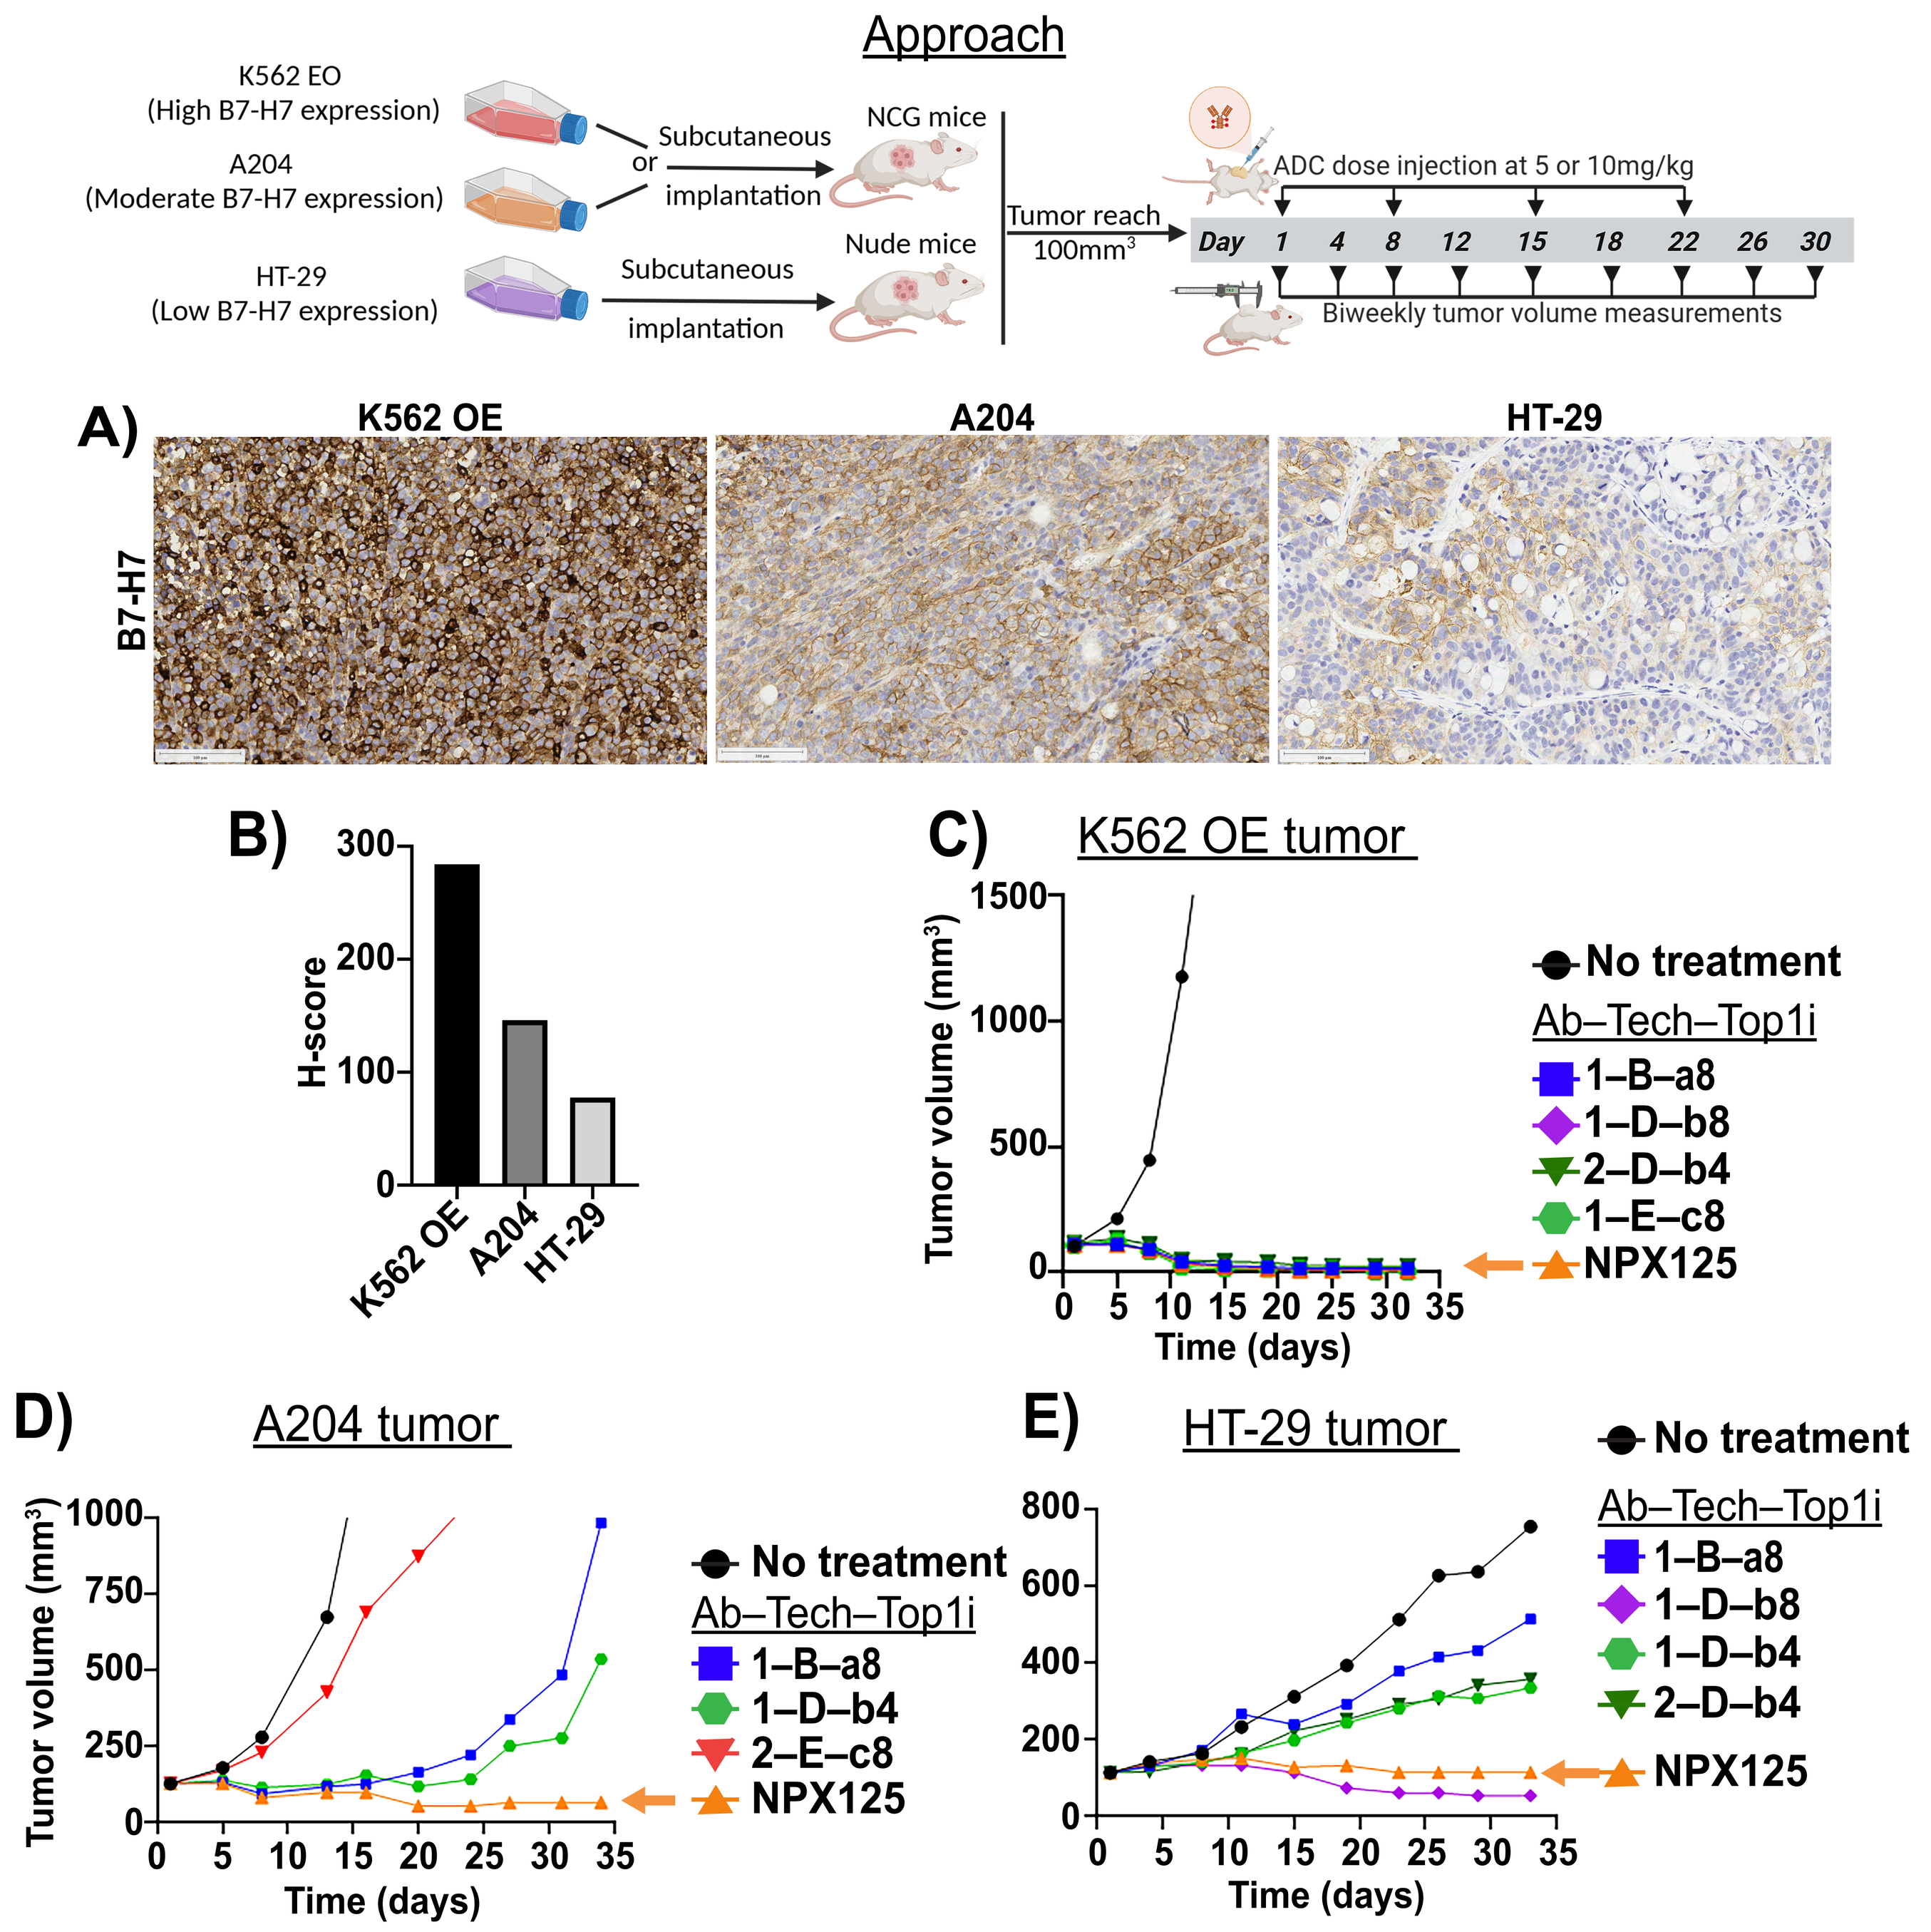
Approach
K562 EO (High B7-H7 expression)
A204 (Moderate B7-H7 expression)
HT-29 (Low B7-H7 expression)
Subcutaneous implantation
NCG mice
Nude mice
Tumor reach 100mm3
ADC dose injection at 5 or 10mg/kg
Day 1 4 8 12 15 18 22 26 30
Biweekly tumor volume measurements
A)
K562 OE
A204
HT-29
B7-H7
B)
H-score
C)
K562 OE tumor
D)
A204 tumor
E)
HT-29 tumor
Tumor volume (mm3)
Time (days)
No treatment
Ab–Tech–Top1i
1–B–a8
1–D–b8
2–D–b4
1–E–c8
1–D–b4
2–E–c8
NPX125

Significant tumor regressions are achieved with NPX125, across diverse tumor types and different B7-H7 expression levels. (A–B) B7-H7 expression in preclinical mouse tumor models by IHC. Representative tumor images showing B7-H7 expression in implanted human tumor cells (A) and quantification by H-score (B). **(C–E)** Tumor growth kinetics in murine models of human xenograft tumors with subcutaneous implantation, treated with four weekly doses of different ADCs. Data represent mean ± SD; n=6. K562 OE treated at 10 mg/kg (C), A204 treated at 5 mg/kg (D) and HT-29 treated at 10 mg/kg (E).

Conclusions

NPX125 is the lead candidate of B7-H7 targeting ADCs tested and demonstrated:

- Similar on-cell binding to the parental antibody and superior to other ADC formats
- Similar internalization kinetics to its parental antibody, and superior to a clinically-relevant ADC
- Higher potency (both direct and bystander cytotoxic activity) than other ADC formats tested, and across various tumor cell lines
- Similar or higher *in vivo* stability compared to other constructs
- Potent anti-tumor activity in multiple preclinical mouse models, including against tumors with lower target antigen expression (B7-H7)

Collectively, these preclinical data establish B7-H7 as a promising ADC target and demonstrate its activity across multiple clinically relevant payload-linker technologies. These findings support further development of lead compounds as potential treatments for patients with B7-H7–expressing tumors.

nextpoint

nextpoint

AACR 2025

#1556

B7-H7-CD3 Bispecific T cell Engaging Antibodies Demonstrate Potent Anti-Tumor Activity in B7-H7+ Preclinical Tumor Models

Karishma Vekaria[1], Susannah Hewitt[1], Melissa W. Hayes[1], Emilien Loeuillard[1], Jamie Strand[1], Riale Gilligan[1], Maja Razlog[1], Emma Aeschliman[1], Quangvinh Tran[1], McKenna Lamb[1], Bijan Etemad-Gilbertson[1], Silvia Ferrati[1], Tatiana Novobrantseva[1], and Matthew Rausch[1]. [1]NextPoint Therapeutics, Cambridge, MA

B7-H7 is a Novel Tumor Antigen that Modulates an Immune Checkpoint Axis

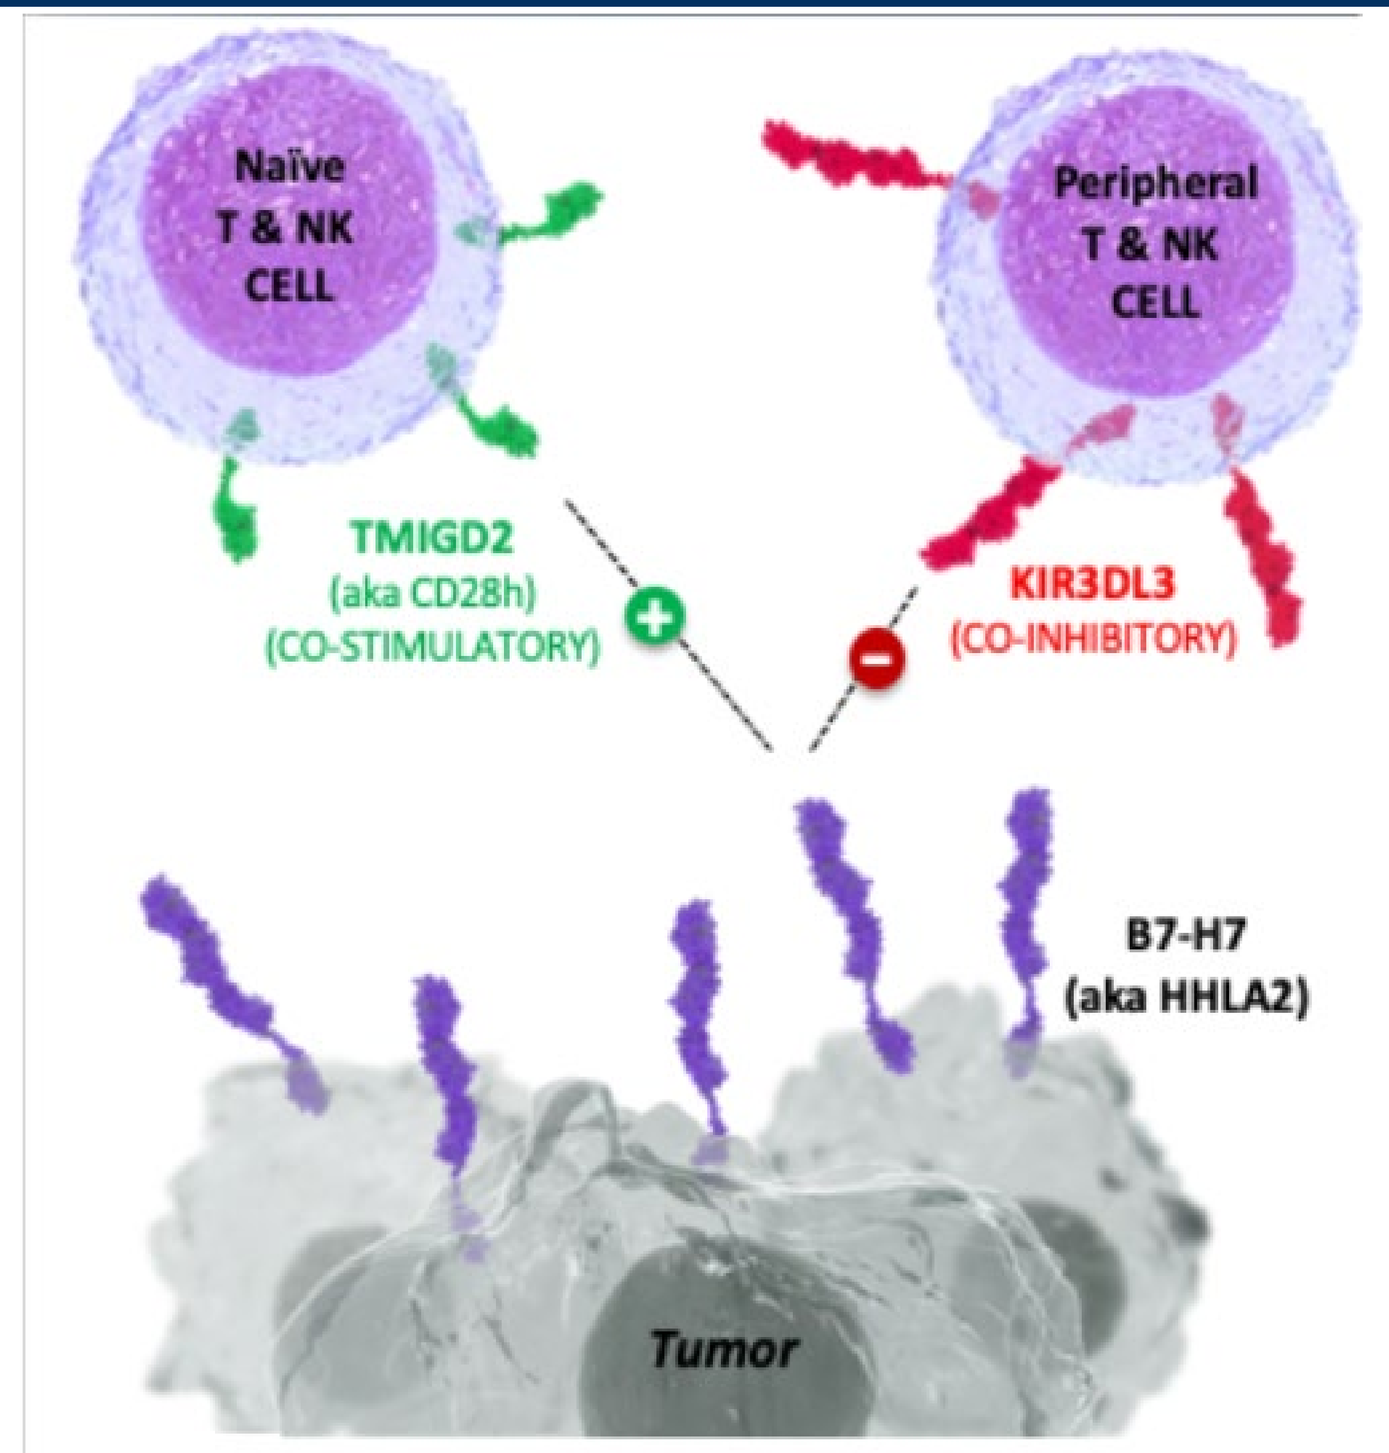
Naïve T & NK CELL
Peripheral T & NK CELL
TMIGD2 (aka CD28h) (CO-STIMULATORY)
KIR3DL3 (CO-INHIBITORY)
B7-H7 (aka HHLA2)
Tumor

Background: B7-H7 is a B7 family member that suppresses T and NK cell activation through an inhibitory receptor, KIR3DL3, and stimulates immune cell activation through a distinct receptor, TMIGD2. B7-H7 is upregulated on tumor cells of many histologic subtypes and is, therefore, explored as a novel tumor targeting antigen. NextPoint has developed two B7-H7-targeted CD3 bispecific antibodies (bsAb), NPX372 and NPX387, to redirect T cell-mediated immunity toward B7-H7+ tumors. Both antibodies bind B7-H7 with similar avidity and block interaction with KIR3DL3, but induce the formation of distinct immune synapses.

B7-H7-CD3 Bispecific Antibody Characteristics

B7-H7-CD3 Bispecific Ab Features

- 2 + 1 KiH bispecific format
- Limited Fc effector function due to LALAPA mutations
- Both arms are cross-reactive to cynomolgus monkey
- Multiple B7-H7 avidity-based epitopes evaluated

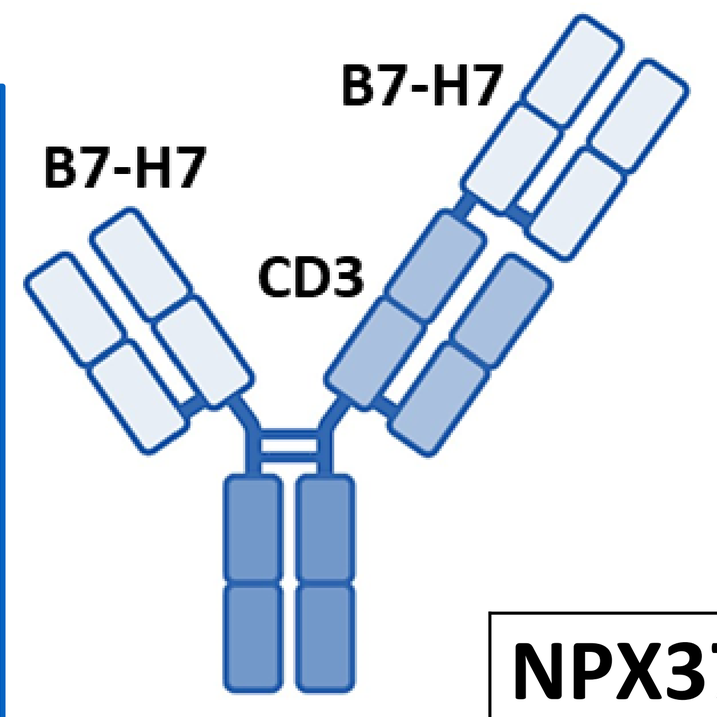
B7-H7
B7-H7
CD3

	On-Cell Binding Affinity/Avidity	
	CD3 EC_{50}	B7-H7 EC_{50}
NPX372	8.8 nM	0.55 nM
NPX387	27.1 nM	1.1 nM

NPX372 and NPX387 Induce Primary CD8+ T Cell Activation After B7-H7 Crosslinking

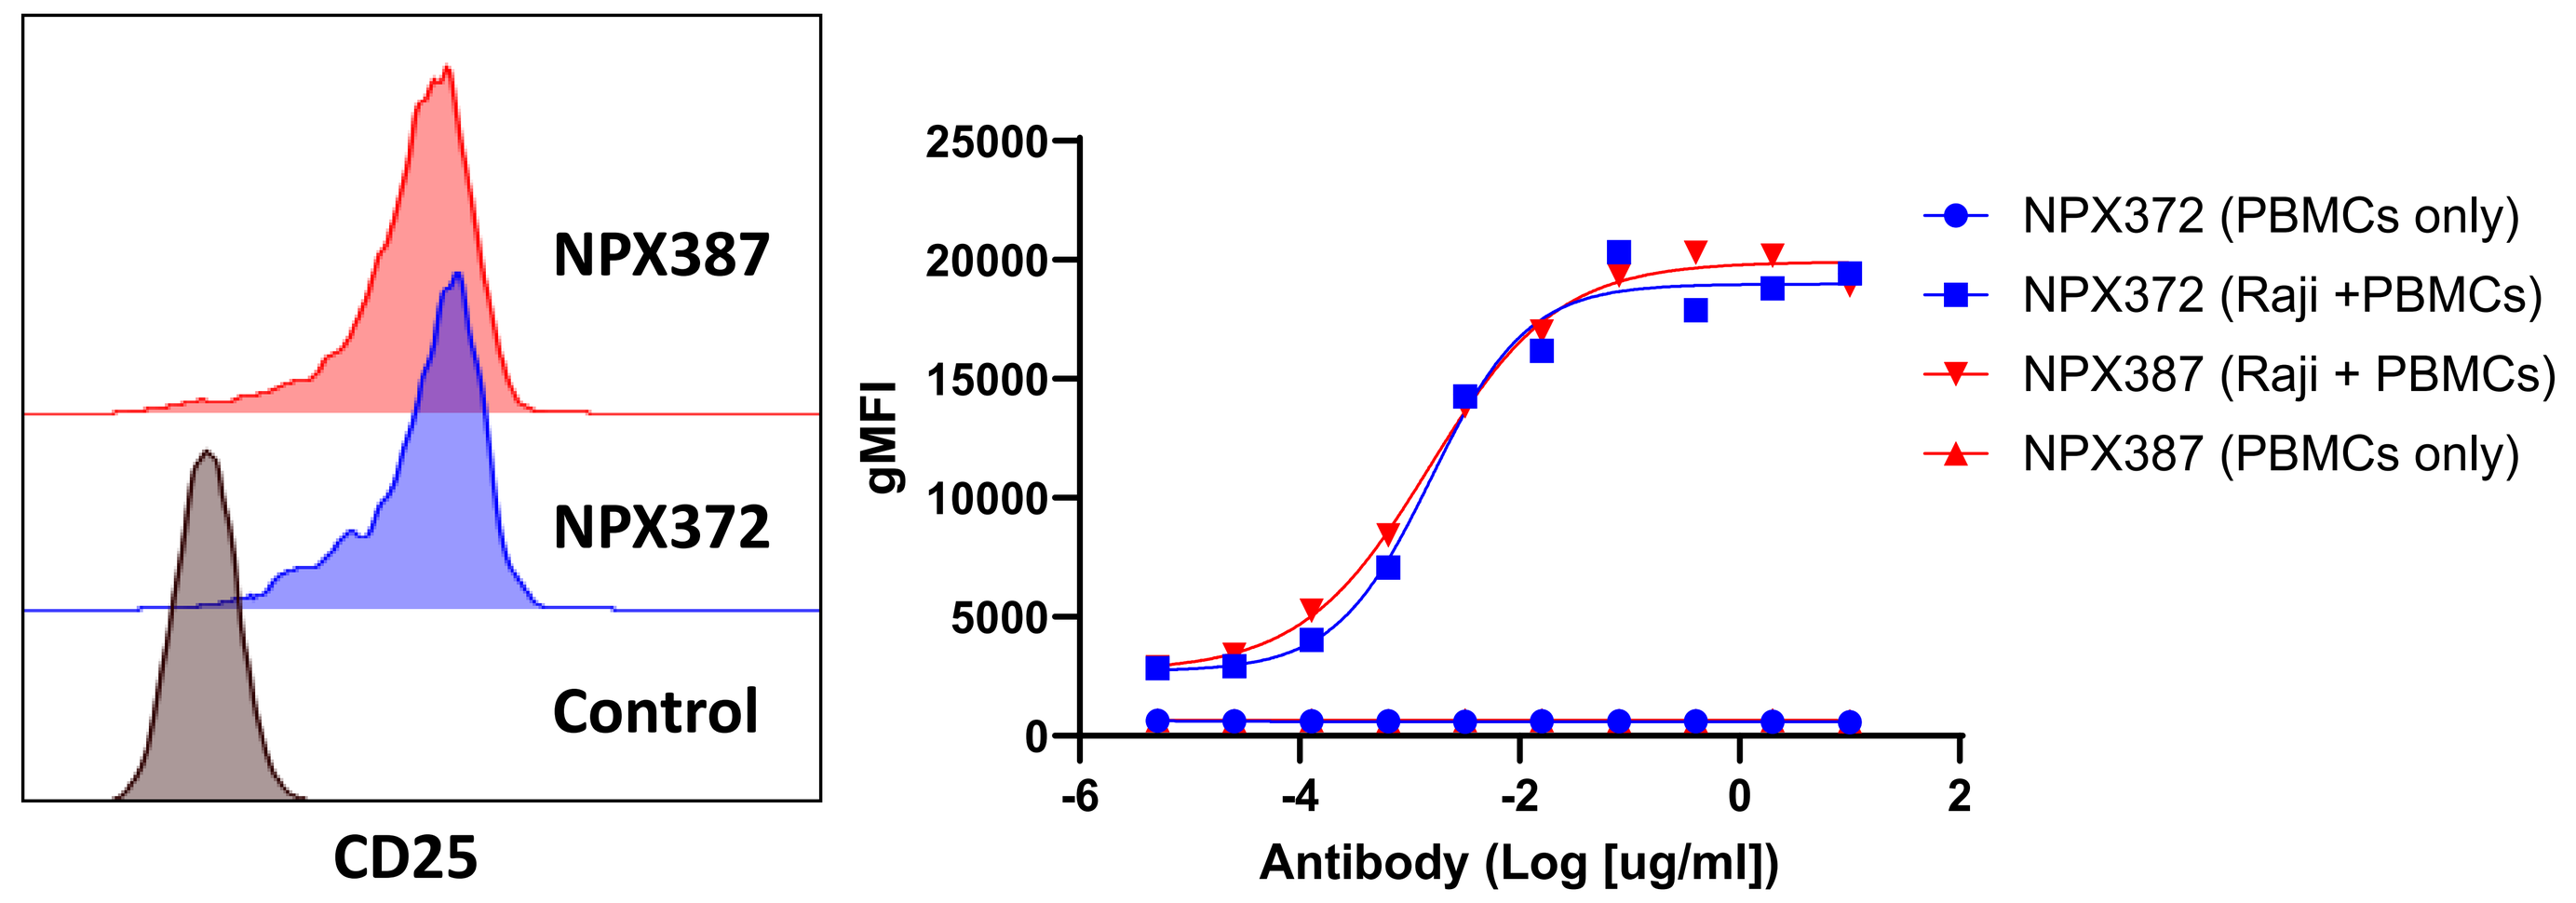
NPX387
NPX372
Control
CD25
gMFI
Antibody (Log [ug/ml])
NPX372 (PBMCs only)
NPX372 (Raji +PBMCs)
NPX387 (Raji + PBMCs)
NPX387 (PBMCs only)

Raji-B7-H7 cells were plated with serially diluted bsAbs for 30 minutes followed by addition of human PBMCs. After 24 hours, upregulation of CD25 was measured by flow cytometry on CD8+ T cells.

NPX372 and NPX387 Induce T Cell Redirected Killing of Multiple B7-H7+ Tumor Cell Lines

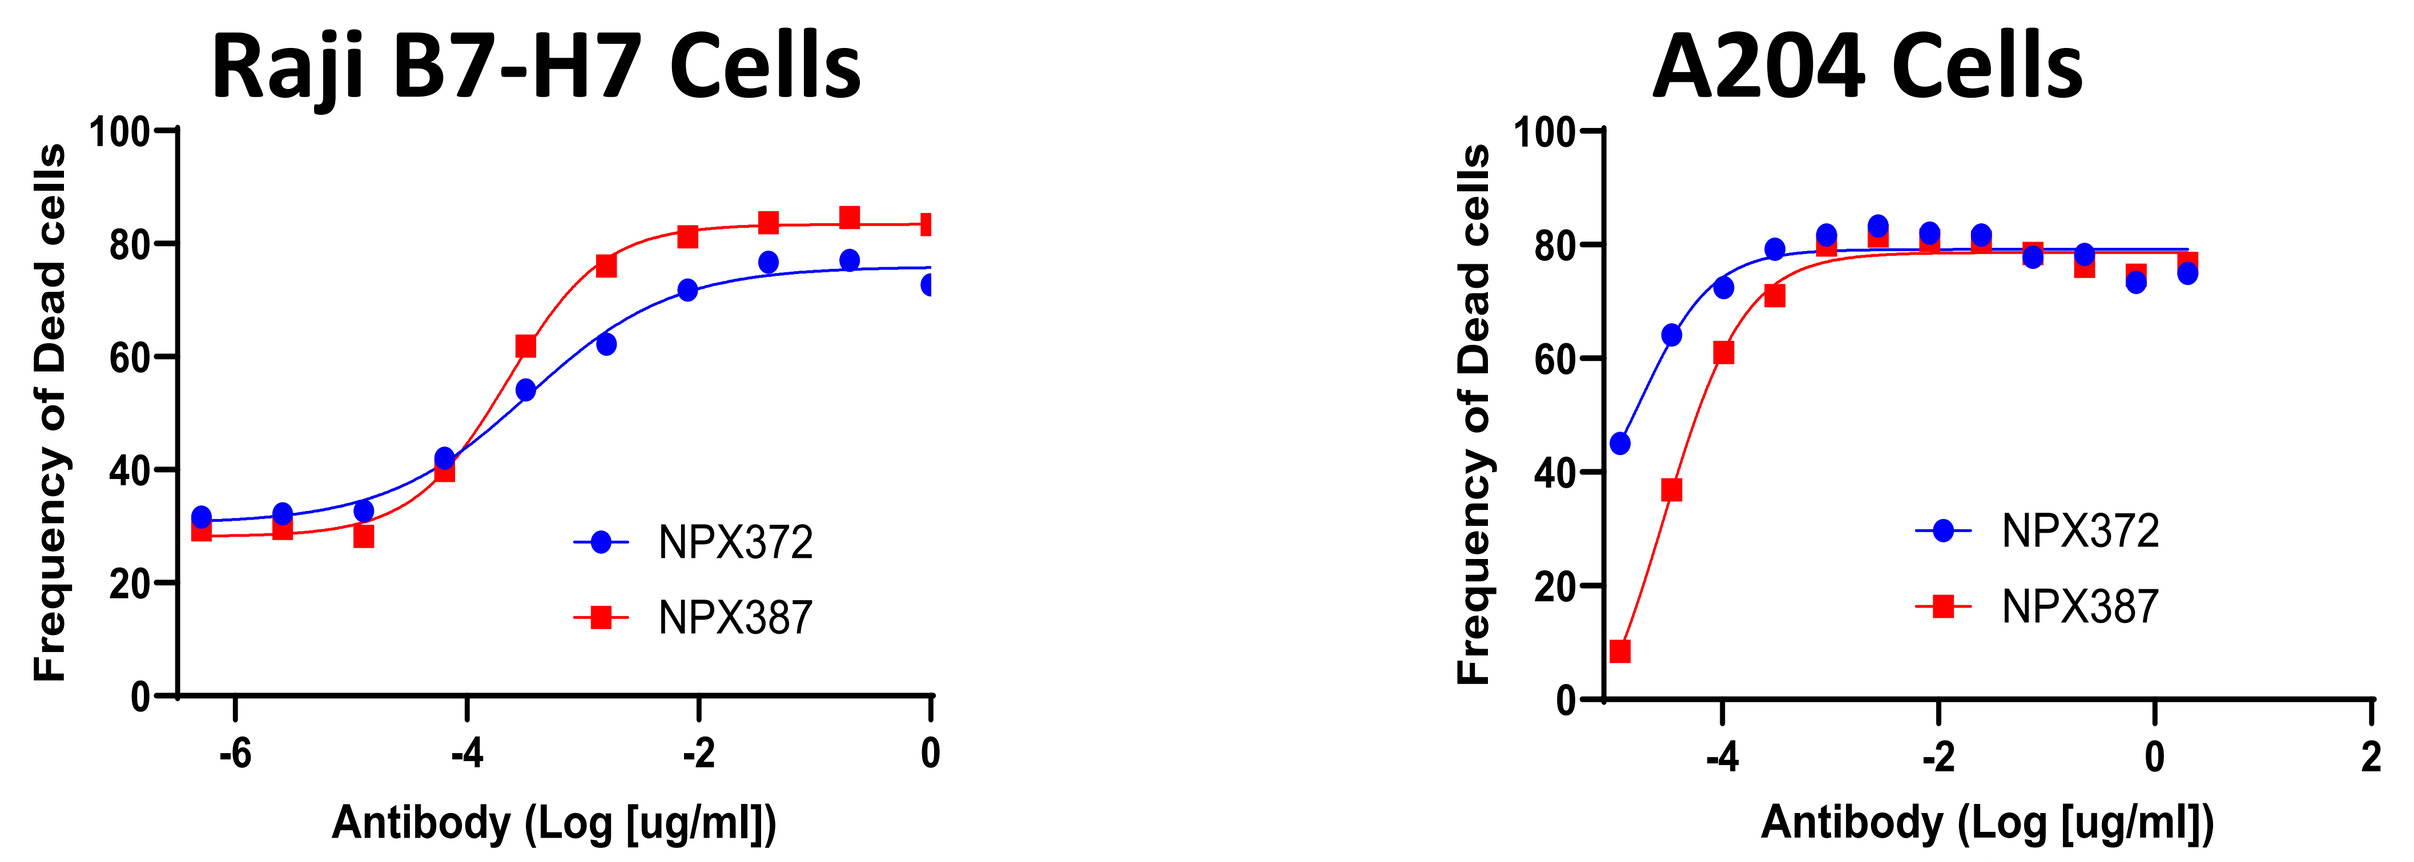
Raji B7-H7 Cells
A204 Cells
Frequency of Dead cells
NPX372
NPX387
Antibody (Log [ug/ml])

Raji B7-H7 and A204 tumor cells were stained with cell trace violet (CTV) and co-cultured with primary human PBMCs at an Effector to Target ratio of 5:1 in the presence of serially diluted bsAb. After 24 hours, the frequency of dead target cells (CTV+ viability dye+ cells) was measured by flow cytometry.

NPX372 and NPX387 Induce B7-H7 Dependent Cytokine Production in vitro

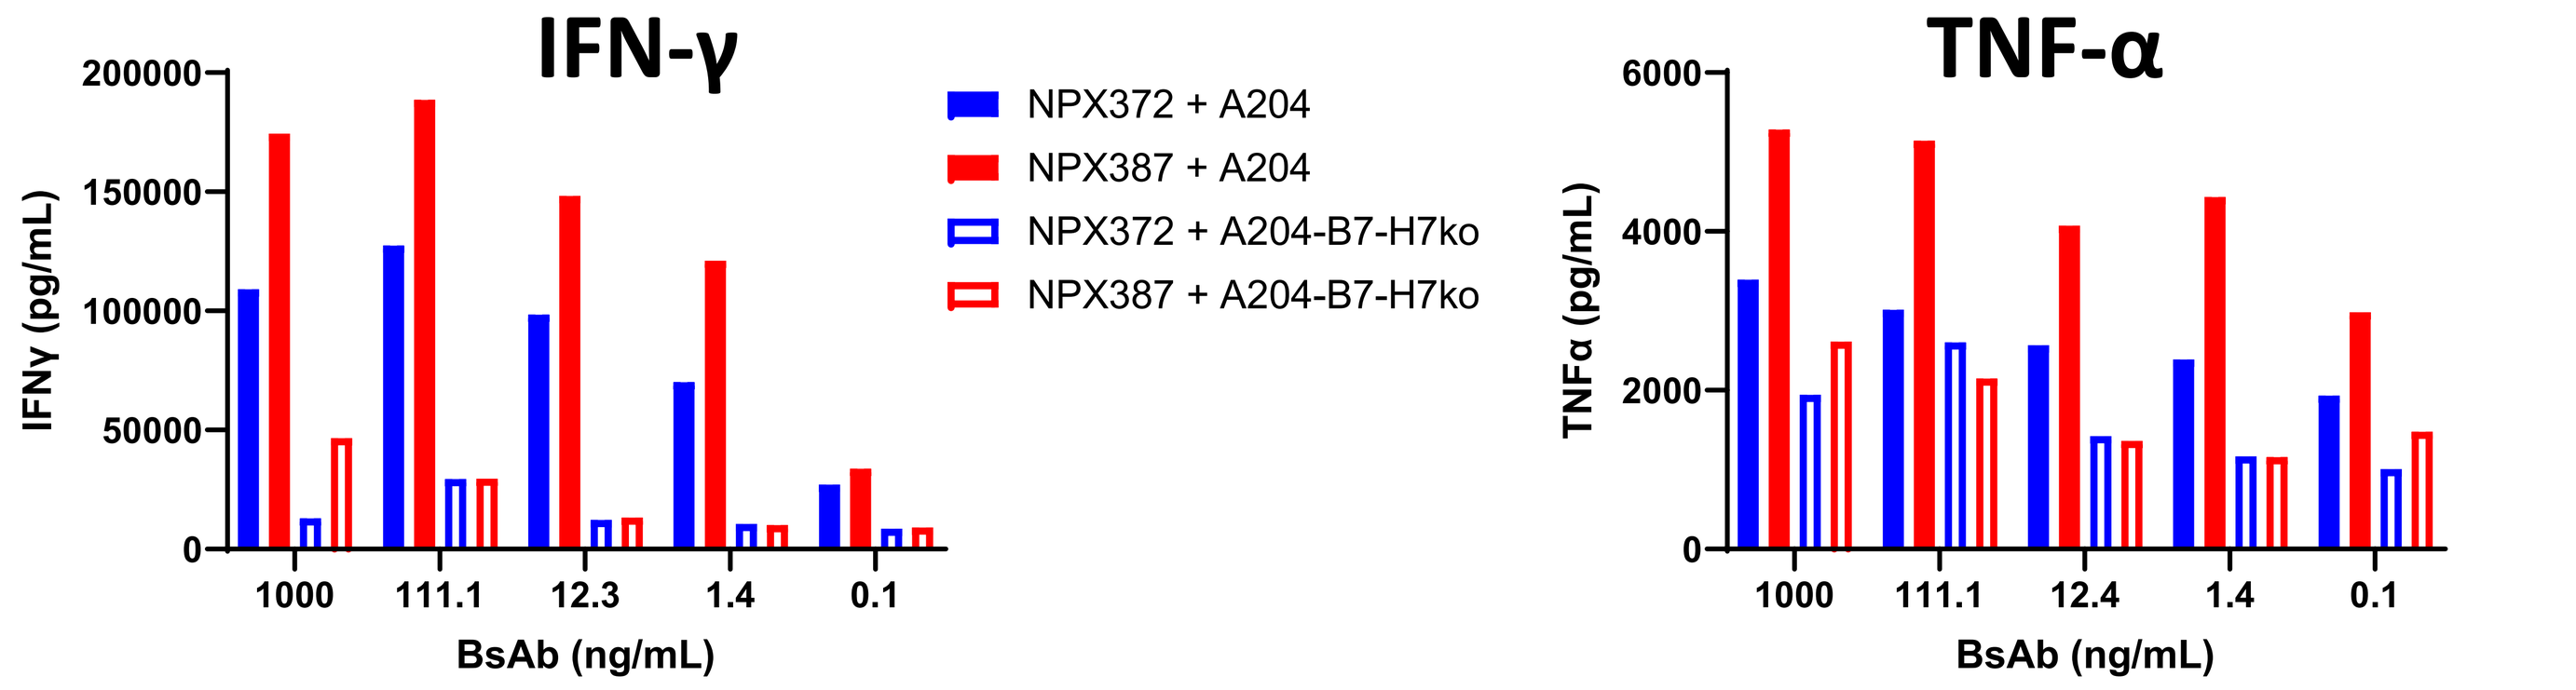
IFN-γ
TNF-α
IFNγ (pg/mL)
TNFα (pg/mL)
NPX372 + A204
NPX387 + A204
NPX372 + A204-B7-H7ko
NPX387 + A204-B7-H7ko
BsAb (ng/mL)

A204 and A204-B7-H7 deficient (A204-B7-H7ko) cells were plated with serially diluted bsAbs for 30 minutes followed by addition of human PBMCs. After 24 hours, supernatants were collected and cytokine levels were measured by MSD.

Treatment With NPX372 and NPX387 Induces A204 Tumor Regression in an Activated T Cell Humanized Mouse Model

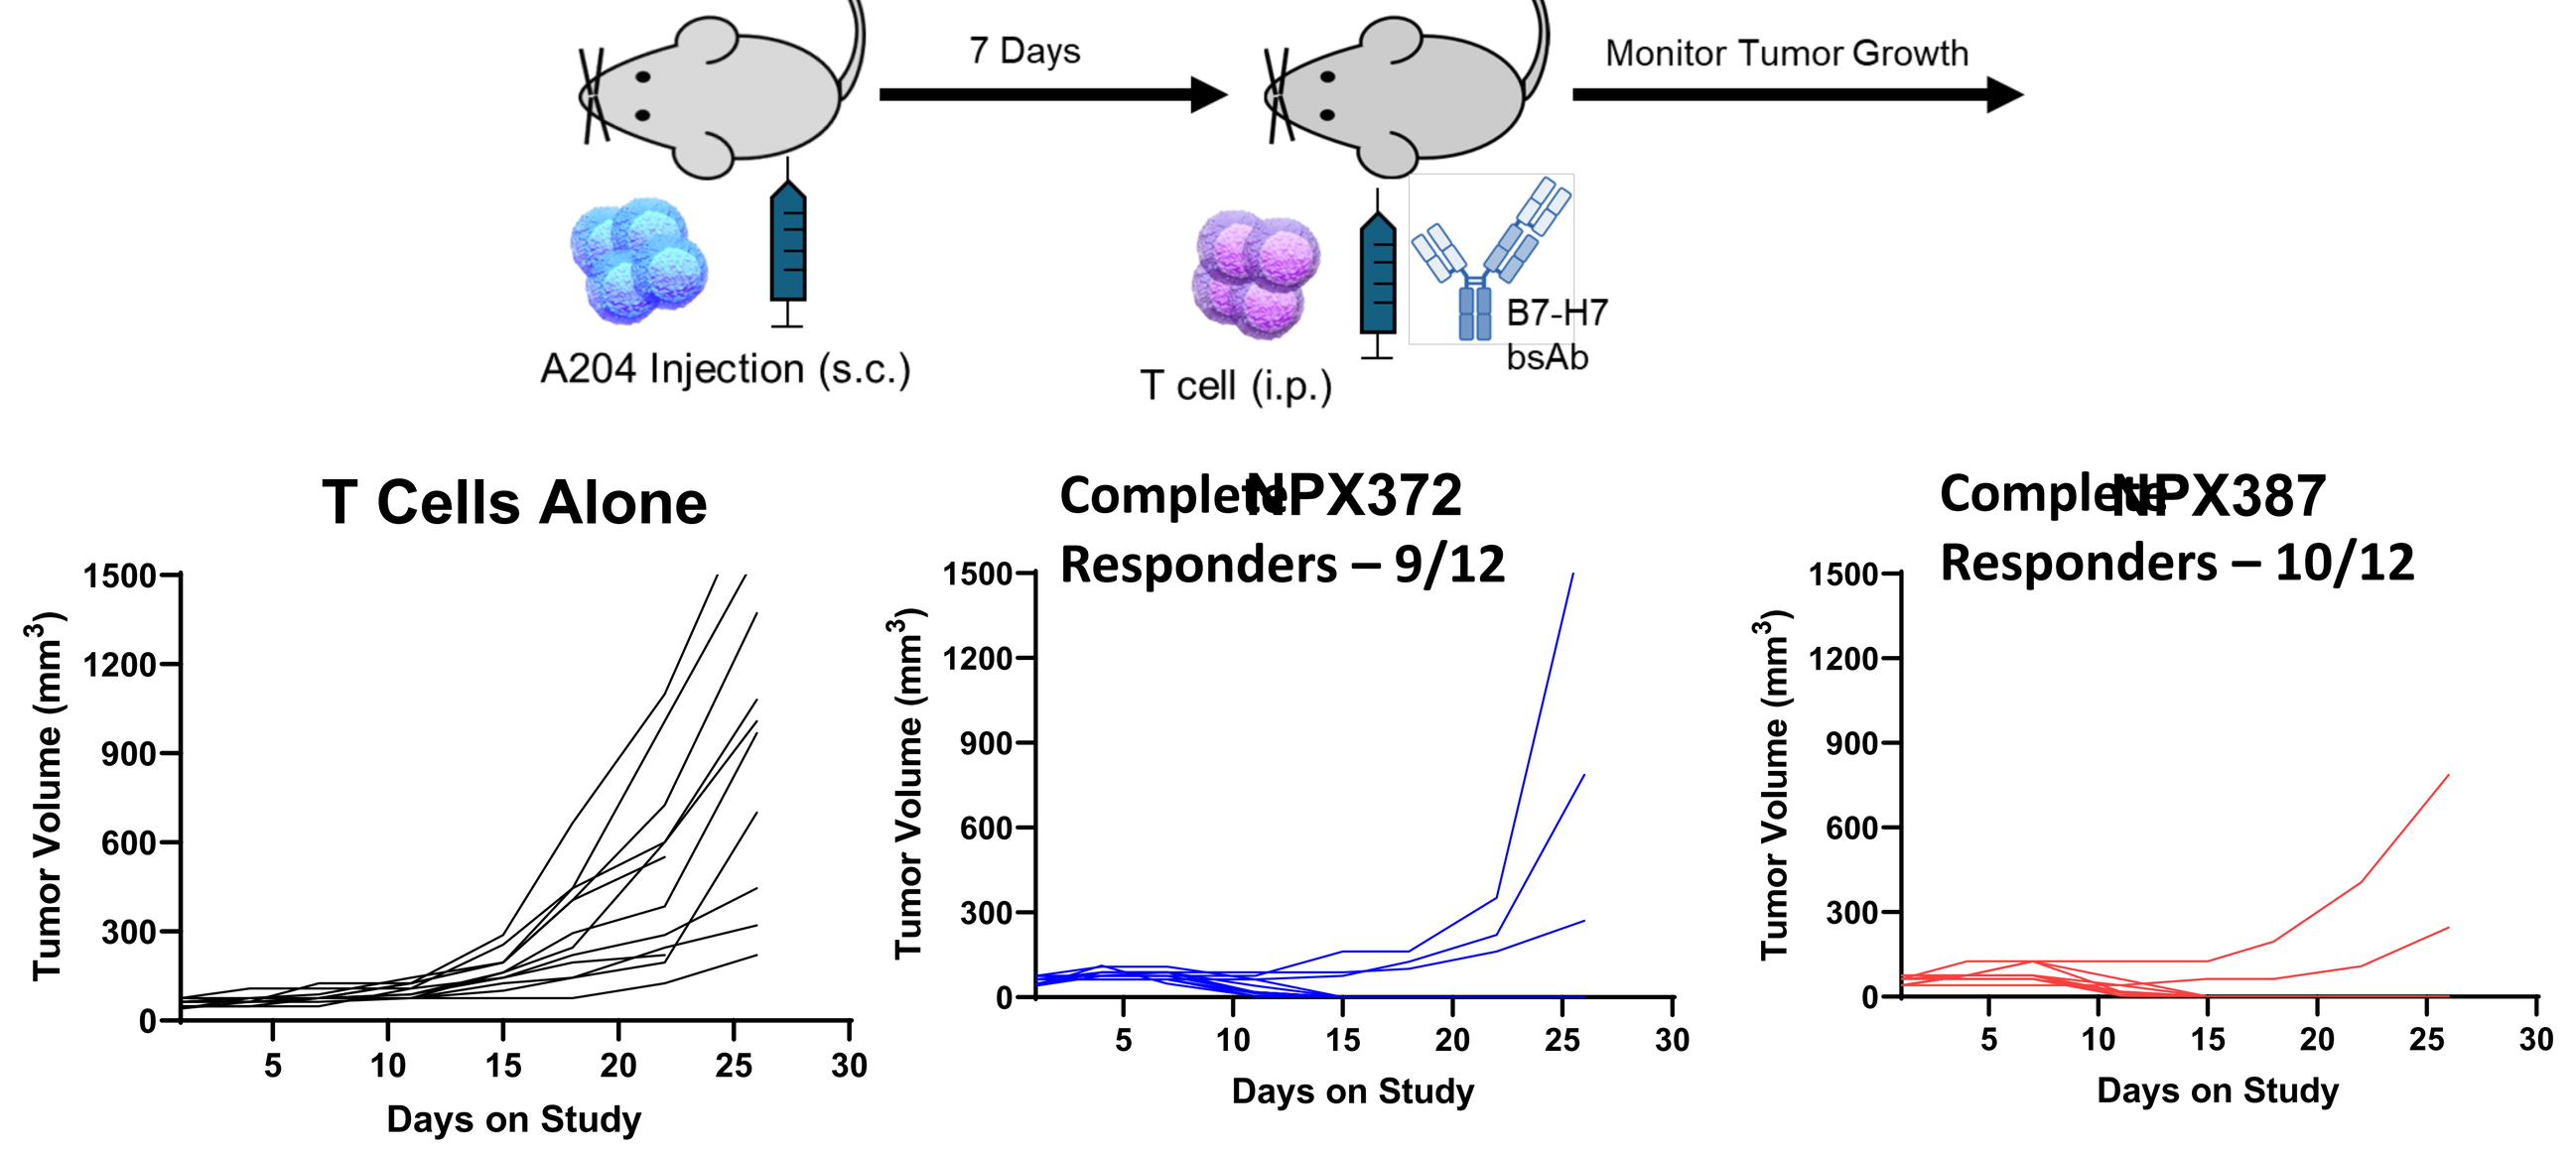
7 Days
Monitor Tumor Growth
A204 Injection (s.c.)
T cell (i.p.)
B7-H7 bsAb
T Cells Alone
NPX372 Complete Responders – 9/12
NPX387 Complete Responders – 10/12
Tumor Volume (mm³)
Days on Study

Female NCG mice with established s.c. A204 tumors were injected with activated T cells ($2x10^7$ cells) i.p. from two separate donors and treated with NPX372 or NPX387 (10mg/kg) i.v. Treatments were continued weekly for a total of four doses.

B7H7-CD3 Bispecific Antibody Screening Summary

- **NPX372 and NPX387 induce B7-H7 dependent T cell activation in vitro**
- **NPX372 and NPX387 induce T cell redirected killing of B7-H7+ tumor cell lines in vitro**
- **NPX387 induces higher levels of IFNγ and TNFα in vitro compared to NPX372**
- **NPX372 and NPX387 induce B7-H7+ tumor regression in humanized mice**
- **NPX372 was selected for further development**

NPX372 Induces PBMC Redirected Killing of B7-H7+ Tumor Cell Lines Spanning Multiple B7-H7 Expression Levels

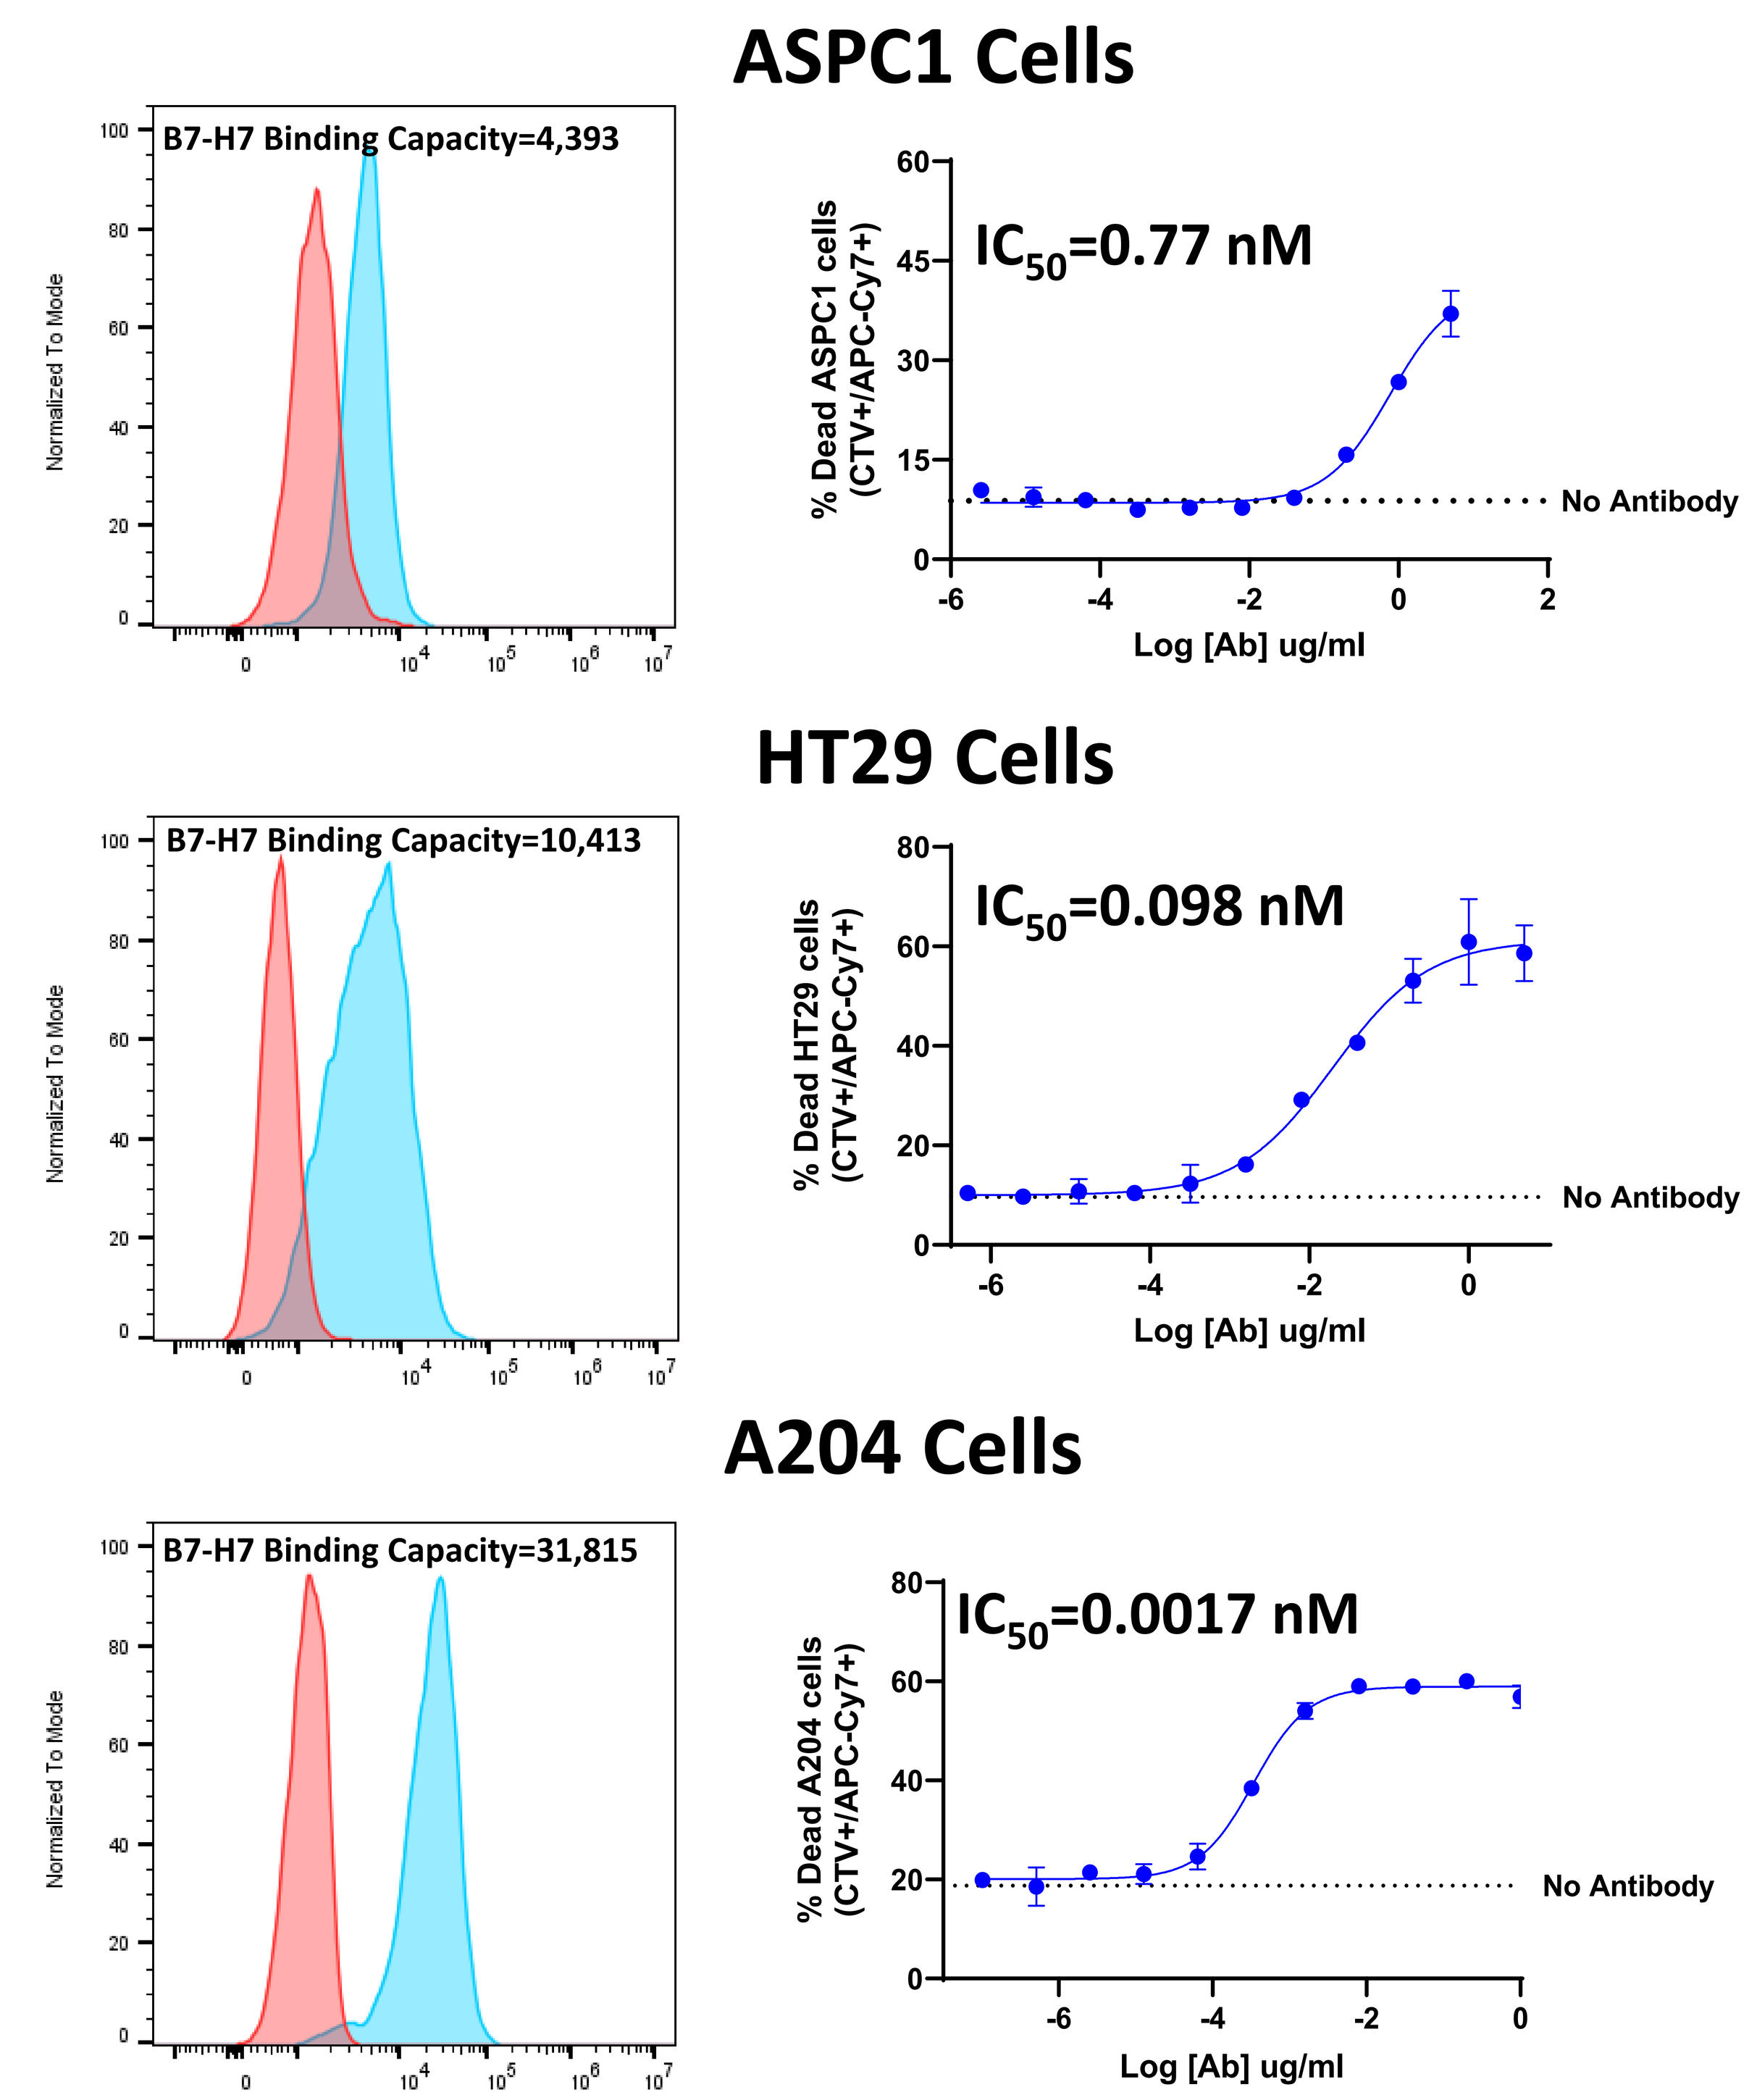
ASPC1 Cells
B7-H7 Binding Capacity=4,393
IC_{50}=0.77 nM
HT29 Cells
B7-H7 Binding Capacity=10,413
IC_{50}=0.098 nM
A204 Cells
B7-H7 Binding Capacity=31,815
IC_{50}=0.0017 nM
No Antibody
Log [Ab] ug/ml
Normalized To Mode

Tumor cells were stained with cell trace violet (CTV) and co-cultured with primary human PBMCs at an E to T ratio of 10:1 in the presence of serially diluted bsAb. After 24 (A204) or 48 Hours (ASPC1 and HT29) hours, the frequency of dead target cells (CTV+ viability dye+ cells) was measured by flow cytometry.

NPX372 Induces A204 Tumor Regression in an Activated T Cell Humanized Mouse Model at Clinically Relevant Doses

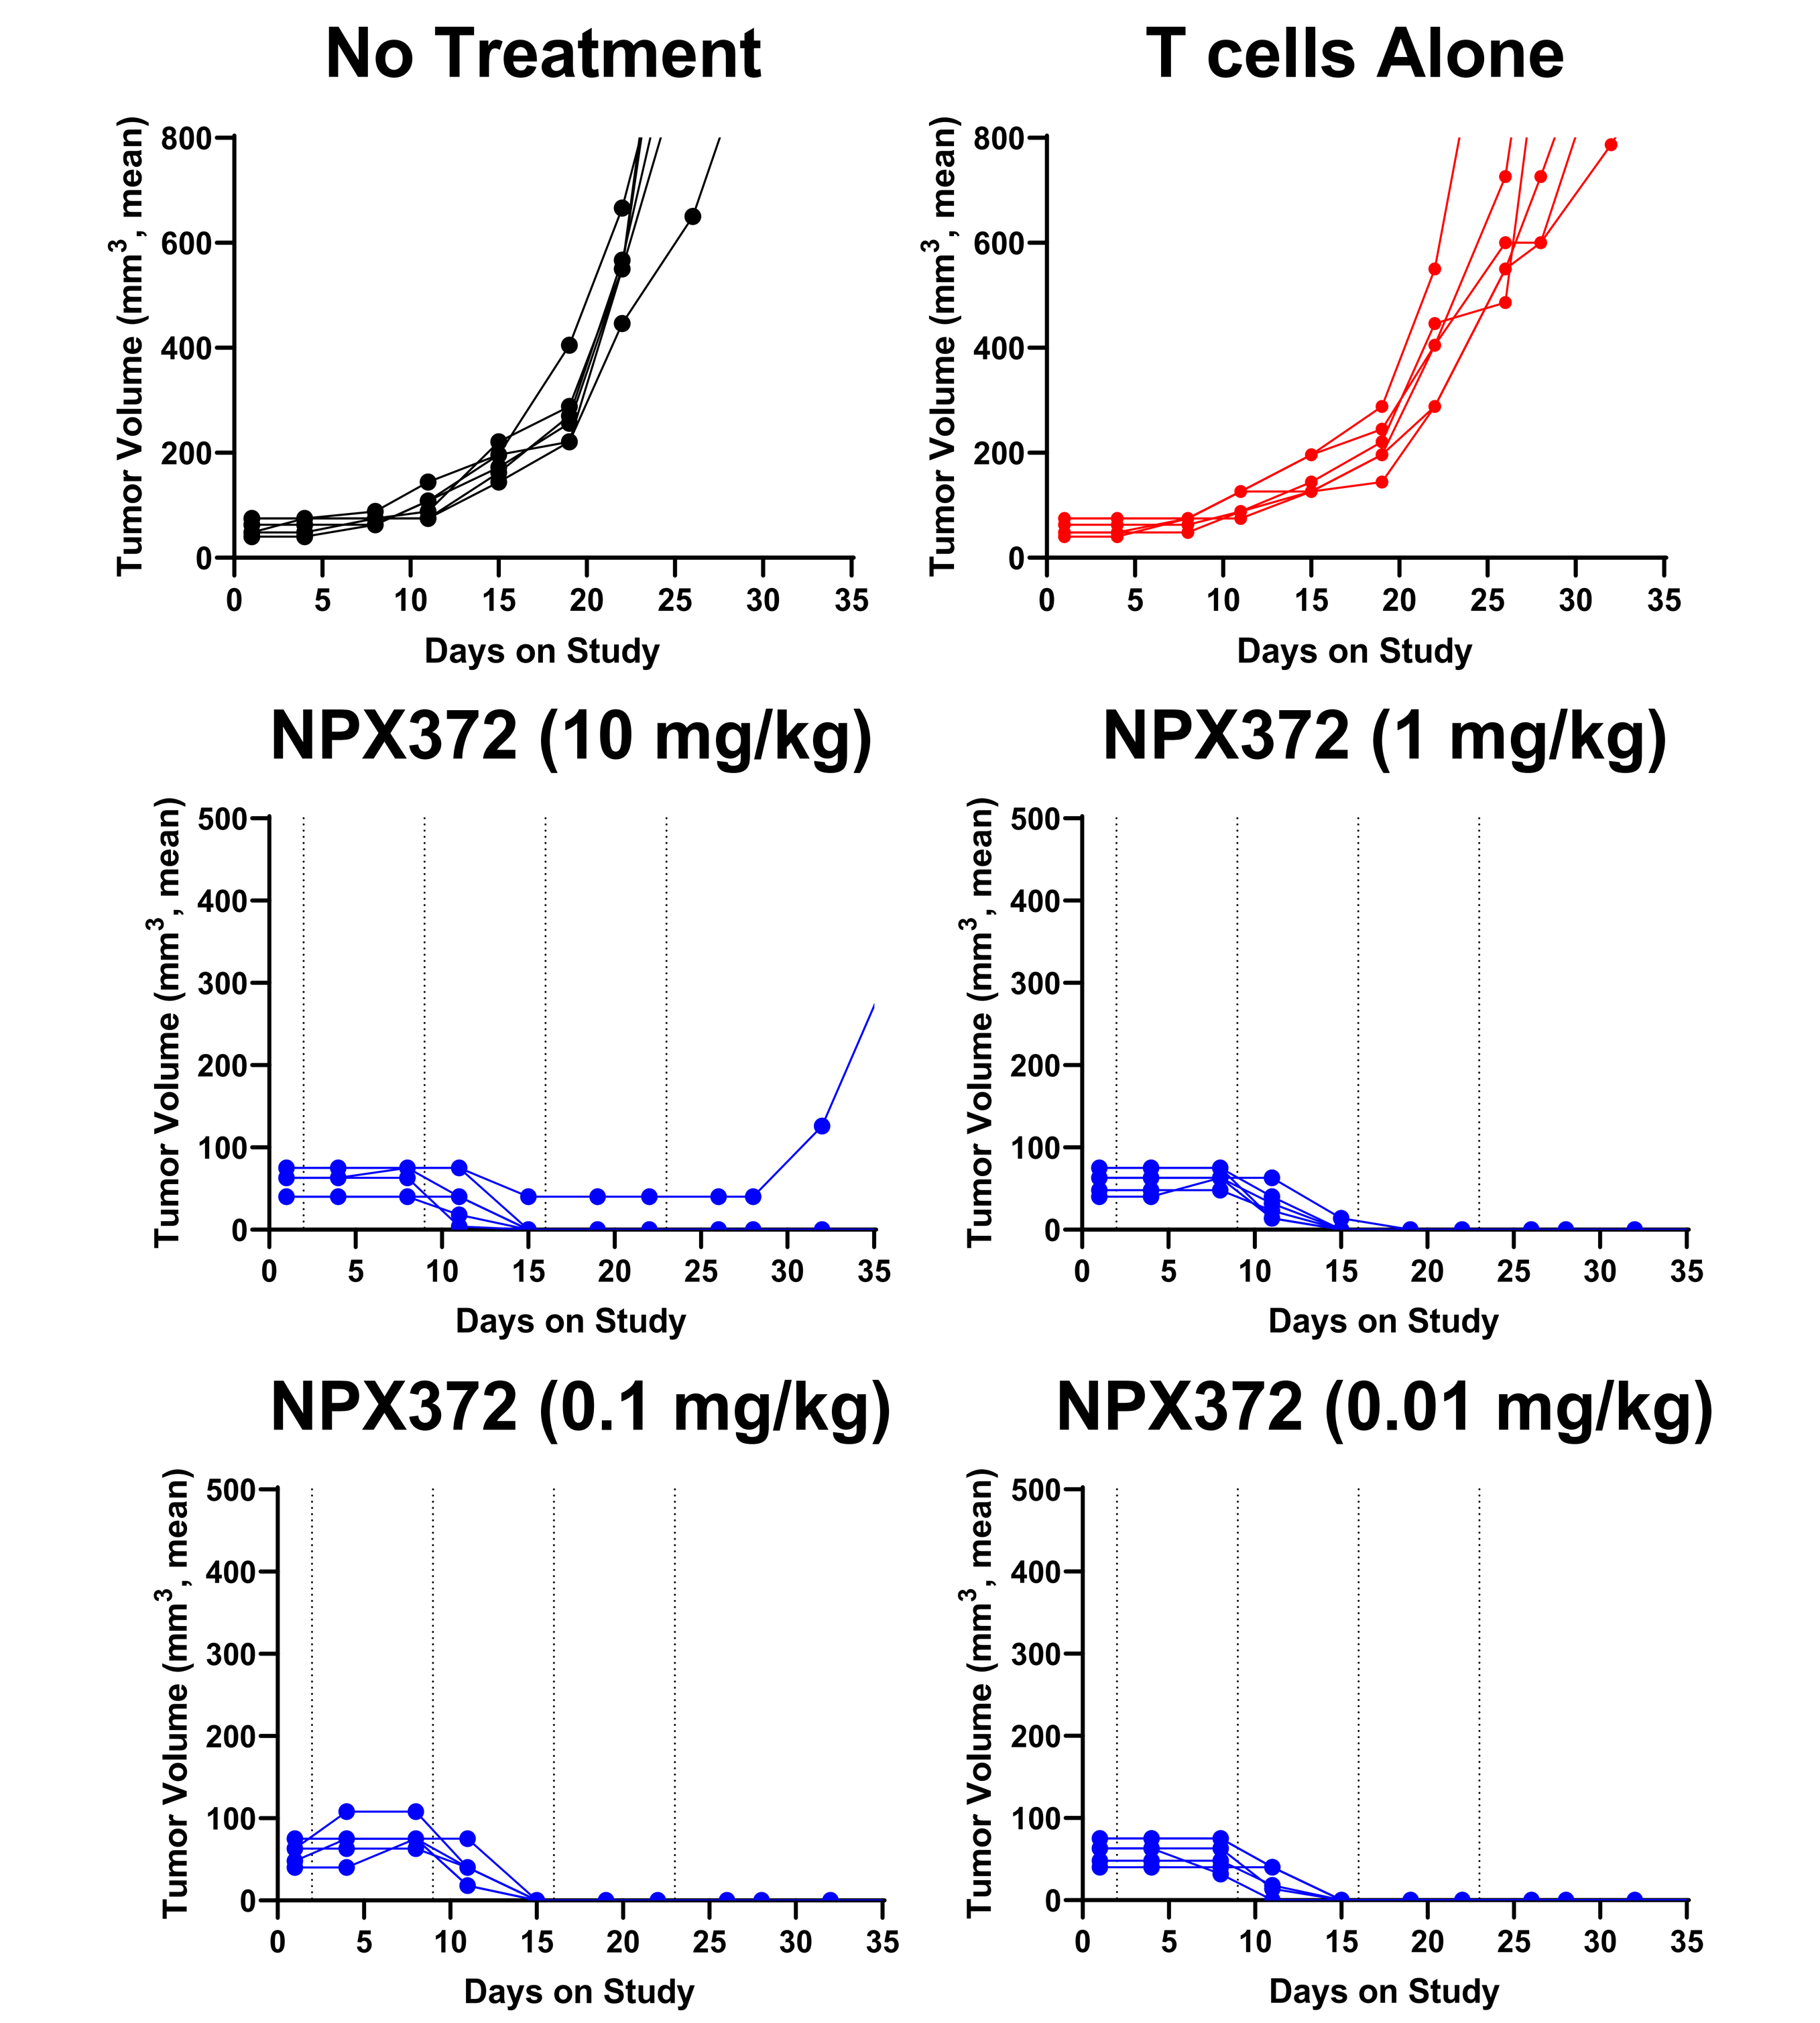
No Treatment
T cells Alone
NPX372 (10 mg/kg)
NPX372 (1 mg/kg)
NPX372 (0.1 mg/kg)
NPX372 (0.01 mg/kg)
Tumor Volume (mm³, mean)
Days on Study

Female NCG mice with established s.c. A204 tumors were injected with activated T cells ($2x10^7$ cells) i.p. from two separate donors and treated with NPX372 i.v. Treatments were continued weekly for a total of four doses.

Conclusions

- **NPX372 induces killing of B7-H7+ tumors across a range of B7-H7 expression levels and induces B7-H7+ tumor regression in humanized mice at clinically relevant doses**
- **See our other posters (#3302, 4354, and 7336) for more information on our B7-H7 targeting strategies in cancer**

AACR 2025

Comprehensive Analysis of B7-H7/HHLA2 Expression in Pan-solid Tumors and its Potential Significance in Anti-tumor Immunity

#3302

Yasmin Hashamboy-Ramsay[1], Karishma Vekaria[1], Emilien Loeuillard[1], Melissa Hayes[1], Emma Ya-Ting Wang[1], Matthew Rausch[1], Tatiana Novobrantseva[1], Katalin Kis-Toth[1], Leena Gandhi[1]

[1]*NextPoint Therapeutics, Cambridge, MA*

Background

B7-H7/HHLA2 is a B7 family member expressed on select normal epithelium and upregulated in many tumors. B7-H7 suppresses T and NK cell function through an inhibitory receptor, KIR3DL3, and stimulates lymphocyte activation through a distinct receptor, TMIGD2. High levels of B7-H7 expression in tumor cells are frequently associated with poor prognosis and it may represent a novel target for therapeutic interventions. We have previously demonstrated that higher levels of B7-H7 are associated with increased tumor cell killing by a B7-H7-targeting T cell engager. Here, we systematically characterized B7-H7 expression across solid tumor histologies to assess the potential of B7-H7 as a biomarker for use in therapeutic targeting of B7-H7.

Methods

B7-H7 protein expression was assessed by immunohistochemistry (IHC) using a monoclonal antibody that has been widely used in the literature (566.1) and a commercially available monoclonal antibody (E1U6X). Hundreds of tumors across many indications and select normal tissues were characterized for B7-H7 expression. The performance of 566.1 and E1U6X was compared to develop a B7-H7 IHC assay that could be used for patient selection for B7-H7 targeting therapeutics including NPX887, a monoclonal antibody currently in clinical trials. Quantitative flow cytometry (qFC) was used to measure the number of molecules per cell across cell lines displaying different levels of B7-H7, and concordance of qFC and IHC measurements by E1U6X was established.

NextPoint Therapeutics Pipeline Targeting the B7-H7 Axis

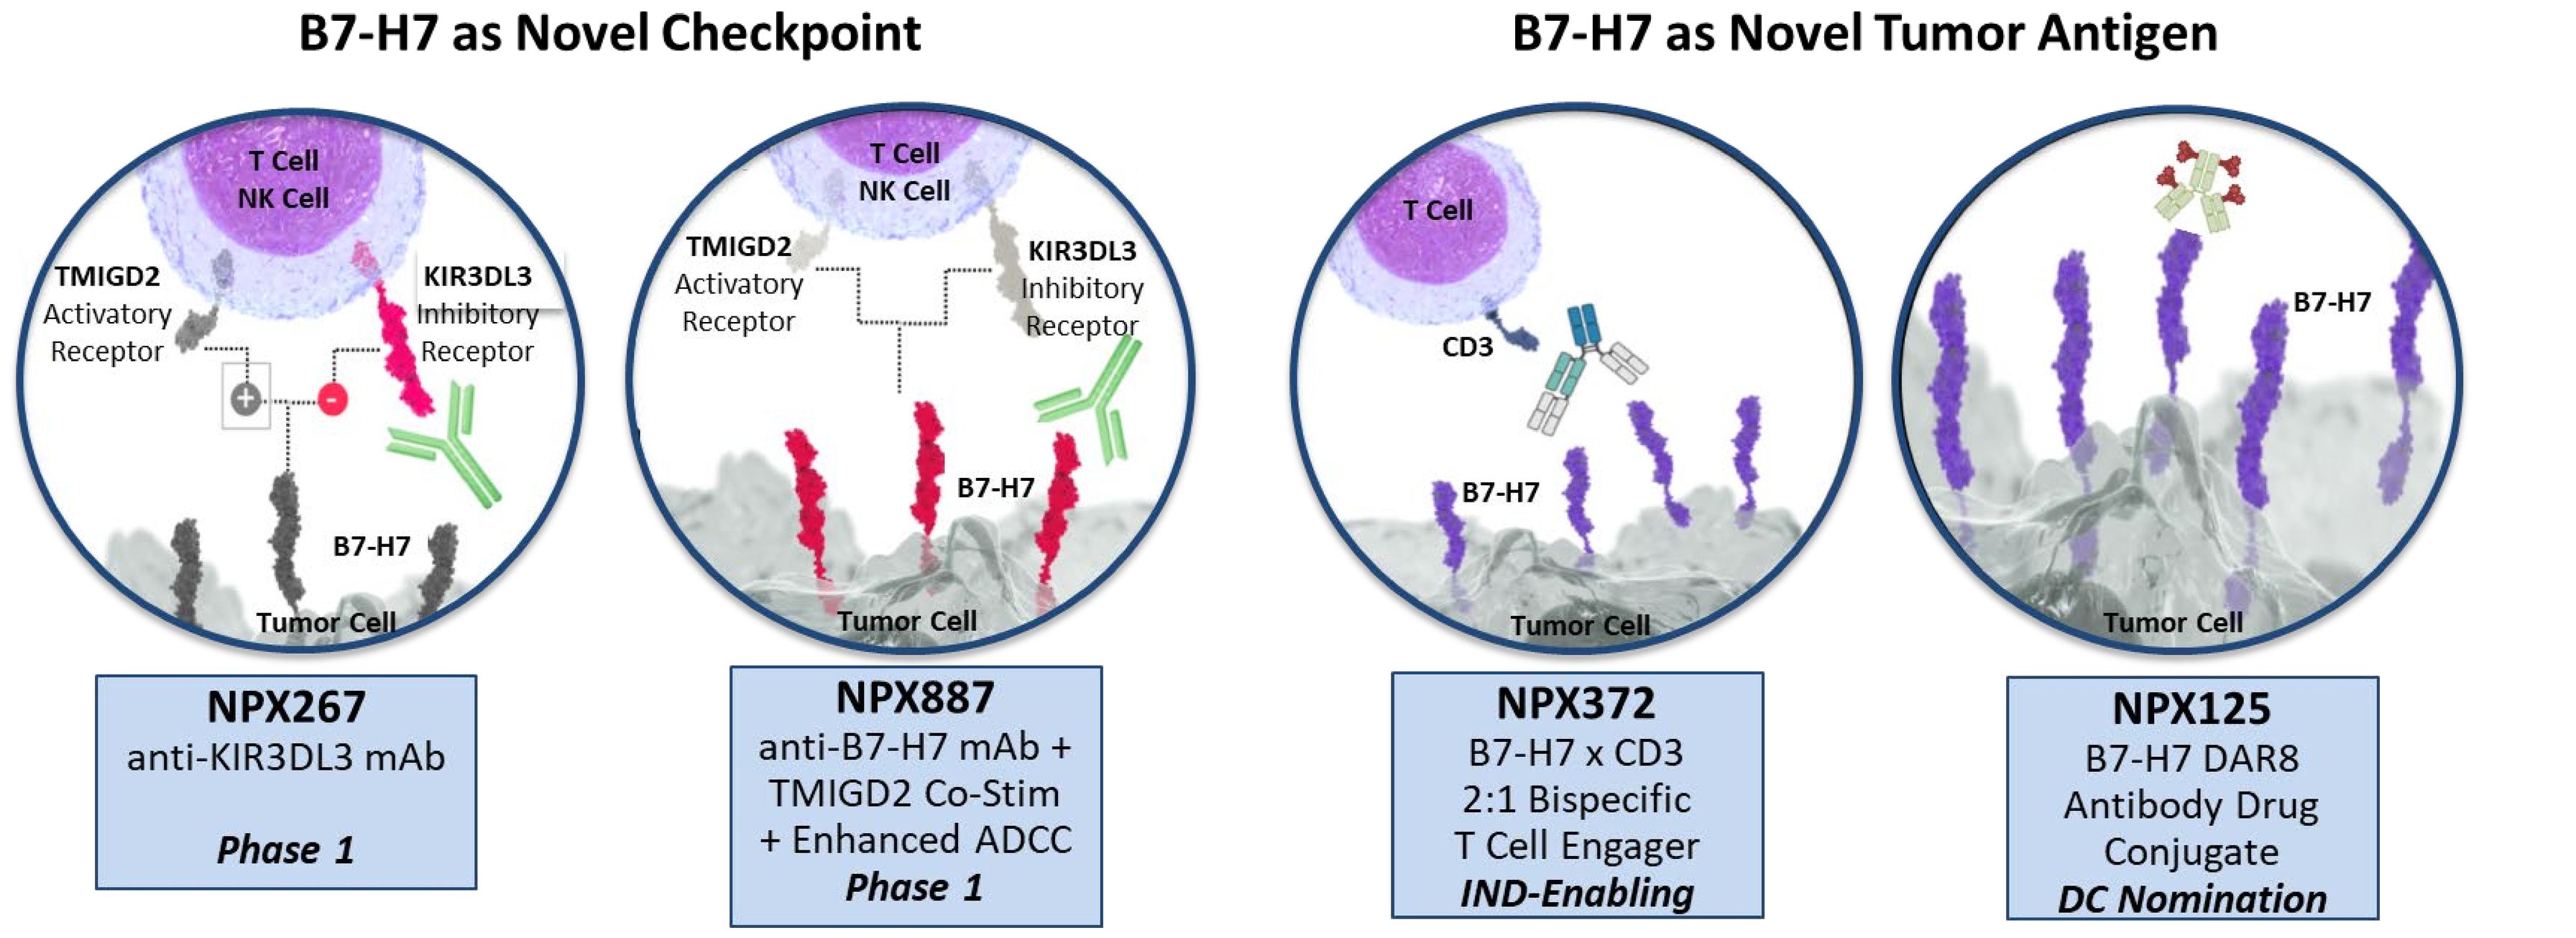
B7-H7 as Novel Checkpoint
B7-H7 as Novel Tumor Antigen
T Cell
NK Cell
TMIGD2 Activatory Receptor
KIR3DL3 Inhibitory Receptor
B7-H7
Tumor Cell
CD3
NPX267
anti-KIR3DL3 mAb
Phase 1
NPX887
anti-B7-H7 mAb + TMIGD2 Co-Stim + Enhanced ADCC
Phase 1
NPX372
B7-H7 x CD3 2:1 Bispecific T Cell Engager
IND-Enabling
NPX125
B7-H7 DAR8 Antibody Drug Conjugate
DC Nomination

Figure 1: NextPoint Therapeutics` antibody-based pipeline is supported by B7-H7 as clinical biomarker

Combined Literature and Internally Generated Data on Protein Expression Highlights Broad Expression of B7-H7 on Tumors

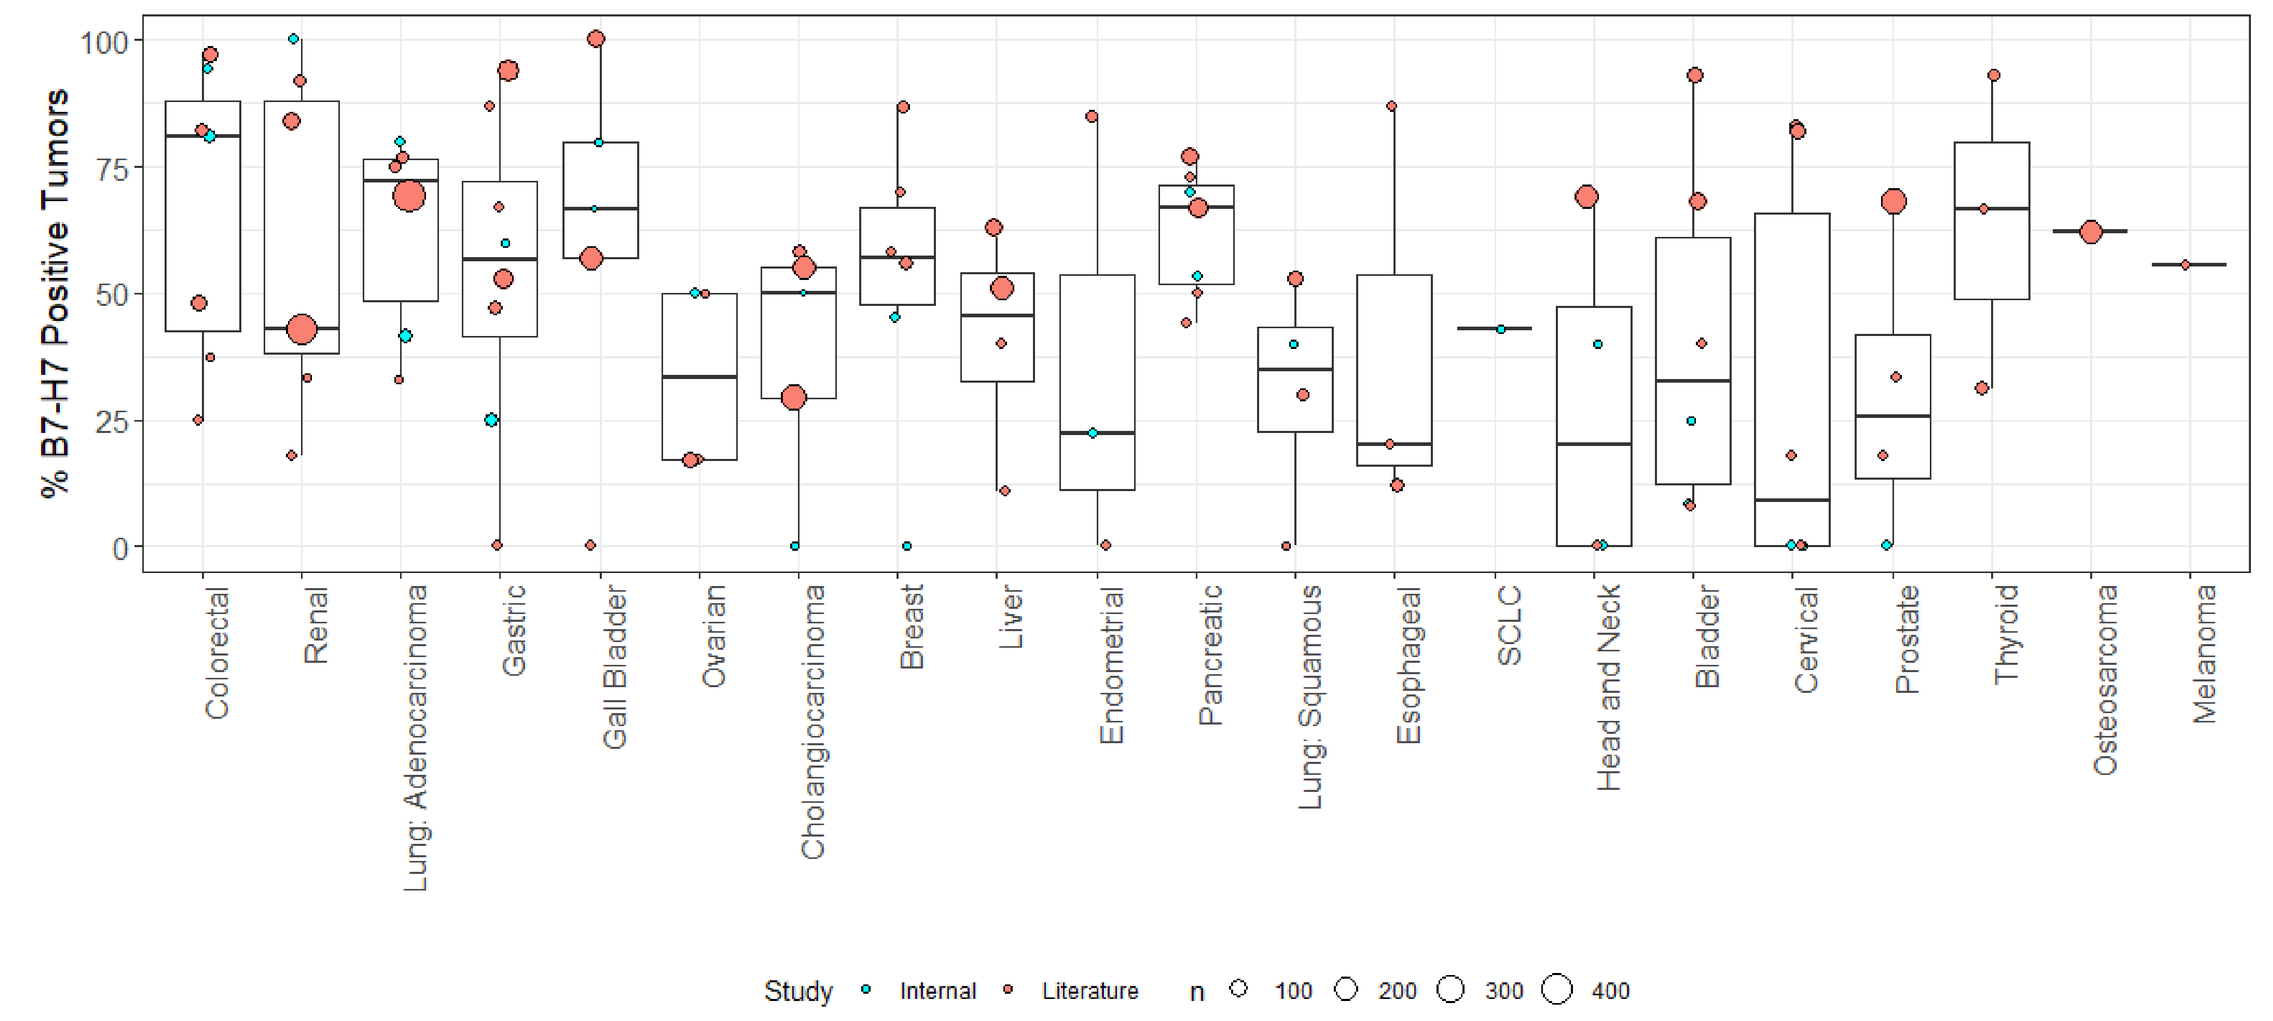
% B7-H7 Positive Tumors
Colorectal
Renal
Lung: Adenocarcinoma
Gastric
Gall Bladder
Ovarian
Cholangiocarcinoma
Breast
Liver
Endometrial
Pancreatic
Lung: Squamous
Esophageal
SCLC
Head and Neck
Bladder
Cervical
Prostate
Thyroid
Osteosarcoma
Melanoma
Study Internal Literature
n 100 200 300 400

Tool Ab Used	Number of Studies
566.1 mAb	11[1-8]
E1U6X (CST) mAb	1
Ab214327 (Abcam) pAb	8[9-16]
HPA055478 (Sigma) pAb	4[17-20]
Other polyclonal Abs	4[21-24]
Unknown	2[25-26]

Figure 2: B7-H7 protein expression was assessed across 30 studies by IHC using a variety of monoclonal (mAb) and polyclonal (pAb) antibodies.

Tumor Cells from CRC, NSCLC, Pancreatic, Renal and Gastric Cancer Display the Highest Protein Levels of B7-H7

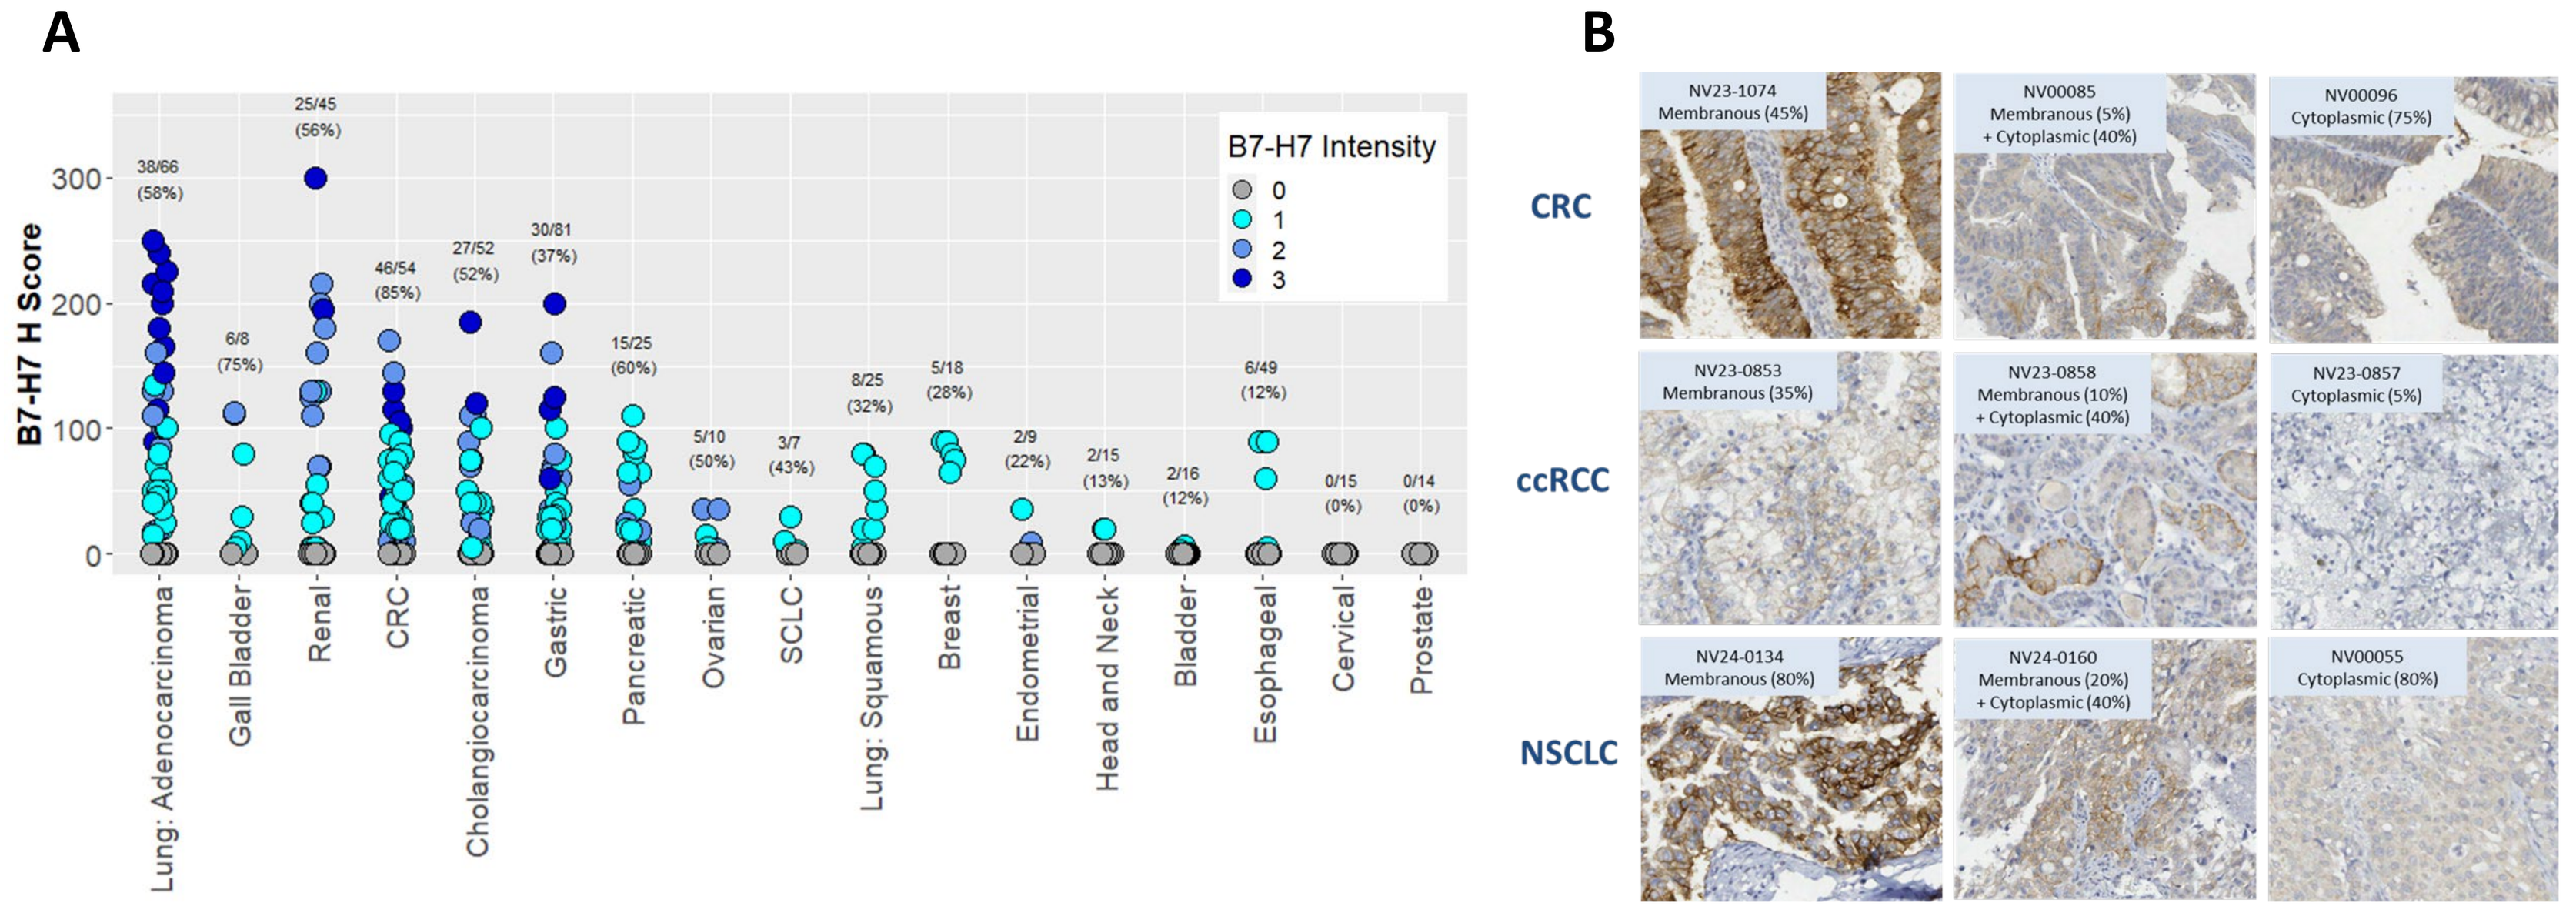
A
B
B7-H7 H Score
B7-H7 Intensity
38/66 (58%)
6/8 (75%)
25/45 (56%)
46/54 (85%)
27/52 (52%)
30/81 (37%)
15/25 (60%)
5/10 (50%)
3/7 (43%)
8/25 (32%)
5/18 (28%)
2/9 (22%)
2/15 (13%)
2/16 (12%)
6/49 (12%)
0/15 (0%)
0/14 (0%)
Lung: Adenocarcinoma
Gall Bladder
Renal
CRC
Cholangiocarcinoma
Gastric
Pancreatic
Ovarian
SCLC
Lung: Squamous
Breast
Endometrial
Head and Neck
Bladder
Esophageal
Cervical
Prostate
CRC
ccRCC
NSCLC
NV23-1074 Membranous (45%)
NV00085 Membranous (5%) + Cytoplasmic (40%)
NV00096 Cytoplasmic (75%)
NV23-0853 Membranous (35%)
NV23-0858 Membranous (10%) + Cytoplasmic (40%)
NV23-0857 Cytoplasmic (5%)
NV24-0134 Membranous (80%)
NV24-0160 Membranous (20%) + Cytoplasmic (40%)
NV00055 Cytoplasmic (80%)

Figure 3: B7-H7 protein expression was assessed by IHC using two monoclonal antibodies, either 566.1 or E1U6X. 509 tumors across 17 indications were characterized for B7-H7 expression. (A) H score and predominant intensity is plotted for each tumor sample, and the % of B7-H7 positive tumor samples is reported for each indication. (B) Representative tumor images using 566.1. B7-H7 is detected on the cytoplasm and the membrane of tumor cells.

Normal Tissue Expression of B7-H7 is Restricted to Organ Substructures within the Intestine, Lung and Kidney, and is Absent in Peripheral Blood

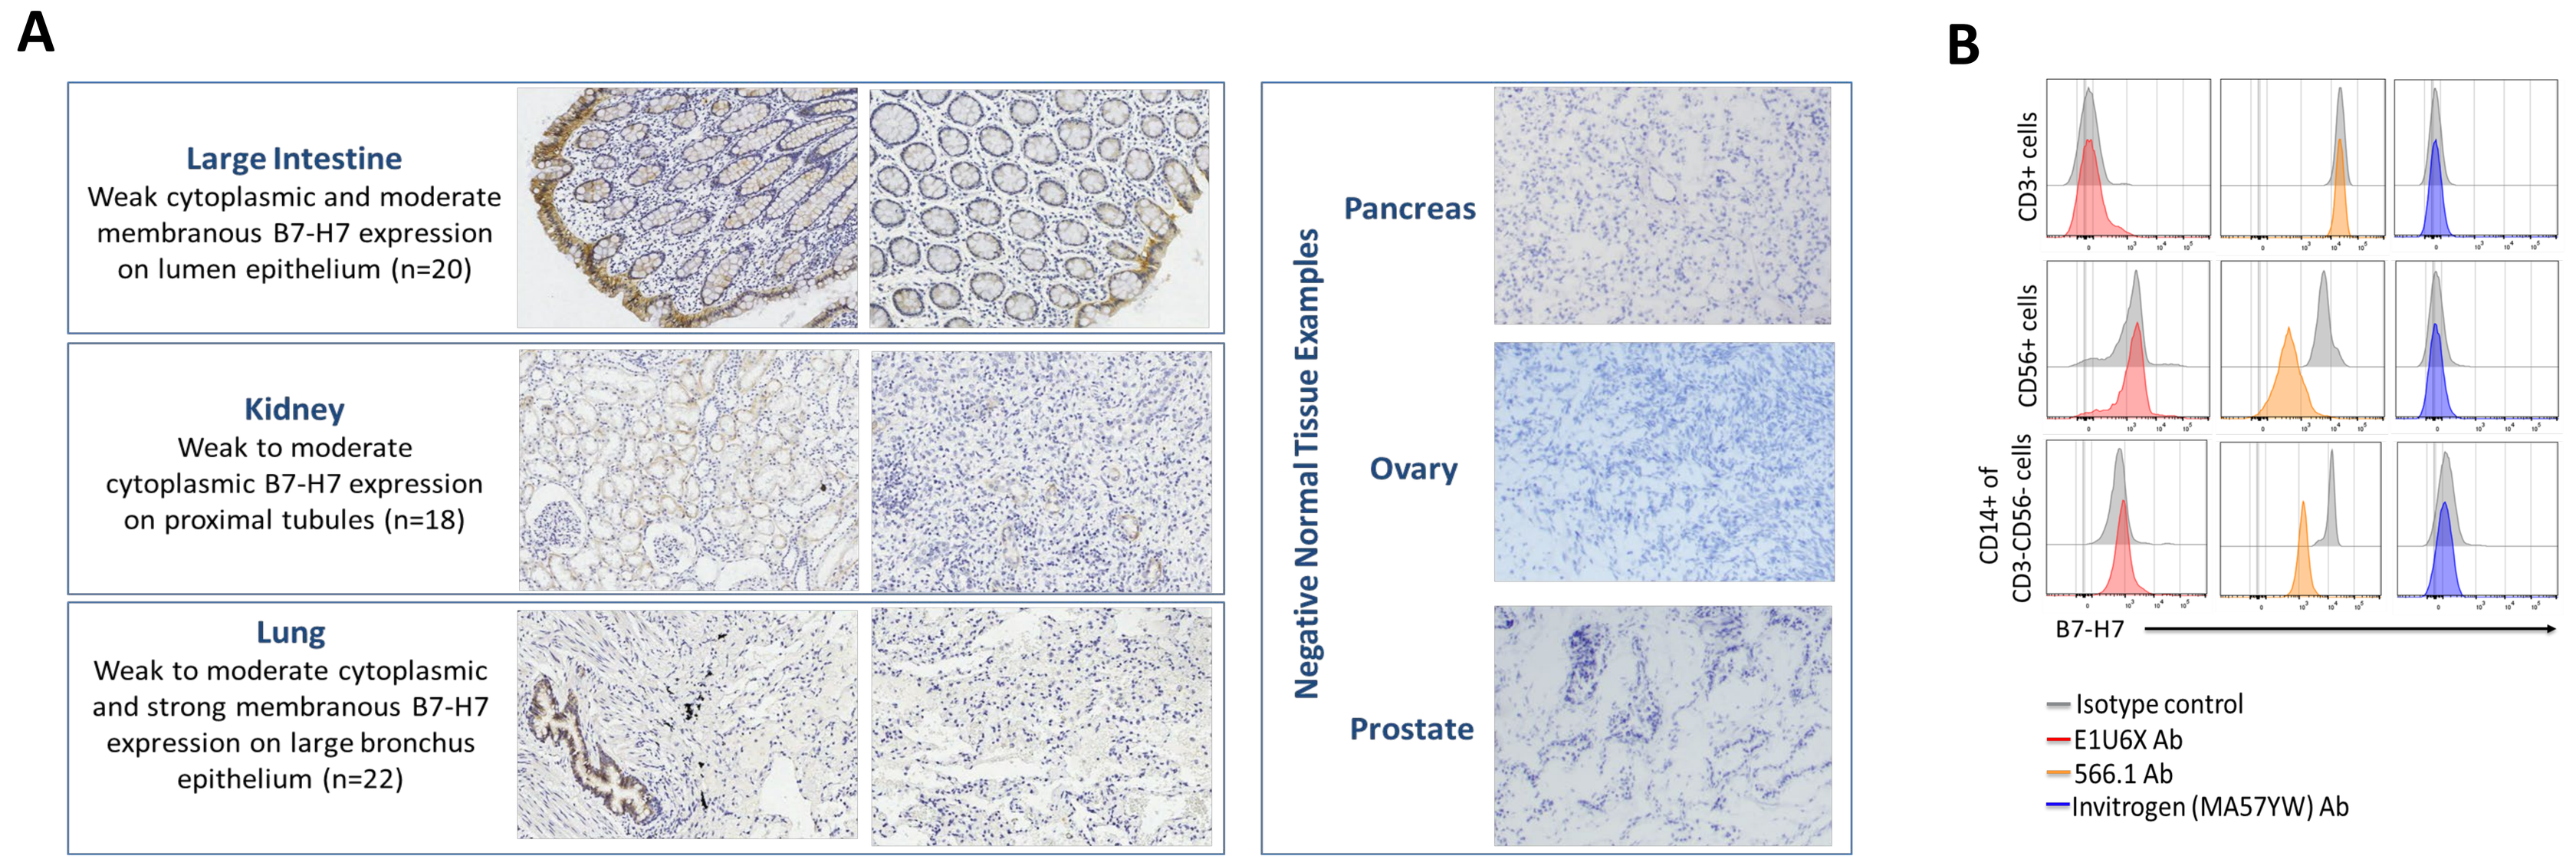
A
Large Intestine
Weak cytoplasmic and moderate membranous B7-H7 expression on lumen epithelium (n=20)
Kidney
Weak to moderate cytoplasmic B7-H7 expression on proximal tubules (n=18)
Lung
Weak to moderate cytoplasmic and strong membranous B7-H7 expression on large bronchus epithelium (n=22)
Negative Normal Tissue Examples
Pancreas
Ovary
Prostate
B
CD3+ cells
CD56+ cells
CD14+ of CD3-CD56- cells
B7-H7
Isotype control
E1U6X Ab
566.1 Ab
Invitrogen (MA57YW) Ab

Figure 4: (A) Normal B7-H7 expression was restricted to substructures of intestine (n=20) and lung (n=22) and showed only faint cytoplasmic staining in the kidney (n=18) and was absent in other normal tissues that were tested. (B) Representative histograms showing no detectable B7-H7 expression on T cells (CD3+), NK cells (CD56+) or monocytes (CD14+ of CD3-CD56-) in PBMCs by flow cytometry.

Similar Levels of B7-H7 Expression are Detected using Monoclonal Antibodies 566.1 and E1U6X

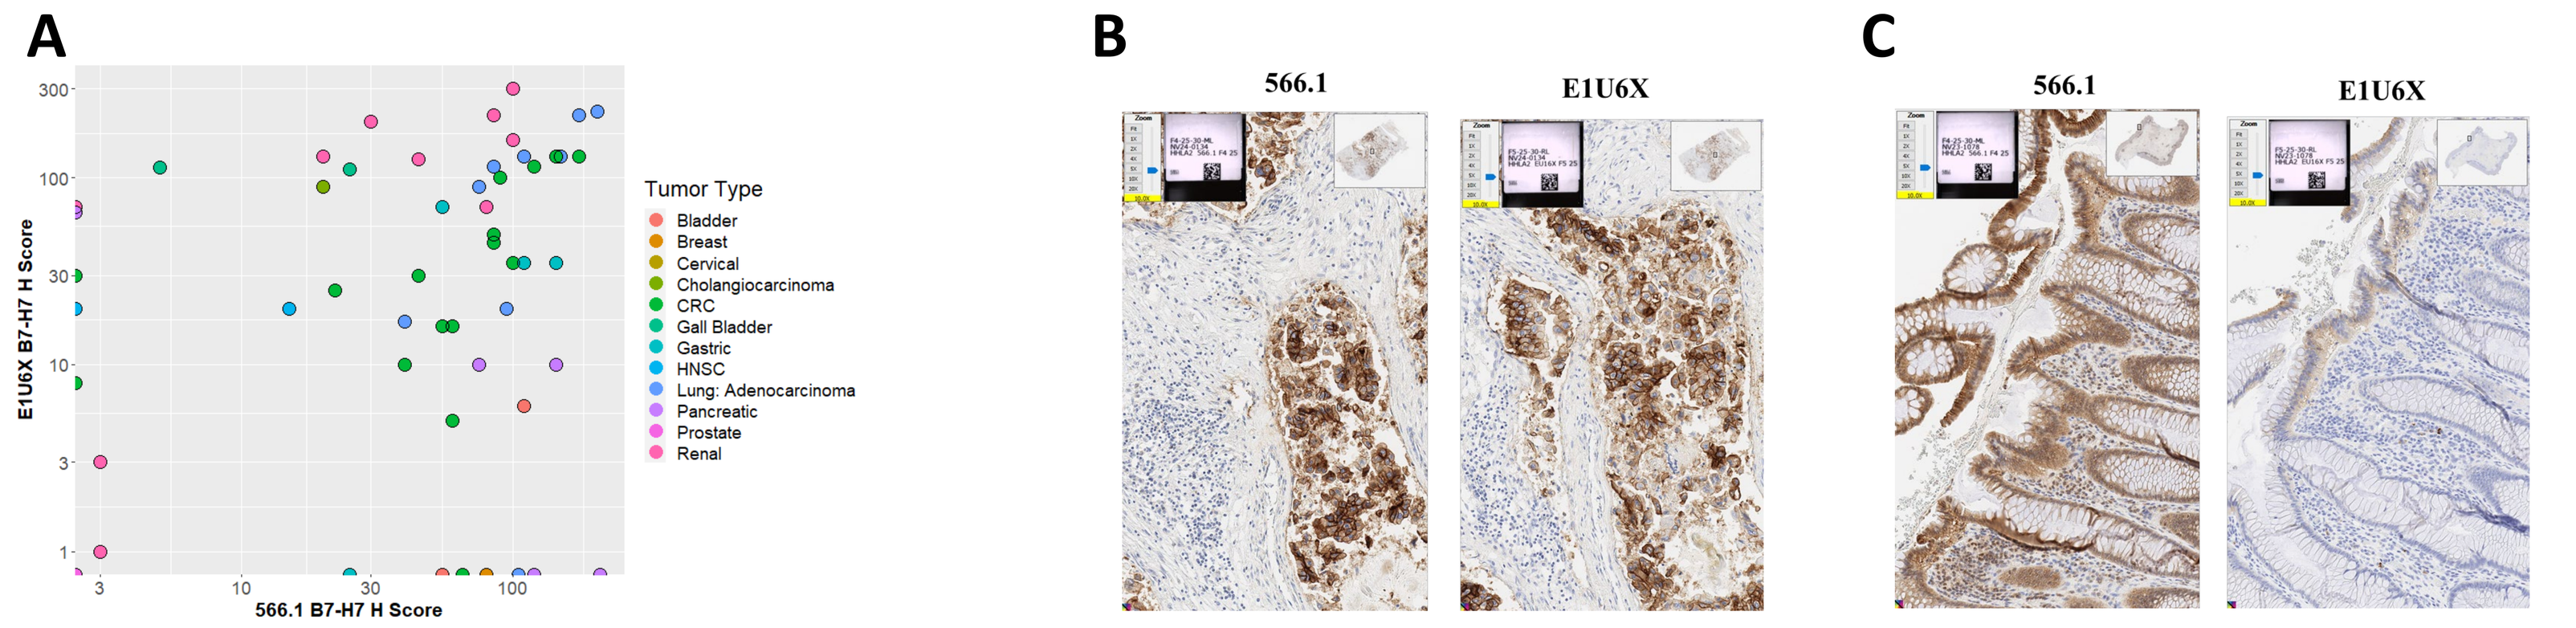
A
B
C
E1U6X B7-H7 H Score
566.1 B7-H7 H Score
Tumor Type
Bladder
Breast
Cervical
Cholangiocarcinoma
CRC
Gall Bladder
Gastric
HNSC
Lung: Adenocarcinoma
Pancreatic
Prostate
Renal
566.1
E1U6X

Figure 5: To develop a B7-H7 IHC assay for patient selection for B7-H7 targeting therapeutics including NPX887, a monoclonal antibody currently in clinical trials, the performance of 566.1 and E1U6X were compared. (A) B7-H7 H Scores assessed by 566.1 and E1U6X were compared in a subset of tumors (n=76). (B) During assay development, similar patterns of B7-H7 expression were detected using both antibodies, as shown in representative matched NSCLC samples. (C) The optimized assay using E1U6X displayed superior performance displaying less background in the positive control normal colon samples.

B7-H7 Molecule per Cell Assessment on Tumor Cell Lines Aligns Well with Immunohistochemistry Assessment of Human Tumor Samples

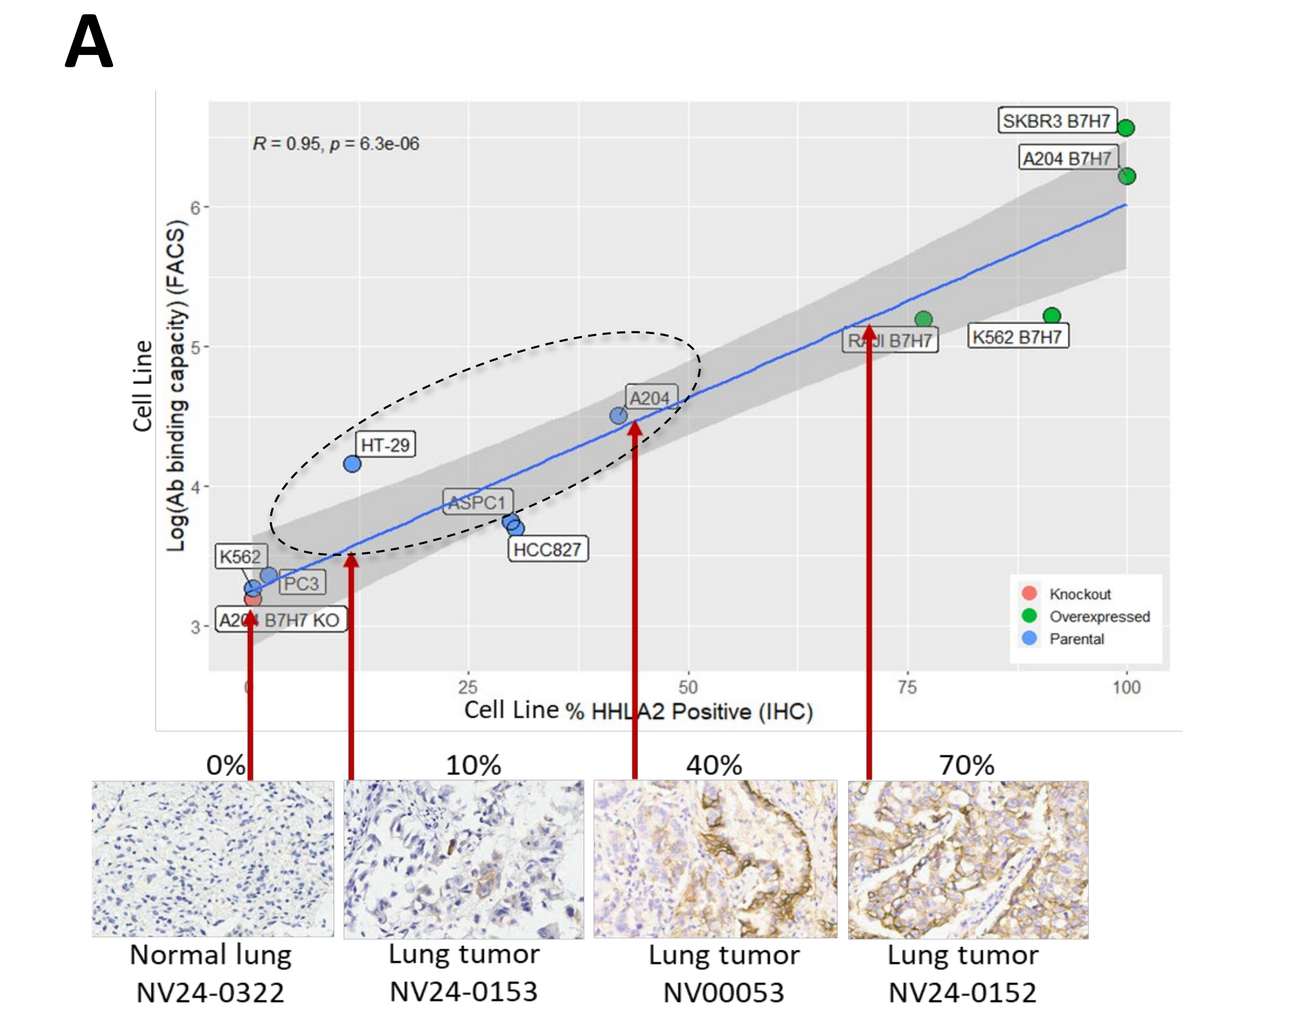
A
R = 0.95, p = 6.3e-06
Cell Line Log(Ab binding capacity) (FACS)
Cell Line % HHLA2 Positive (IHC)
SKBR3 B7H7
A204 B7H7
RKO B7H7
K562 B7H7
A204
HT-29
ASPC1
HCC827
K562
PC3
A204 B7H7 KO
Knockout
Overexpressed
Parental
0%
10%
40%
70%
Normal lung NV24-0322
Lung tumor NV24-0153
Lung tumor NV00053
Lung tumor NV24-0152

B

Tumor Type	Distribution of B7-H7 Positivity (% of Samples): Equal to 0	Between 0 and 15	Between 15 and 40	Between 40 and 100	*Number of samples*
Lung: Adenocarcinoma	43	6	14	37	*65*
CRC	18	18	29	35	*55*
Breast	72	0	0	28	*18*
Gall Bladder	25	25	25	25	*8*
Kidney	47	16	13	24	*45*
Pancreatic	52	20	4	24	*25*
Cholangiocarcinoma	48	15	21	15	*52*
Lung: Squamous	69	4	12	15	*26*
Esophageal	88	4	0	8	*49*
Gastric	73	2	20	5	*44*
Head & Neck: Squamous	88	0	13	0	*16*
Prostate	100	0	0	0	*14*
Cervical	100	0	0	0	*15*
Bladder	88	13	0	0	*16*
Lung: SCLC	57	29	14	0	*7*
Ovarian	60	20	20	0	*10*
Mesothelioma	100	0	0	0	*3*
Endometrial	78	11	11	0	*9*

Figure 6: (A) qFC was used to measure the number of molecules per cell across cancer cell lines displaying different levels of B7-H7, and concordance of qFC and IHC measurements by E1U6X was established. B7-H7 levels on tumor samples are comparable to tumor cell lines effectively killed by immune cells in the presence of anti-B7-H7 therapeutic antibodies, as previously presented[27]. In vitro data for cell lines highlighted in dashed circle is also shown in AACR 2025 poster #1556. (B) Distribution of positive B7-H7 tumor samples based on IHC assessment using E1U6X or 566.1 tool antibodies.

Conclusions

We demonstrated that normal tissue expression of B7-H7 is restricted to organ substructures within the intestine, lung, and kidney. We observed significant expression of B7-H7 across tumor cells of varying histologies, highlighting patient populations for B7-H7 targeting therapies. The IHC assay using E1U6X was selected for investigational use in ongoing (NPX887-001; NCT06240728) and potential future clinical studies of therapeutic molecules targeting the B7-H7 axis.

References

1. Zhang et al, *Front in Cell and Develop Biol,* 2020
2. Cheng et al, *Clin Can Res*, 2017
3. Janakiram et al, *Clin Can Res,* 2015
4. Boor et al, *Brit J of Cancer*, 2020
5. Zhou et al, *J for Immun of Cancer*, 2021
6. Xu et al, *Cancer Cell Int*, 2021
7. Koirala et al, *Sci Reports*, 2016
8. Wang et al, *Cancers*, 2023
9. Zhu et al, *OncoTargets and Ther*, 2018
10. Luo et al, *J of Inflammation Res*, 2021
11. Zhang et al, *Int J of Oncology*, 2022
12. Lv et al, *Front in Immun*, 2022
13. Niu et al, *Japanese J of Clin Onc*, 2022
14. Xiao et al, *Disease Markers*, 2019
15. Nishihara et al, *Int Urology and Nephrology,* 2023
16. Sun et al, *World J of Surgical Onc*, 2023
17. Human Protein Atlas
18. Liao et al, *Amer J of Clin Path*, 2022
19. Yan et al, *Cancer Letters*, 2019
20. Zhou et al, *J for Immunotherapy of Cancer,* 2020
21. Kula et al, *Int J of Mol Sci*, 2023
22. Bhatt et al, *Cancer Immun Res*, 2021
23. Byun et al, *Medicine*, 2021
24. Jing et al, *J for Immunotherapy of Cancer*, 2019
25. Wei et al, *Sci Immunology*, 2021
26. Li et al, *Int J of Biol Sci*, 2020
27. Vekaria et al, *SITC Annual Meeting*, 2024

nextpoint

Abstract #4354

Safety and tolerability of NPX372, a novel B7-H7:CD3 bispecific T cell engaging antibody

Silvia Ferrati[1], Matthew Rausch[1], Susannah Hewitt[1], Katalin Kis-Toth[1], Bijan Etemad-Gilbertson[1], Leena Gandhi[1], Tatiana Novobrantseva[1]

[1]*NextPoint Therapeutics, Cambridge, MA*

BACKGROUND

B7-H7 is a B7 family member upregulated on tumor cells of many histologic subtypes with a very restricted normal tissue expression and is, therefore, being explored as a novel tumor targeting antigen.

NextPoint Therapeutics has developed a T cell engager bispecific antibody, NPX372, which simultaneously recognizes B7-H7 and CD3 from both human and non-human primates, providing T cell activation in the proximity of B7-H7 (Poster # 5493).

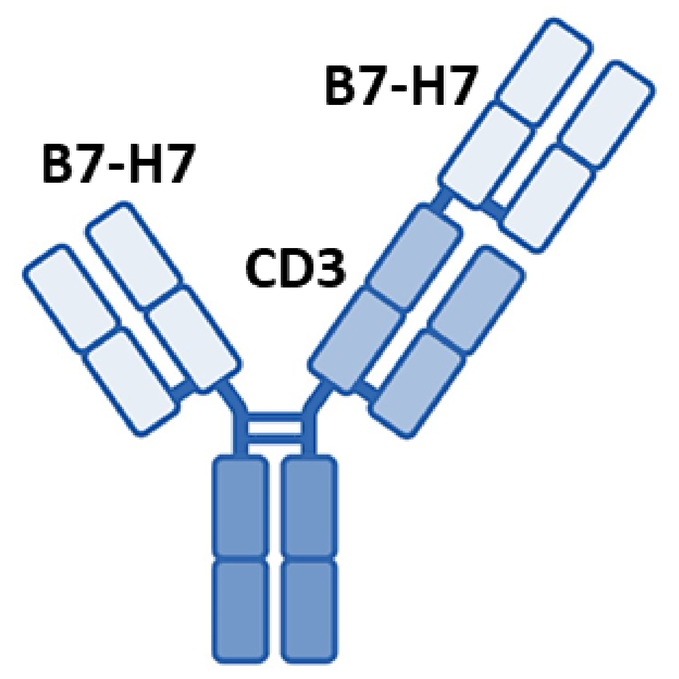
B7-H7
B7-H7
CD3

Mechanism of Action

NPX372 induces primary CD8+ T cell activation after B7-H7 crosslinking

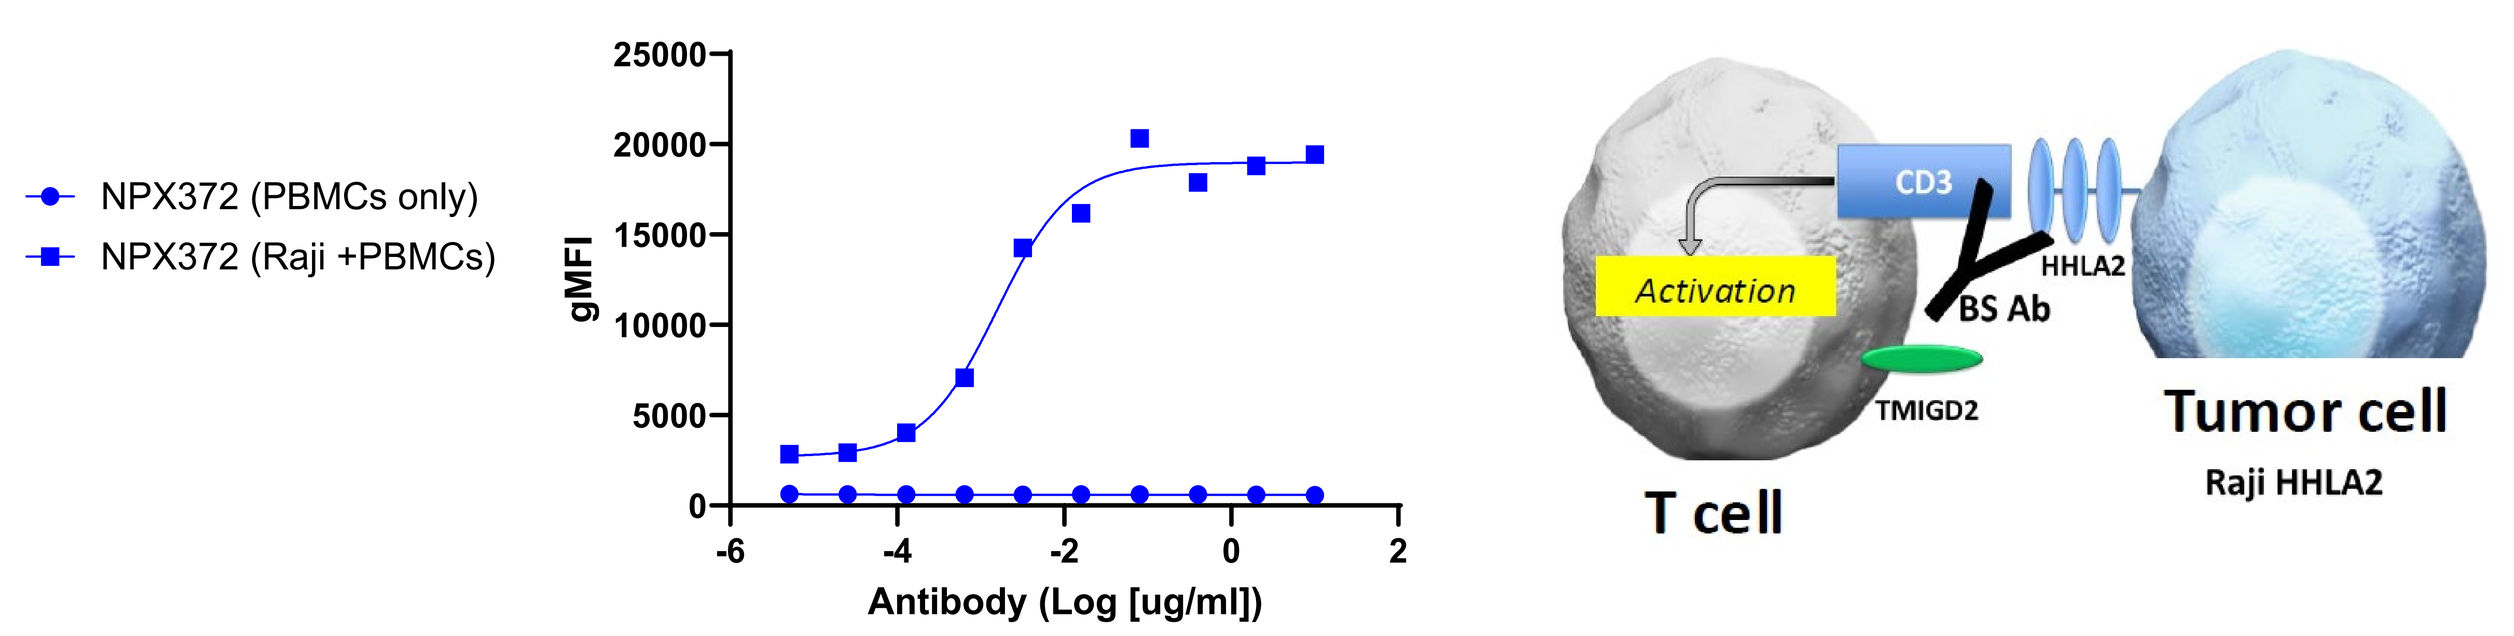
NPX372 (PBMCs only)
NPX372 (Raji +PBMCs)
gMFI
Antibody (Log [ug/ml])
CD3
Activation
HHLA2
BS Ab
TMIGD2
T cell
Tumor cell
Raji HHLA2

Raji-B7-H7 cells were plated with serially diluted NPX372 for 30 minutes followed by addition of human PBMCs. After 24 hours, upregulation of CD25 was measured by flow cytometry on CD8+ T cells.

NPX372 Induces PBMC Redirected Killing of B7-H7+ Tumor Cell Lines Spanning Multiple B7-H7+ Expression Levels

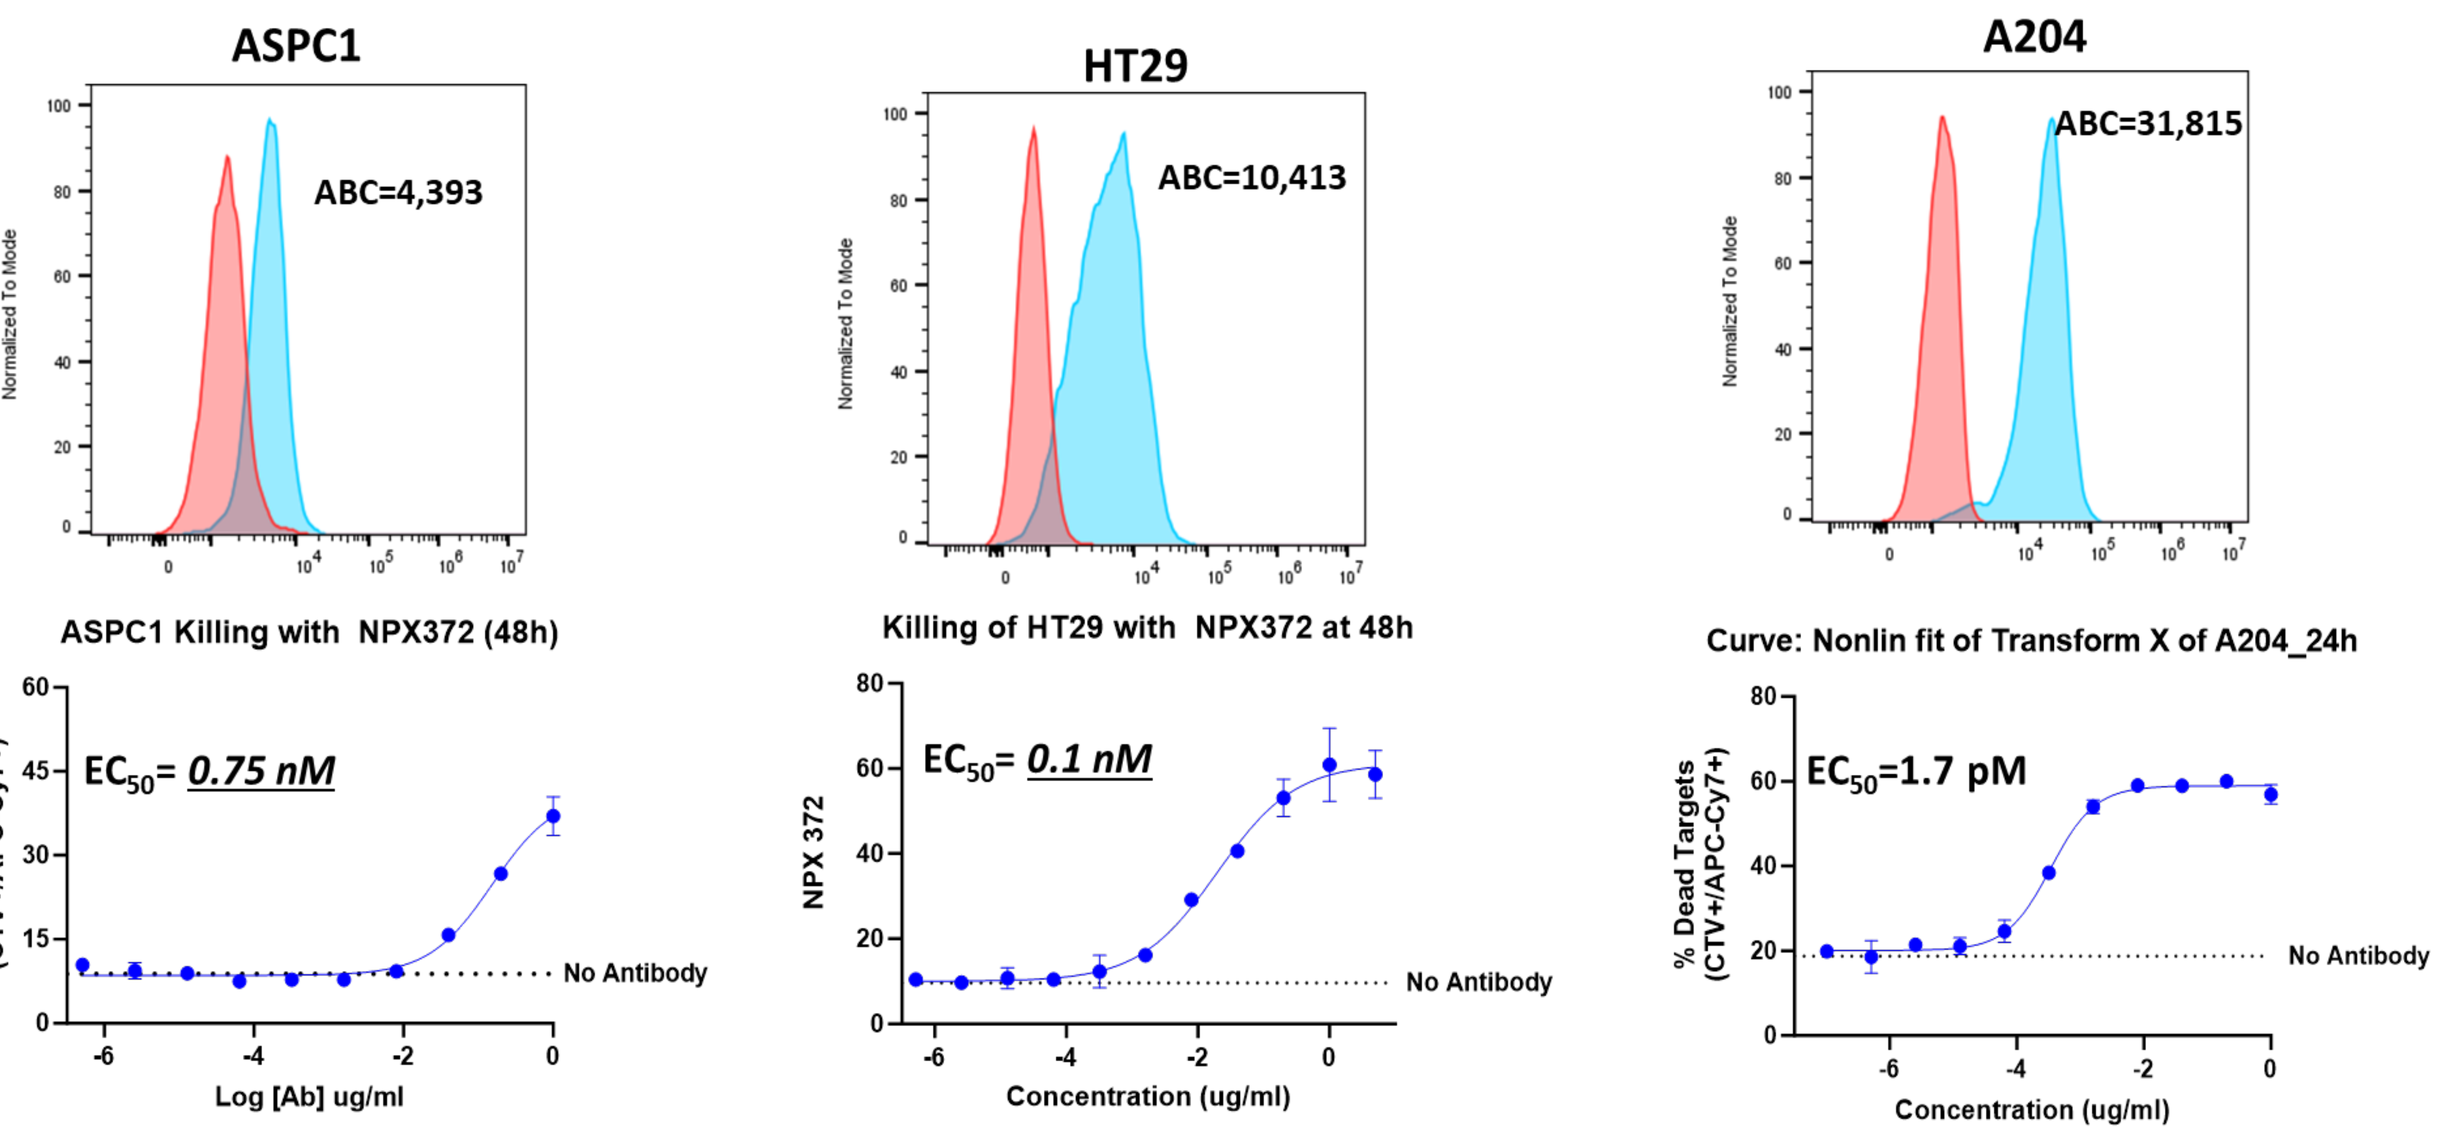
ASPC1
ABC=4,393
HT29
ABC=10,413
A204
ABC=31,815
ASPC1 Killing with NPX372 (48h)
Killing of HT29 with NPX372 at 48h
Curve: Nonlin fit of Transform X of A204_24h
EC50= 0.75 nM
EC50= 0.1 nM
EC50=1.7 pM
No Antibody
% Dead Targets (CTV+/APC-Cy7+)
Log [Ab] ug/ml
Concentration (ug/ml)

Tumor cells were stained with cell trace violet (CTV) and co-cultured with primary human PBMCs at an E to T ratio of 10:1 in the presence of serially diluted NPX372. After 24 hours (A204) or 48 hours (ASPC1 and HT29), the frequency of dead target cells (CTV+ viability dye+ cells) was measured by flow cytometry.

NPX372 Induces A204 Tumor Regression in an Activated T Cell Humanized Mouse Model at Clinically Relevant Doses

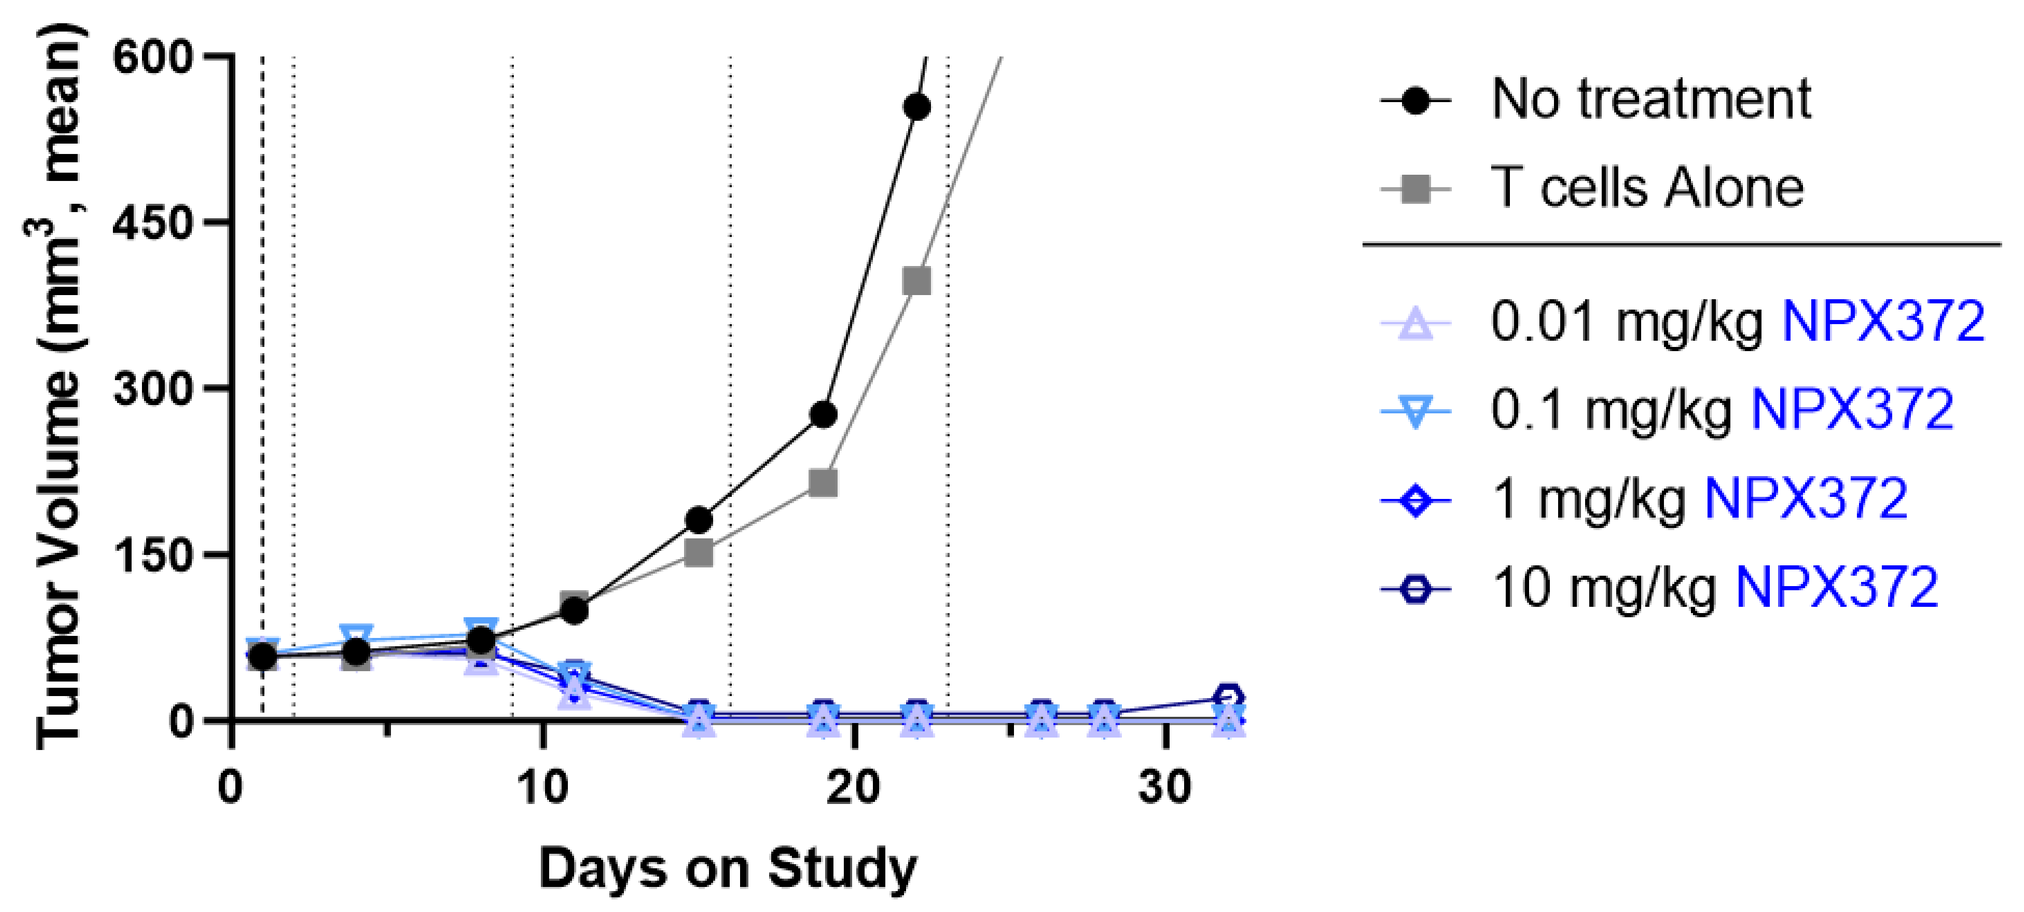
Tumor Volume (mm3, mean)
Days on Study
No treatment
T cells Alone
0.01 mg/kg NPX372
0.1 mg/kg NPX372
1 mg/kg NPX372
10 mg/kg NPX372

Female NCG mice with established subcutaneous A204 tumors were injected with activated T cells (2x10^7 cells) i.p. and NPX372 i.v. Antibody treatments were continued weekly for a total of four doses.

NPX372 Binds to Human and Cynomolgus Monkey B7-H7

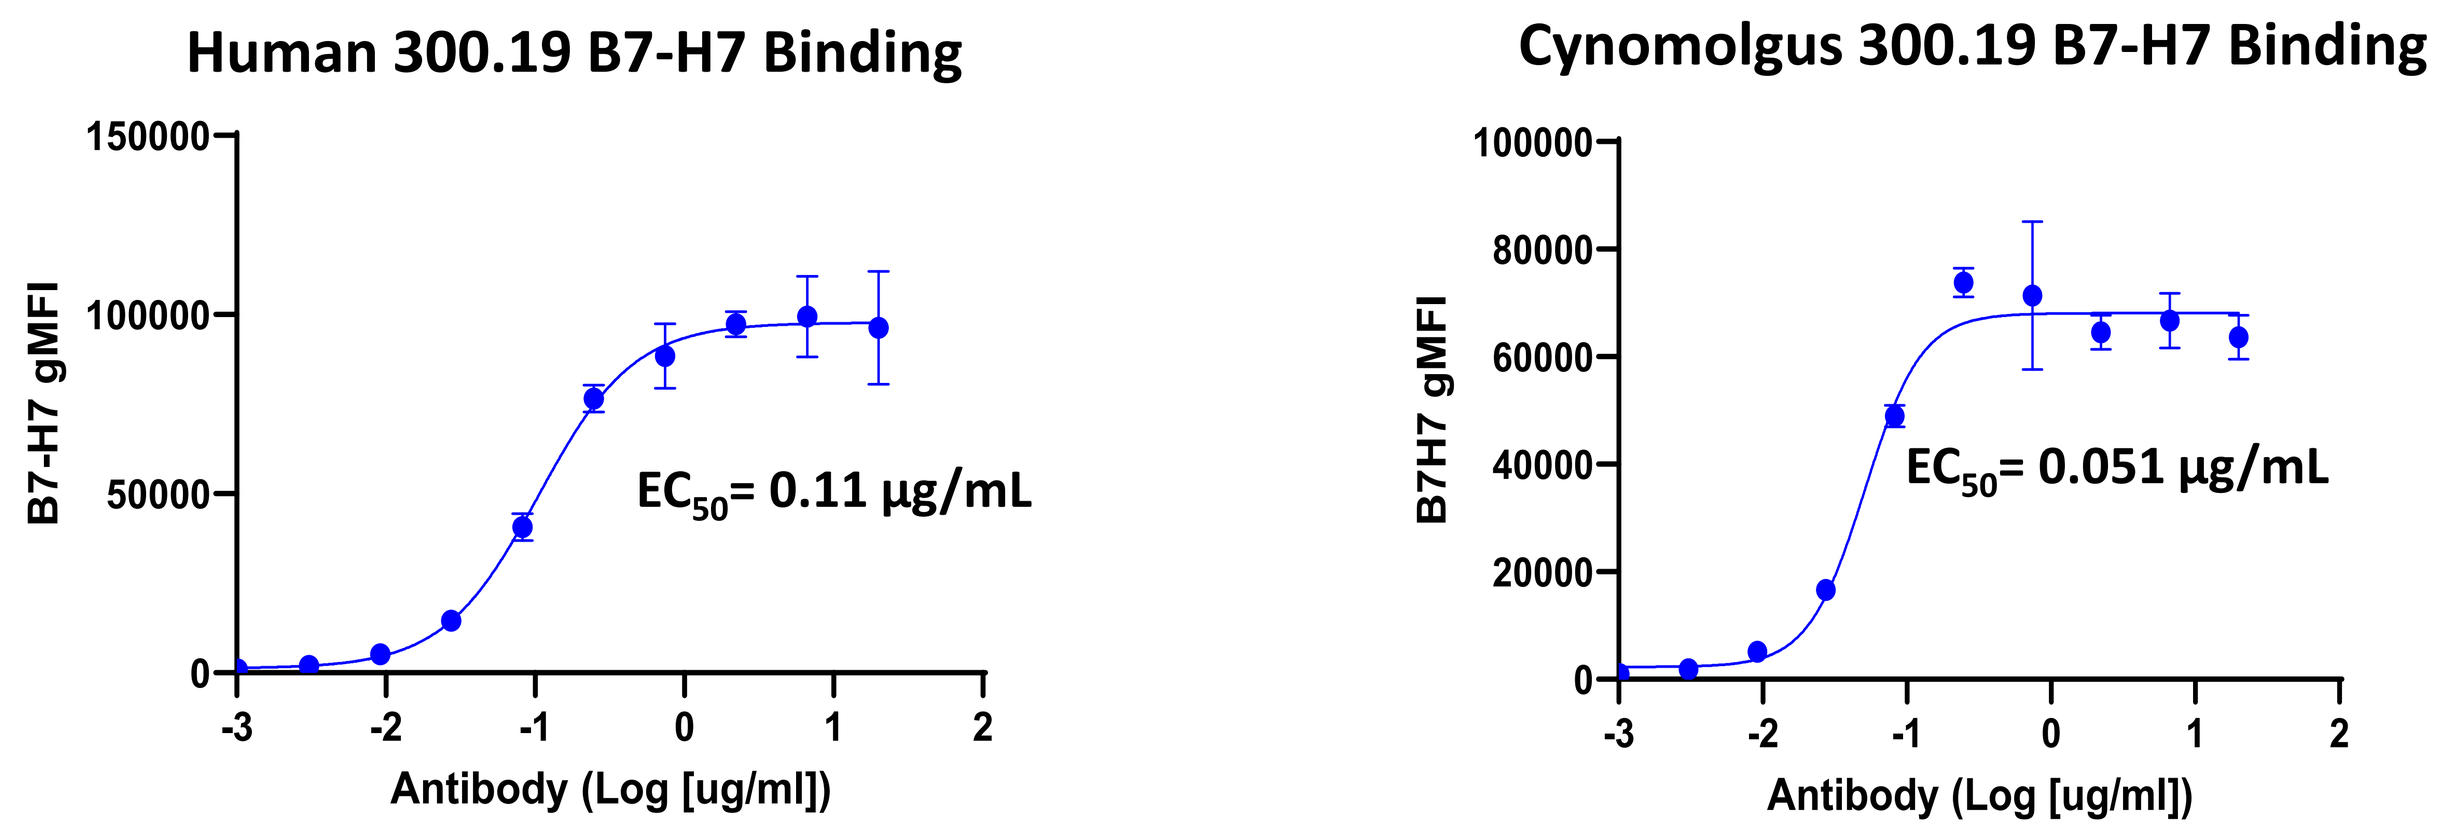
Human 300.19 B7-H7 Binding
B7-H7 gMFI
EC50= 0.11 µg/mL
Antibody (Log [ug/ml])
Cynomolgus 300.19 B7-H7 Binding
B7H7 gMFI
EC50= 0.051 µg/mL
Antibody (Log [ug/ml])

300.19 cells engineered to express human or cynomolgus monkey B7-H7 were treated with serially diluted NPX372, followed by addition of anti-human Fc secondary antibody for detection. Bispecific antibody binding to B7-H7 was analyzed by flow cytometry.

NPX372 Binding Kinetics		
	B7-H7 KD (M)	CD3 KD (M)
Human	3.33E-09	4.76E-09
Cyno	1.53E-09	4.22E-09

Similar NPX372 binding to both human and monkey recombinant B7-H7 and CD3 was also verified by Octet.

B7-H7 Expression Pattern is Similar in Human vs. Monkey Intestine

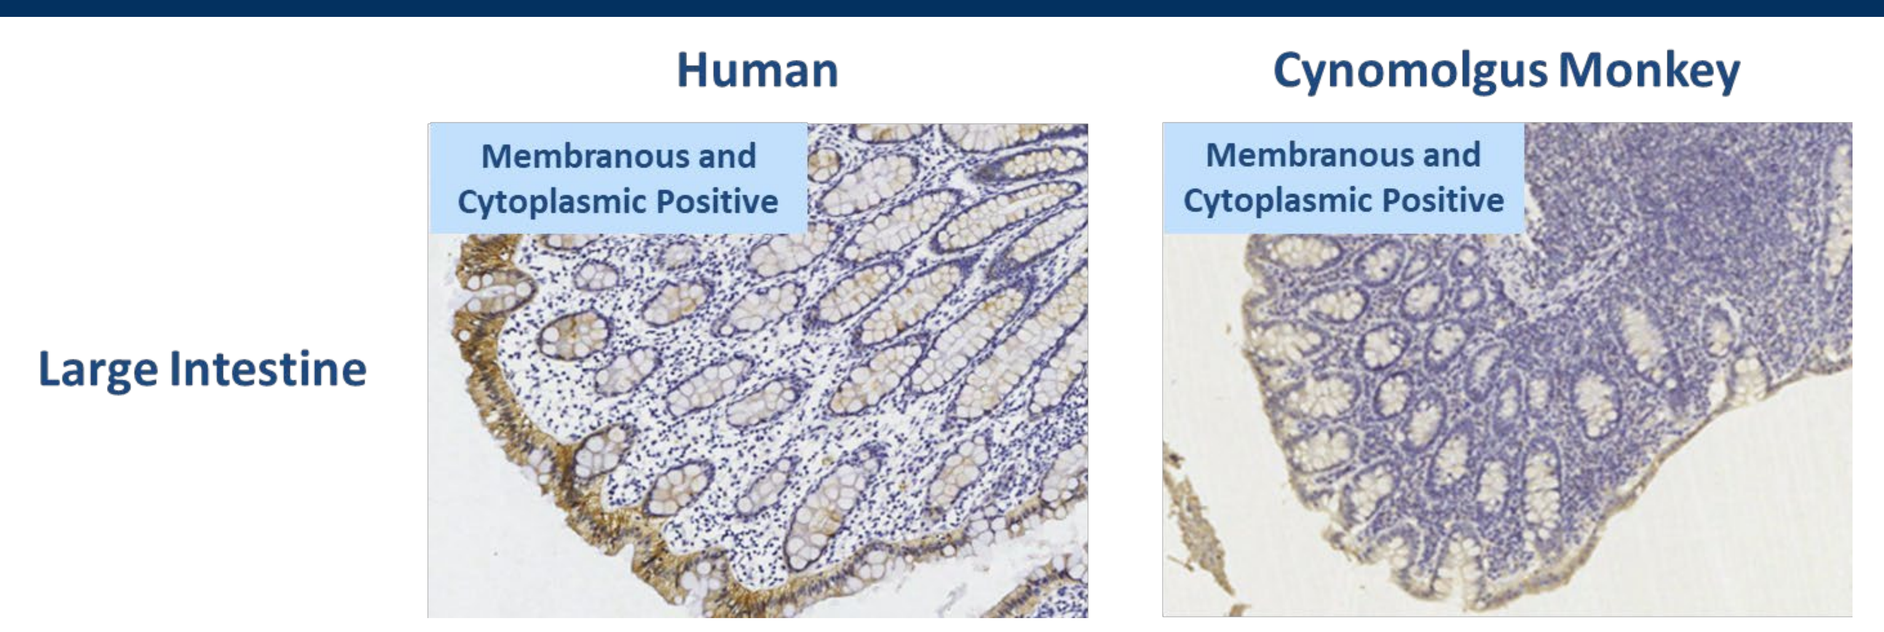
Human
Cynomolgus Monkey
Large Intestine
Membranous and Cytoplasmic Positive
Membranous and Cytoplasmic Positive

B7-H7 expression was evaluated with immunohistochemistry using E1U6X anti-B7-H7 antibody. B7-H7 expression is similar in human and monkey gut and restricted to specific epithelial substructures (Poster # 3302).

Non-GLP Toxicology Study Design in Non-human Primates

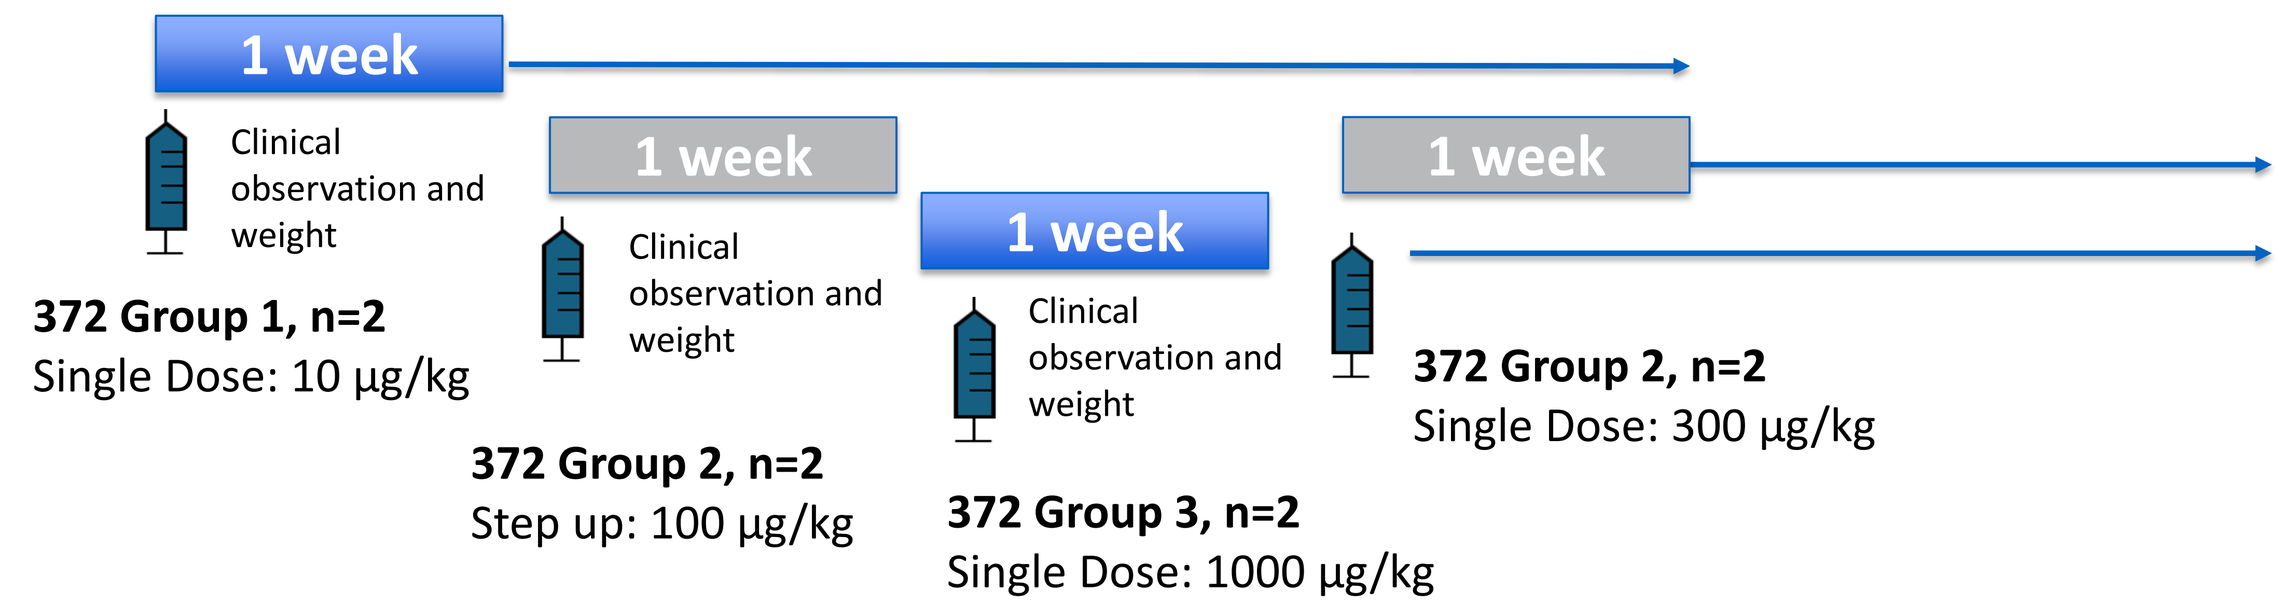
1 week
Clinical observation and weight
372 Group 1, n=2
Single Dose: 10 µg/kg
372 Group 2, n=2
Step up: 100 µg/kg
372 Group 3, n=2
Single Dose: 1000 µg/kg
372 Group 2, n=2
Single Dose: 300 µg/kg

A non-GLP study in non-human primates was performed to evaluate the tolerability and pharmacokinetic profile of NPX372. Monkeys were dosed at 10, 100, 300 or 1000 µg/kg via i.v. bolus, followed by a 28-day observation period for each group.

NPX372 is Safe at Relevant Doses in Non-human Primates

Group	N	Dose (µg/kg)	Cage Observations
1	2	10	No obvious abnormalities were found in both animals.
3	2	1000	Continuous loose to watery stools and loss of appetite were observed from Day 1 to Day 5 post dosing in both animals, and resolved by itself.
2	2	100	No obvious abnormalities were found in both animals.
		300	No obvious abnormalities were found in both animals.

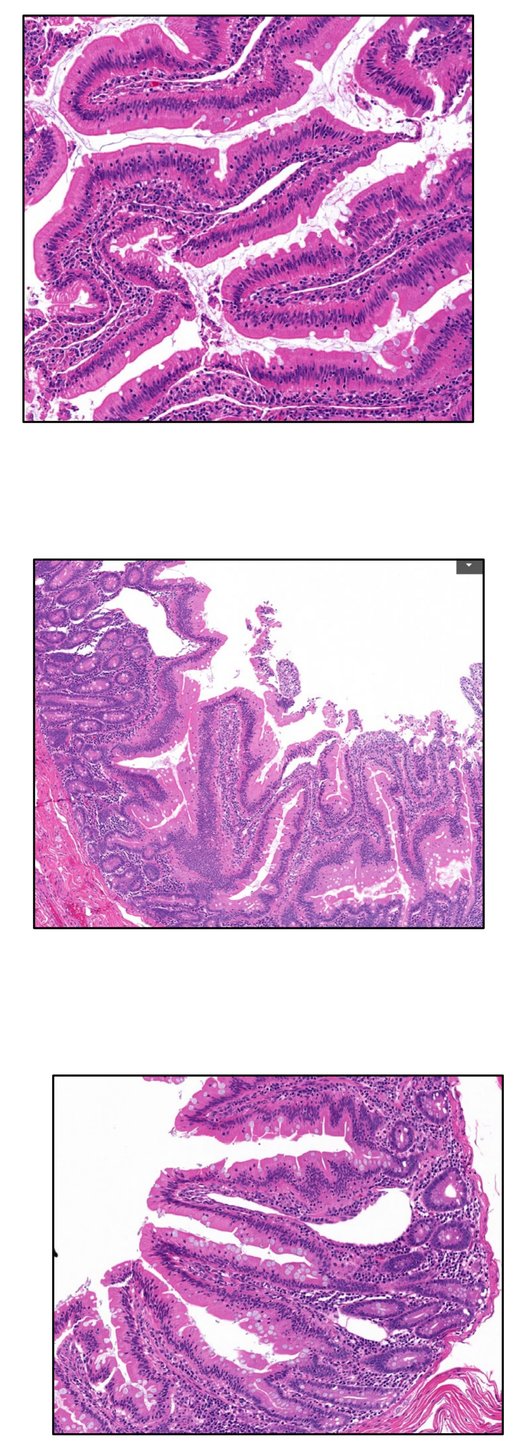

10 µg/kg
Normal Jejunum

100/300 µg/kg
Distinct and healthy villi

1000 µg/kg
Normal jejunum

NPX372 tolerated up to 1000 µg/kg

- No adverse events at/below 300 µg/kg
- GI toxicity and slight weight decrease observed at 1000 µg/kg; clinical symptoms fully resolved without intervention by Day 6

Normal pathology in all 13 tissues evaluated at Day 28 for all animals up to 1000 µg/kg

Dose Proportional Pharmacokinetics

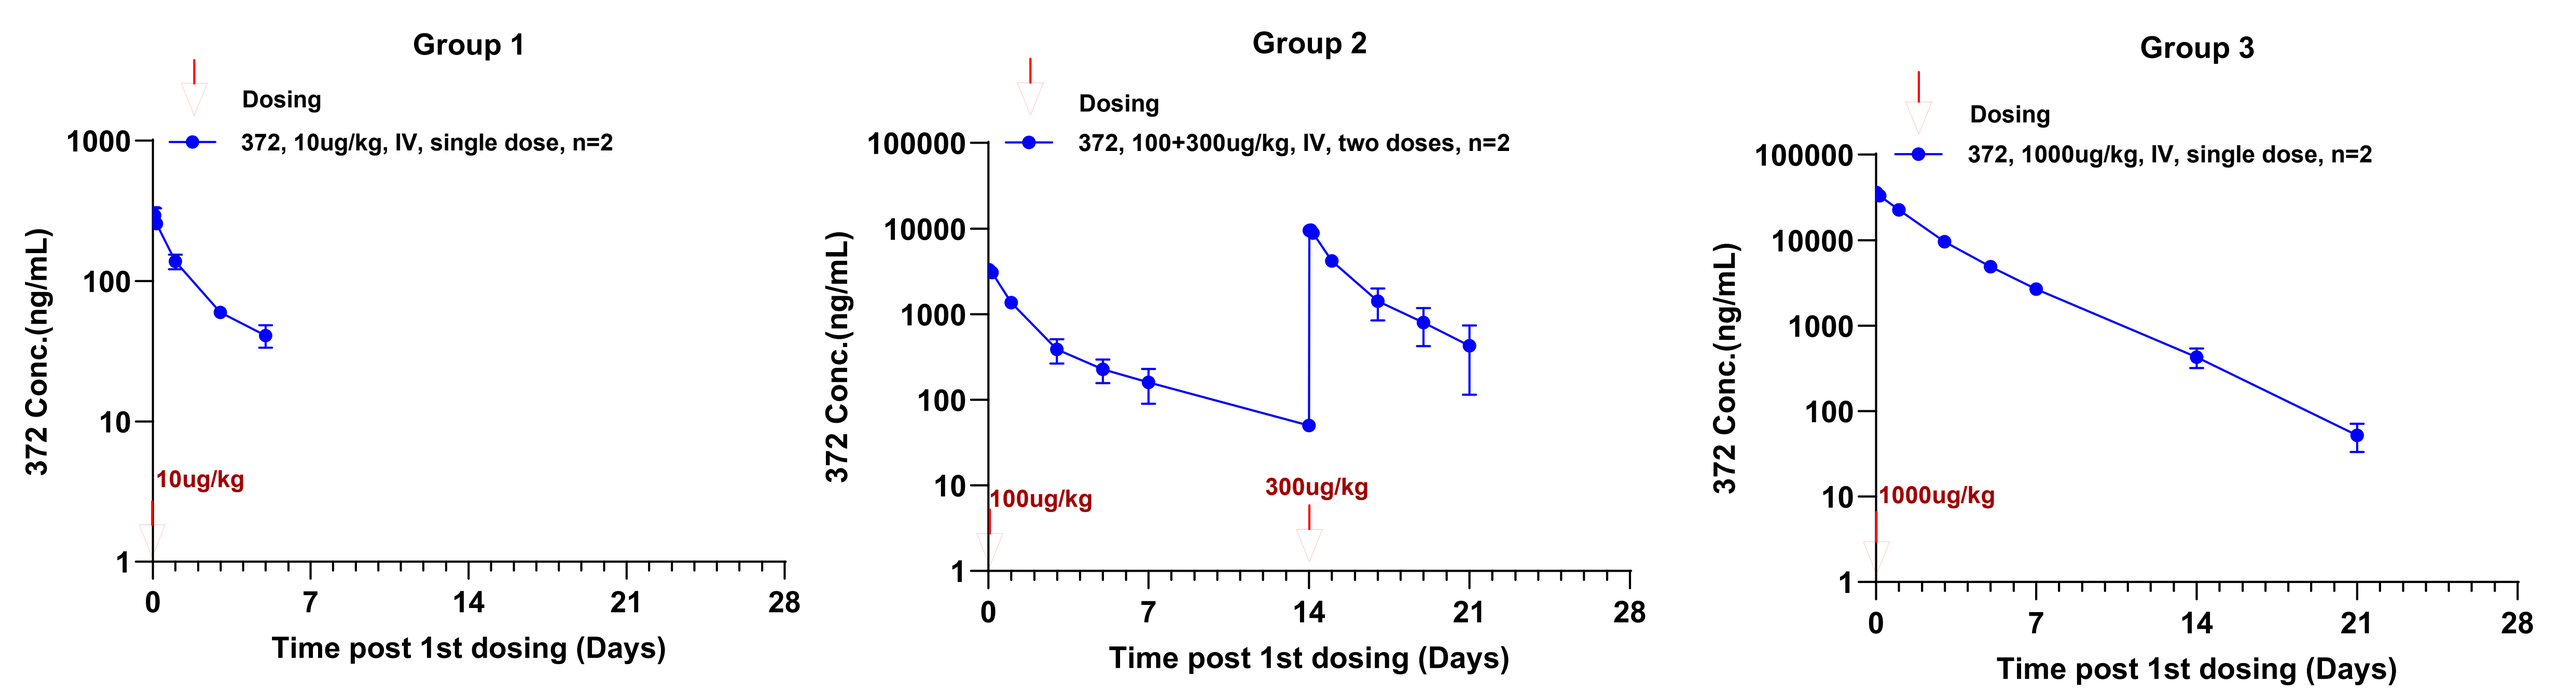
Group 1
Dosing
372, 10ug/kg, IV, single dose, n=2
10ug/kg
Group 2
372, 100+300ug/kg, IV, two doses, n=2
100ug/kg
300ug/kg
Group 3
372, 1000ug/kg, IV, single dose, n=2
1000ug/kg
372 Conc.(ng/mL)
Time post 1st dosing (Days)

NPX372 concentrations were evaluated with a qualified ELISA assay using biotinylated human B7-H7 as capturing reagent and HRP-human anti-CD3 as detection reagent. **Exposure (C_{max} and AUC) appeared dose dependent within the range of doses tested (10-1000 µg/kg).**

Immune Cell Activation Post-treatment; No Incidence of CRS

No abnormalities found in clinical pathology parameters for all doses/animals

- Clinical chemistry, coagulation, and CBC all normal
- Minor increases in CRP in one animal treated with 300 and 1000 µg/kg dose

Transient increases in G-CSF, IL-1RA, IL-8, MCP-1 and MIP-1b with possible dose proportionality

- One animal dosed at 1000 µg/kg showed an increase in IFNγ

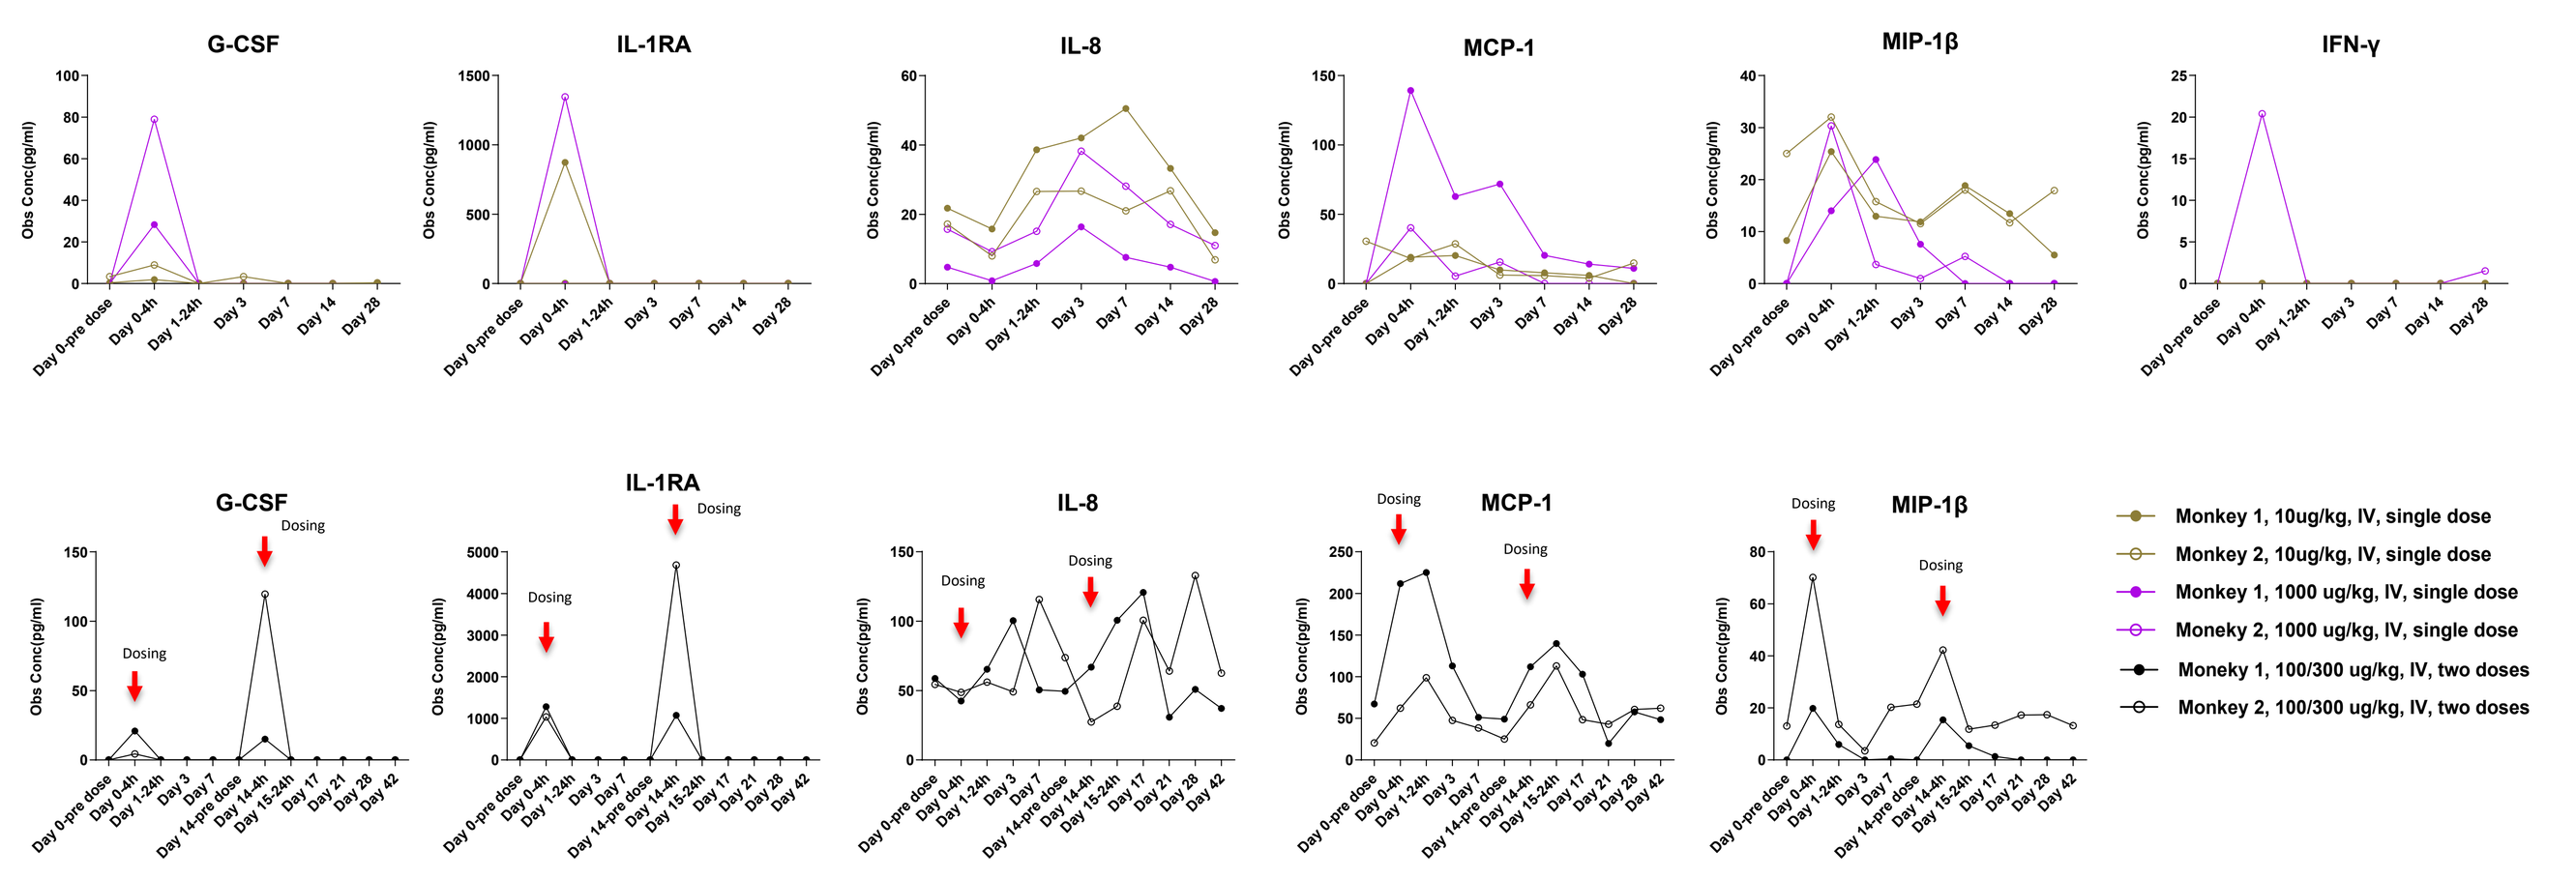
G-CSF
IL-1RA
IL-8
MCP-1
MIP-1β
IFN-γ
Obs Conc(pg/ml)
Dosing
Monkey 1, 10ug/kg, IV, single dose
Monkey 2, 10ug/kg, IV, single dose
Monkey 1, 1000 ug/kg, IV, single dose
Monkey 2, 1000 ug/kg, IV, single dose
Monkey 1, 100/300 ug/kg, IV, two doses
Monkey 2, 100/300 ug/kg, IV, two doses

Activation of T cells after NPX372 treatment

- Blood was collected at multiple timepoints after treatment with NPX372 and analyzed by flow cytometry
- Dose dependent CD4$^+$ and CD8$^+$ T cell activation was observed based on CD25$^+$ marker

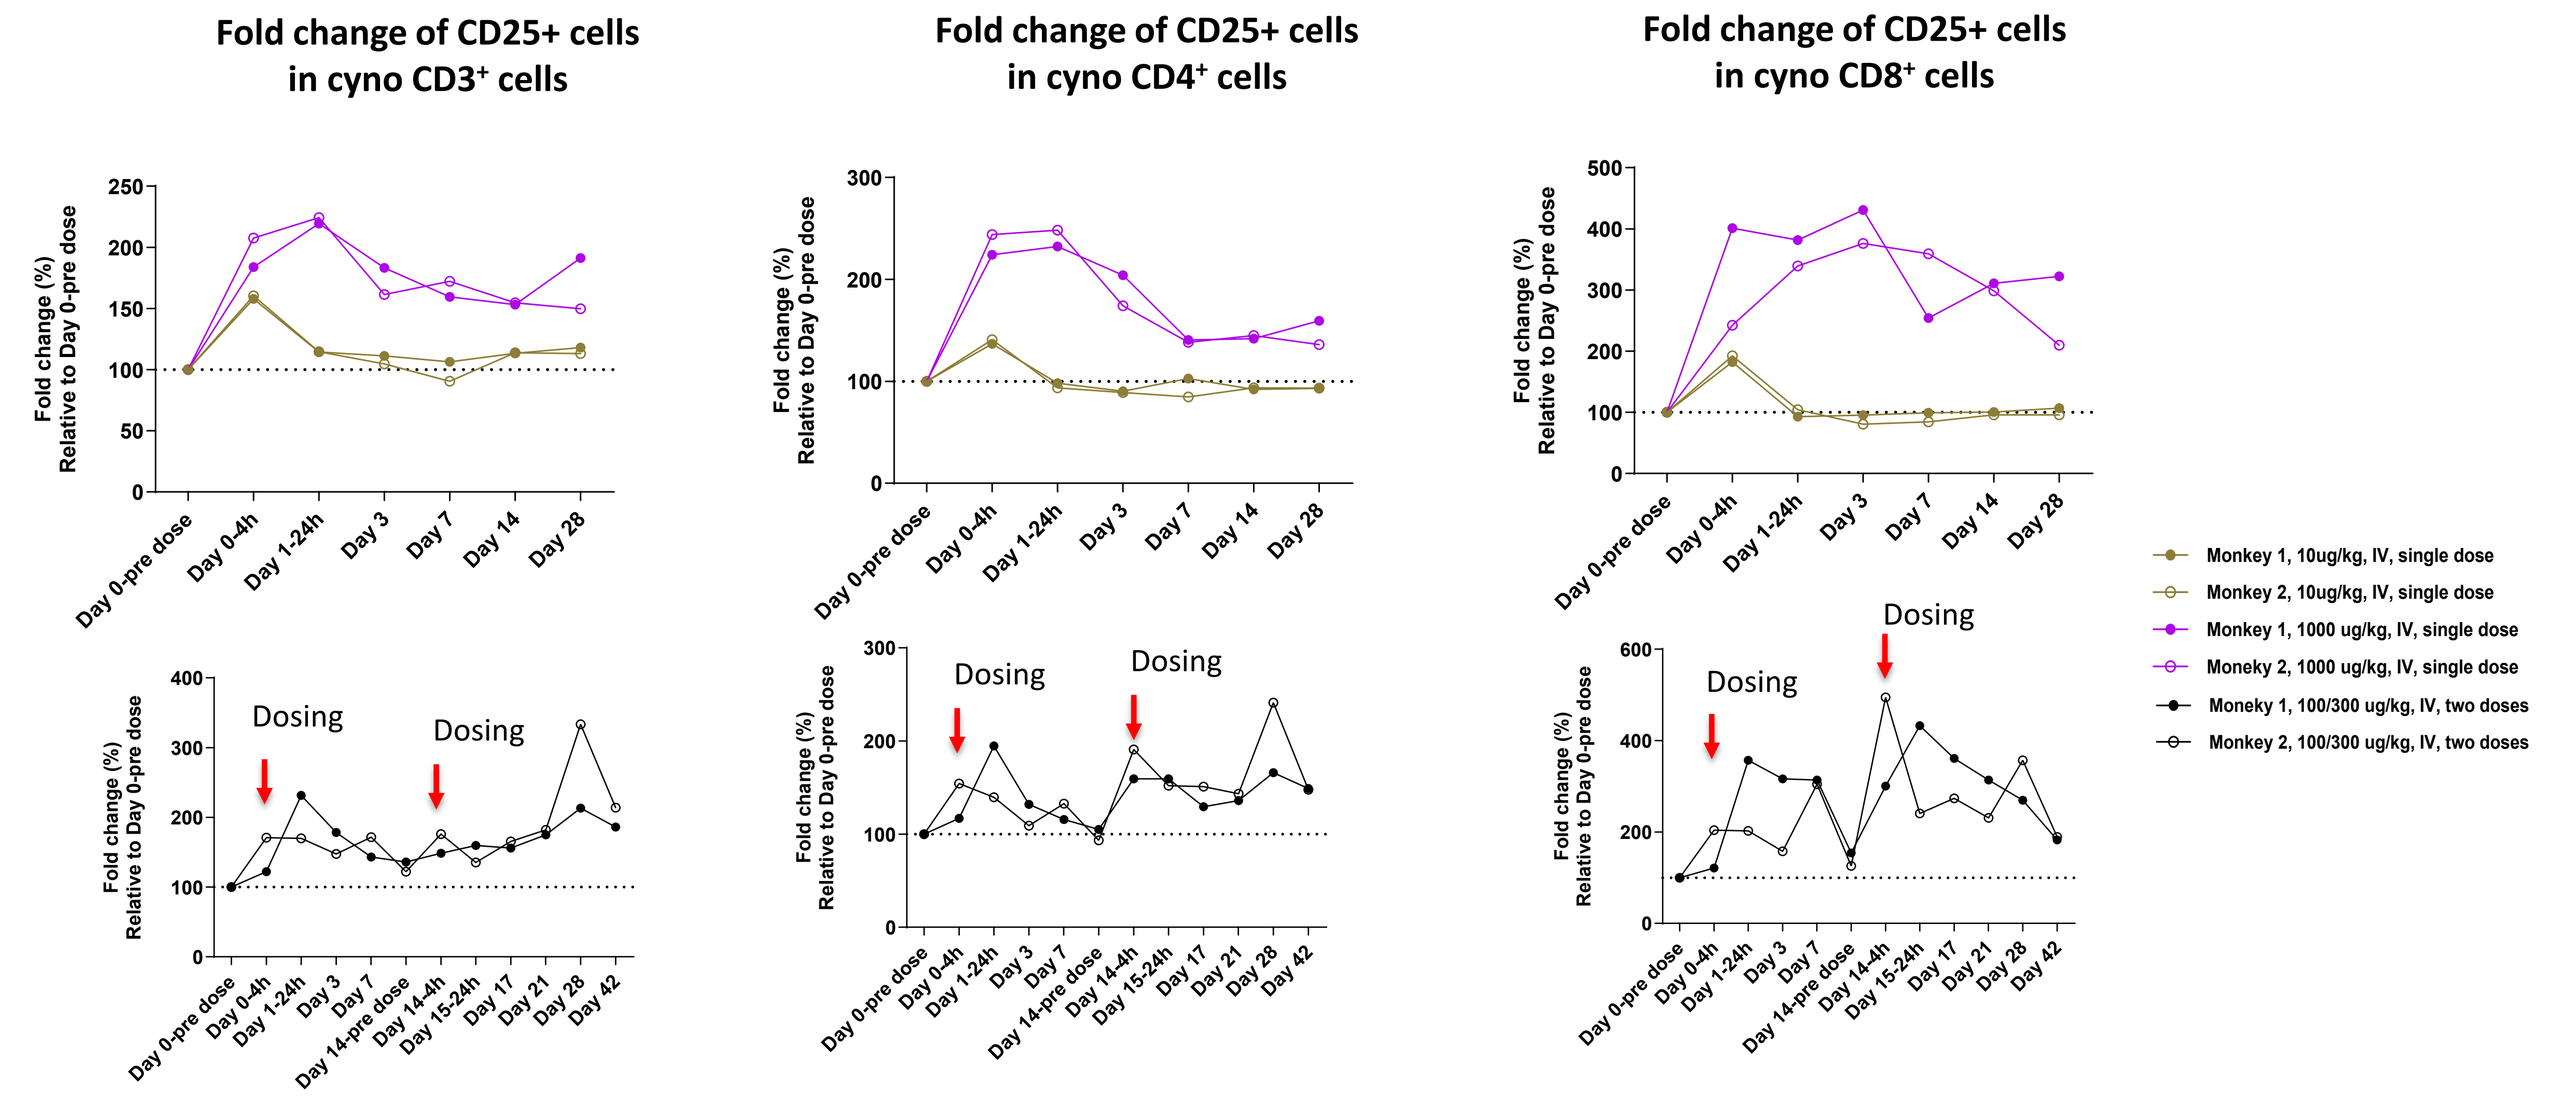
Fold change of CD25+ cells in cyno CD3+ cells
Fold change of CD25+ cells in cyno CD4+ cells
Fold change of CD25+ cells in cyno CD8+ cells
Fold change (%) Relative to Day 0-pre dose
Dosing
Monkey 1, 10ug/kg, IV, single dose
Monkey 2, 10ug/kg, IV, single dose
Monkey 1, 1000 ug/kg, IV, single dose
Monkey 2, 1000 ug/kg, IV, single dose
Monkey 1, 100/300 ug/kg, IV, two doses
Monkey 2, 100/300 ug/kg, IV, two doses

CONCLUSION

- **Cynomolgus monkey is a relevant toxicology species for NPX372 based on cross-reactivity of the bispecific and B7-H7 expression profile**
- **NPX372 induced T cell activation in NHP across doses, but no CRS was observed**
- **NPX372 was tolerated in monkeys at doses much higher than doses needed to induce tumor cell killing in vitro. Coupled with the absence of CRS, we anticipate a meaningful therapeutic window for dosing in humans with projected beneficial biological effects and manageable toxicity.**

nextpoint

AACR 2025